Oral Delivery of Non-live Preparation of *Veillonella parvula* Modulates CNS Inflammation via the Small Intestinal Axis: a Novel Approach to Treating Neuroinflammatory Diseases

Holly E. Ponichtera, Adam N. R. Cartwright, Aula Alami, Seamus Ragan, Will Caffry, Aylwin Ng, Maria Sizova, Nihal Okan, Valeria Kravitz, Kevin Huynh, Kritika Ramani, Christopher Davitt, Mary Abdou, David Emerson, Esra Uckun Kiran, Mehmedalija Jahic, Tanmoy Ganguly, Mark Bodmer, Andrea Itano

Evelo Biosciences, Cambridge MA

Rationale

The small intestinal axis (SINTAX) is a network of functional anatomical connections between the gut and the rest of the body. SINTAX can be targeted with orally-delivered, gut-restricted agents that have systemic anti-inflammatory effects, including in the CNS. EDP1867 is a pharmaceutical preparation of a single strain of *Veillonella parvula* isolated from the ileum of a human donor. During manufacturing it is γ-irradiated after fermentation making it a non-live biotherapeutic product. EDP1867 has pharmacological activity via SINTAX in T cell-mediated murine inflammation models, reducing peripheral inflammation without toxicity. We investigated whether EDP1867 was also efficacious in a model of chronic neuroinflammation and demyelination. This might suggest benefit in multiple sclerosis (MS).

EDP1867

EDP1867 is a non-live, gut-restricted strain of *V. parvula* that modulates human and mouse immune cell responses to resolve inflammation without toxic effects

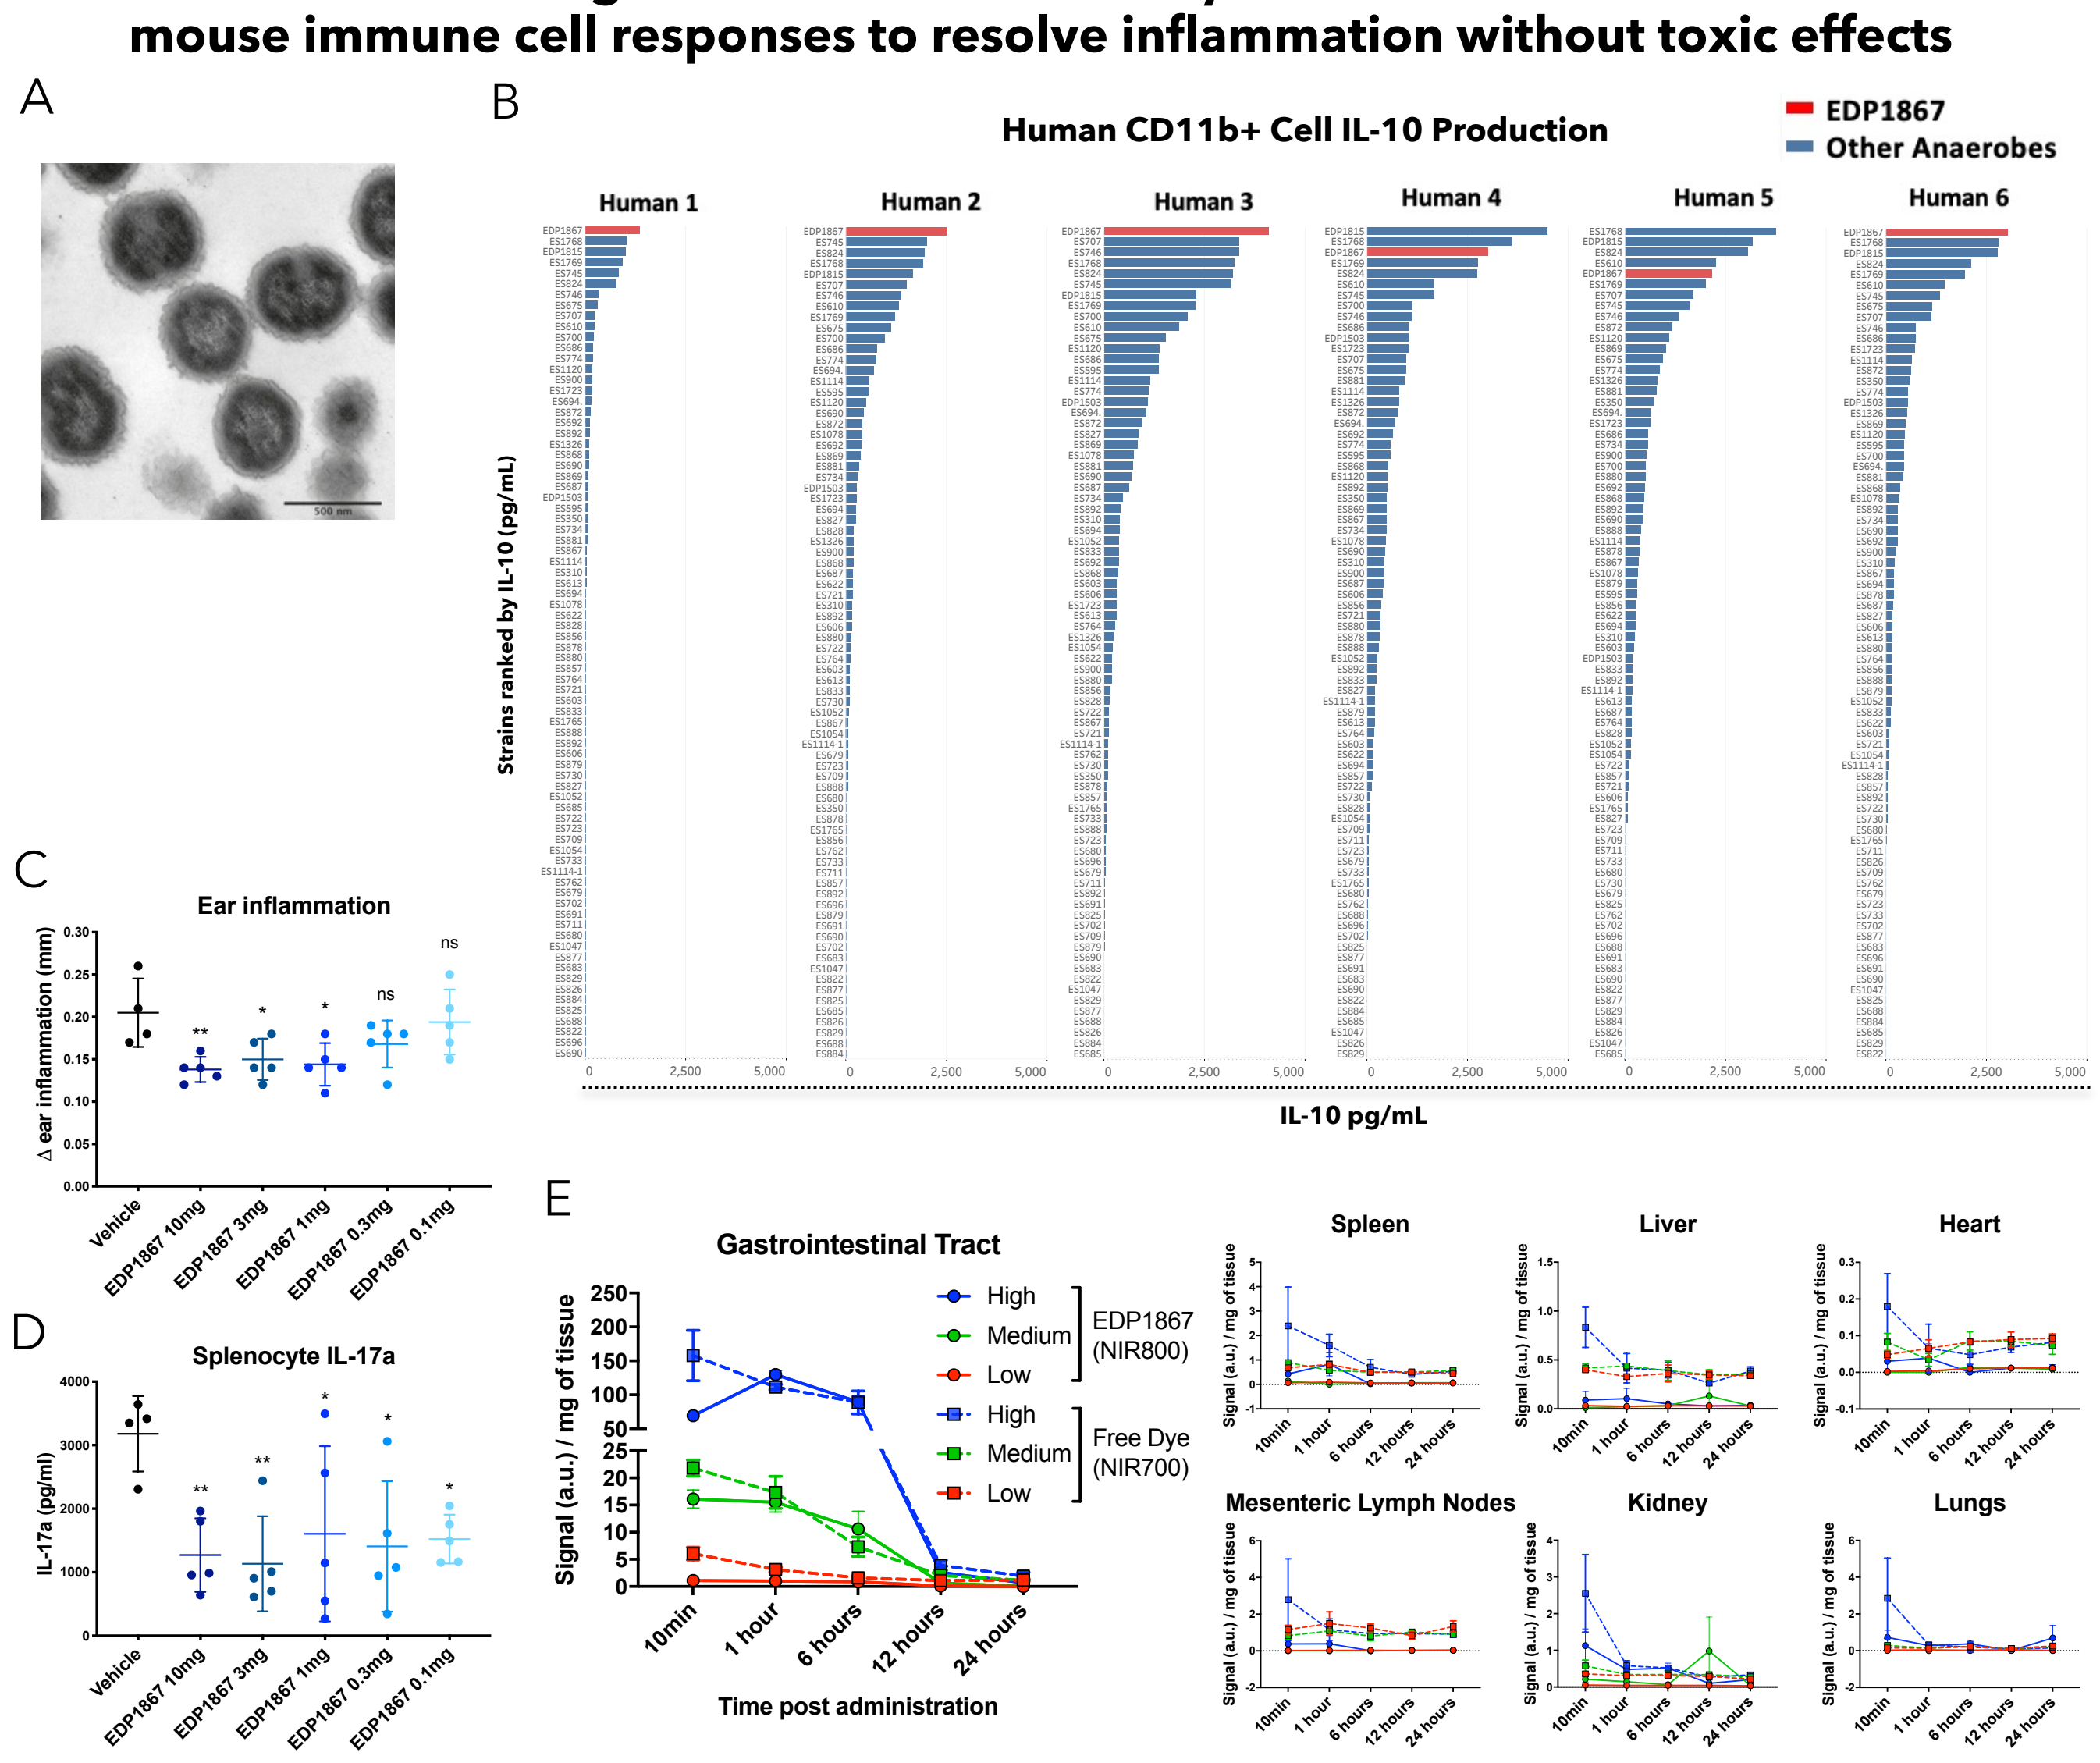
A
B
Human CD11b+ Cell IL-10 Production
EDP1867
Other Anaerobes
Human 1
Human 2
Human 3
Human 4
Human 5
Human 6
Strains ranked by IL-10 (pg/mL)
IL-10 pg/mL
C
Ear inflammation
Δ ear inflammation (mm)
D
Splenocyte IL-17a
IL-17a (pg/ml)
Vehicle
EDP1867 10mg
EDP1867 3mg
EDP1867 1mg
EDP1867 0.3mg
EDP1867 0.1mg
E
Gastrointestinal Tract
High
Medium
Low
EDP1867 (NIR800)
Free Dye (NIR700)
Signal (a.u.) / mg of tissue
Time post administration
10min
1 hour
6 hours
12 hours
24 hours
Spleen
Liver
Heart
Mesenteric Lymph Nodes
Kidney
Lungs

Figure 1. EDP1867 displayed anti-inflammatory effects in pre-clinical human and mouse assays
A) EDP1867 is a single strain of *V. parvula,* a gram negative obligate anaerobic cocci. Transmission electron microscopy image[1].
B) In an *in vitro* assay with primary human CD11b+ peripheral blood mononuclear cells from 6 human donors, EDP1867 induced high levels of the anti-inflammatory cytokine IL-10 compared to 78 other anaerobic microbes.
C) In a Th17-mediated mouse psoriasis model EDP1867 reduced ear inflammation in a dose-dependent manner compared to vehicle treated mice. 5% imiquimod cream was applied to the ears of C57BL/6 mice every 24 hr for 7 days and change in ear swelling was measured using a caliper 24 hr after the last application.
D) In psoriatic mice, EDP1867 treatment reduced IL-17a production by splenocytes after 48 hr of PMA/I stimulation *ex vivo*. Statistics display ordinary one-way ANOVA of treatment vs. vehicle (*p≤0.05, **p≤0.01, ***p≤0.001, p, ****p≤0.0001).
E) A biodistribution study in naïve mice displayed that orally-administered EDP1867 is restricted to the gut lumen and not detectable after 12 hr post-treatment. Naïve mice were orally-administered fluorescently labeled EDP1867 or free dye at high, medium, and low doses and biodistribution was assessed at 10 min, 1 hr, 6 hr, 12 hr and 24hr on the LI-COR Pearl Imager.

Methods

Orally-administered EDP1867 was tested in the proteolipid protein (PLP)-induced relapsing-remitting Experimental Autoimmune Encephalomyelitis (EAE) SJL mouse model. EAE was induced in SJL mice by injection of $PLP_{139\text{-}151}$ emulsified in complete Freund's adjuvant (CFA) on day 0. To test prophylactic treatment mice received a 10 mg oral daily dose of EDP1867 or vehicle from day 0-42. To test therapeutic treatment mice received EDP1867 or vehicle starting on the second day of disease. Clinical score was assessed daily from day 9-42 and statistical analyses of mean maximum score (MMS), and relapse incidence were performed. Histological analysis was carried out to assess inflammation and demyelination in spinal cord. Small intestine tissue was assessed by RNA sequencing and gene set enrichment analysis (GSEA).

Results

Oral treatment with EDP1867 reduced disease severity and incidence of relapse in the relapsing-remitting EAE mouse model of multiple sclerosis

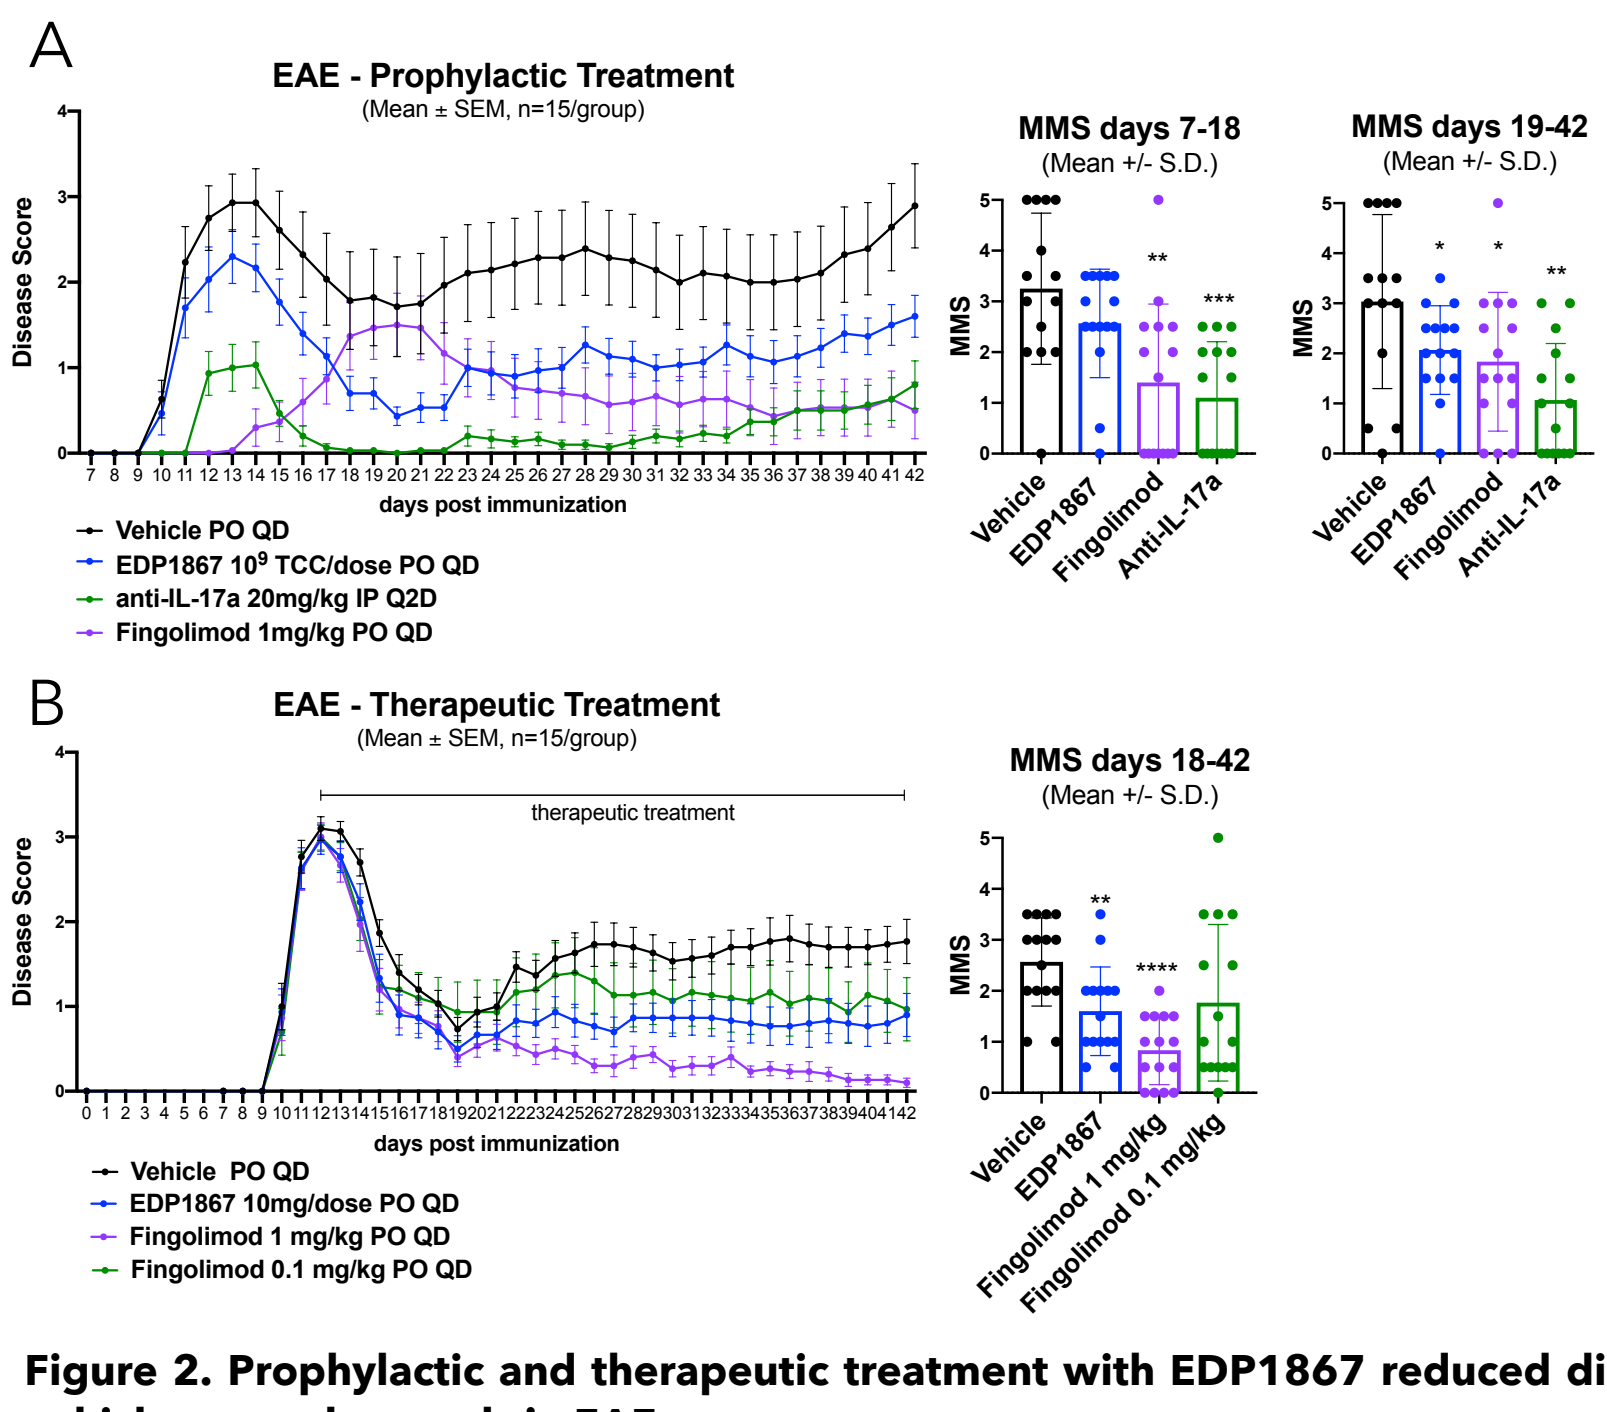
A
EAE - Prophylactic Treatment
(Mean ± SEM, n=15/group)
Disease Score
days post immunization
Vehicle PO QD
EDP1867 10⁹ TCC/dose PO QD
anti-IL-17a 20mg/kg IP Q2D
Fingolimod 1mg/kg PO QD
MMS days 7-18
(Mean +/- S.D.)
MMS days 19-42
(Mean +/- S.D.)
Vehicle
EDP1867
Fingolimod
Anti-IL-17a
B
EAE - Therapeutic Treatment
(Mean ± SEM, n=15/group)
therapeutic treatment
Vehicle PO QD
EDP1867 10mg/dose PO QD
Fingolimod 1 mg/kg PO QD
Fingolimod 0.1 mg/kg PO QD
MMS days 18-42
(Mean +/- S.D.)
Fingolimod 1 mg/kg
Fingolimod 0.1 mg/kg

Table 1. EAE Scores for Prophylactic Treatment

Treatment	EAE incidence until Day 18 (%)	MMS until Day 18 +/- SD	p value	Incidence of relapse	MMS of relapse period +/- SD (Days 19-42)	p value	End score +/- SD	p value
Vehicle	93.3%	3.25 +/- 1.49		77.8%	3.04 +/- 1.74		2.89 +/- 1.84	
EDP1867	93.3%	2.57 +/- 1.07	0.2403	64.3%	2.07 +/- 0.88	0.0407	1.60 +/- 0.95	0.0190
anti-IL-17A	53.3%	1.10 +/- 1.11	0.0005	40.0%	1.07 +/- 1.13	0.0021	0.80 +/- 1.08	0.0027
Fingolimod, 1 mg/kg	53.3%	1.40 +/- 1.55	0.0045	0.0%	1.83 +/- 1.38	0.0326	0.50 +/- 1.28	0.0024

Table 2. EAE Scores for Therapeutic Treatment

Treatment	Mean day of onset +/- SD	Score at enrollment +/- SD	Incidence of relapse	MMS of relapse period +/- SD (Days 18-42)	p value	End score +/- SD	p value
Vehicle	10.4 +/- 0.5	3.03 +/- 0.48	73.3%	2.57 +/- 0.86		1.77 +/- 1.02	
EDP1867	10.5 +/- 0.5	2.90 +/- 0.69	46.7%	1.60 +/- 0.87	0.0069	0.90 +/- 0.99	0.0284
Fingolimod, 1 mg/kg	10.5 +/- 0.7	2.77 +/- 0.53	0.0%	0.83 +/- 0.67	<0.0001	0.10 +/- 0.21	<0.0001
Fingolimod, 0.1 mg/kg	10.5 +/- 0.7	3.00 +/- 0.46	28.6%	1.77 +/- 1.53	0.0814	0.97 +/- 1.45	0.0285

Figure 2. Prophylactic and therapeutic treatment with EDP1867 reduced disease severity and incidence of relapse compared to vehicle-treated controls in EAE
A) Prophylactic EDP1867 treatment reduced disease score and MMS in both the acute phase between days 7-18 as well as the relapse period from day 19-42. Incidence of relapse was reduced to 64.3% in EDP1867-treated mice compared to 77.8% in vehicle-treated mice **(Table 1)**. Statistics compare treatment to vehicle using Wilcoxon's non-parametric test.
B) Therapeutic EDP1867 treatment reduced disease score and MMS in the relapse period from day 18-42. Incidence of relapse was reduced to 46.7% in EDP1867-treated mice compared to 73.3% in vehicle-treated mice **(Table 2)**.

References
1. Front Microbiol (2017); 8: 1215. https://doi.org/10.3389/fmicb.2017.01215
2. J Exp Med (2006) 203 (3): 497–500. https://doi.org/10.1084/jem.20060227

Oral treatment with EDP1867 reduced inflammation and demyelination in the CNS

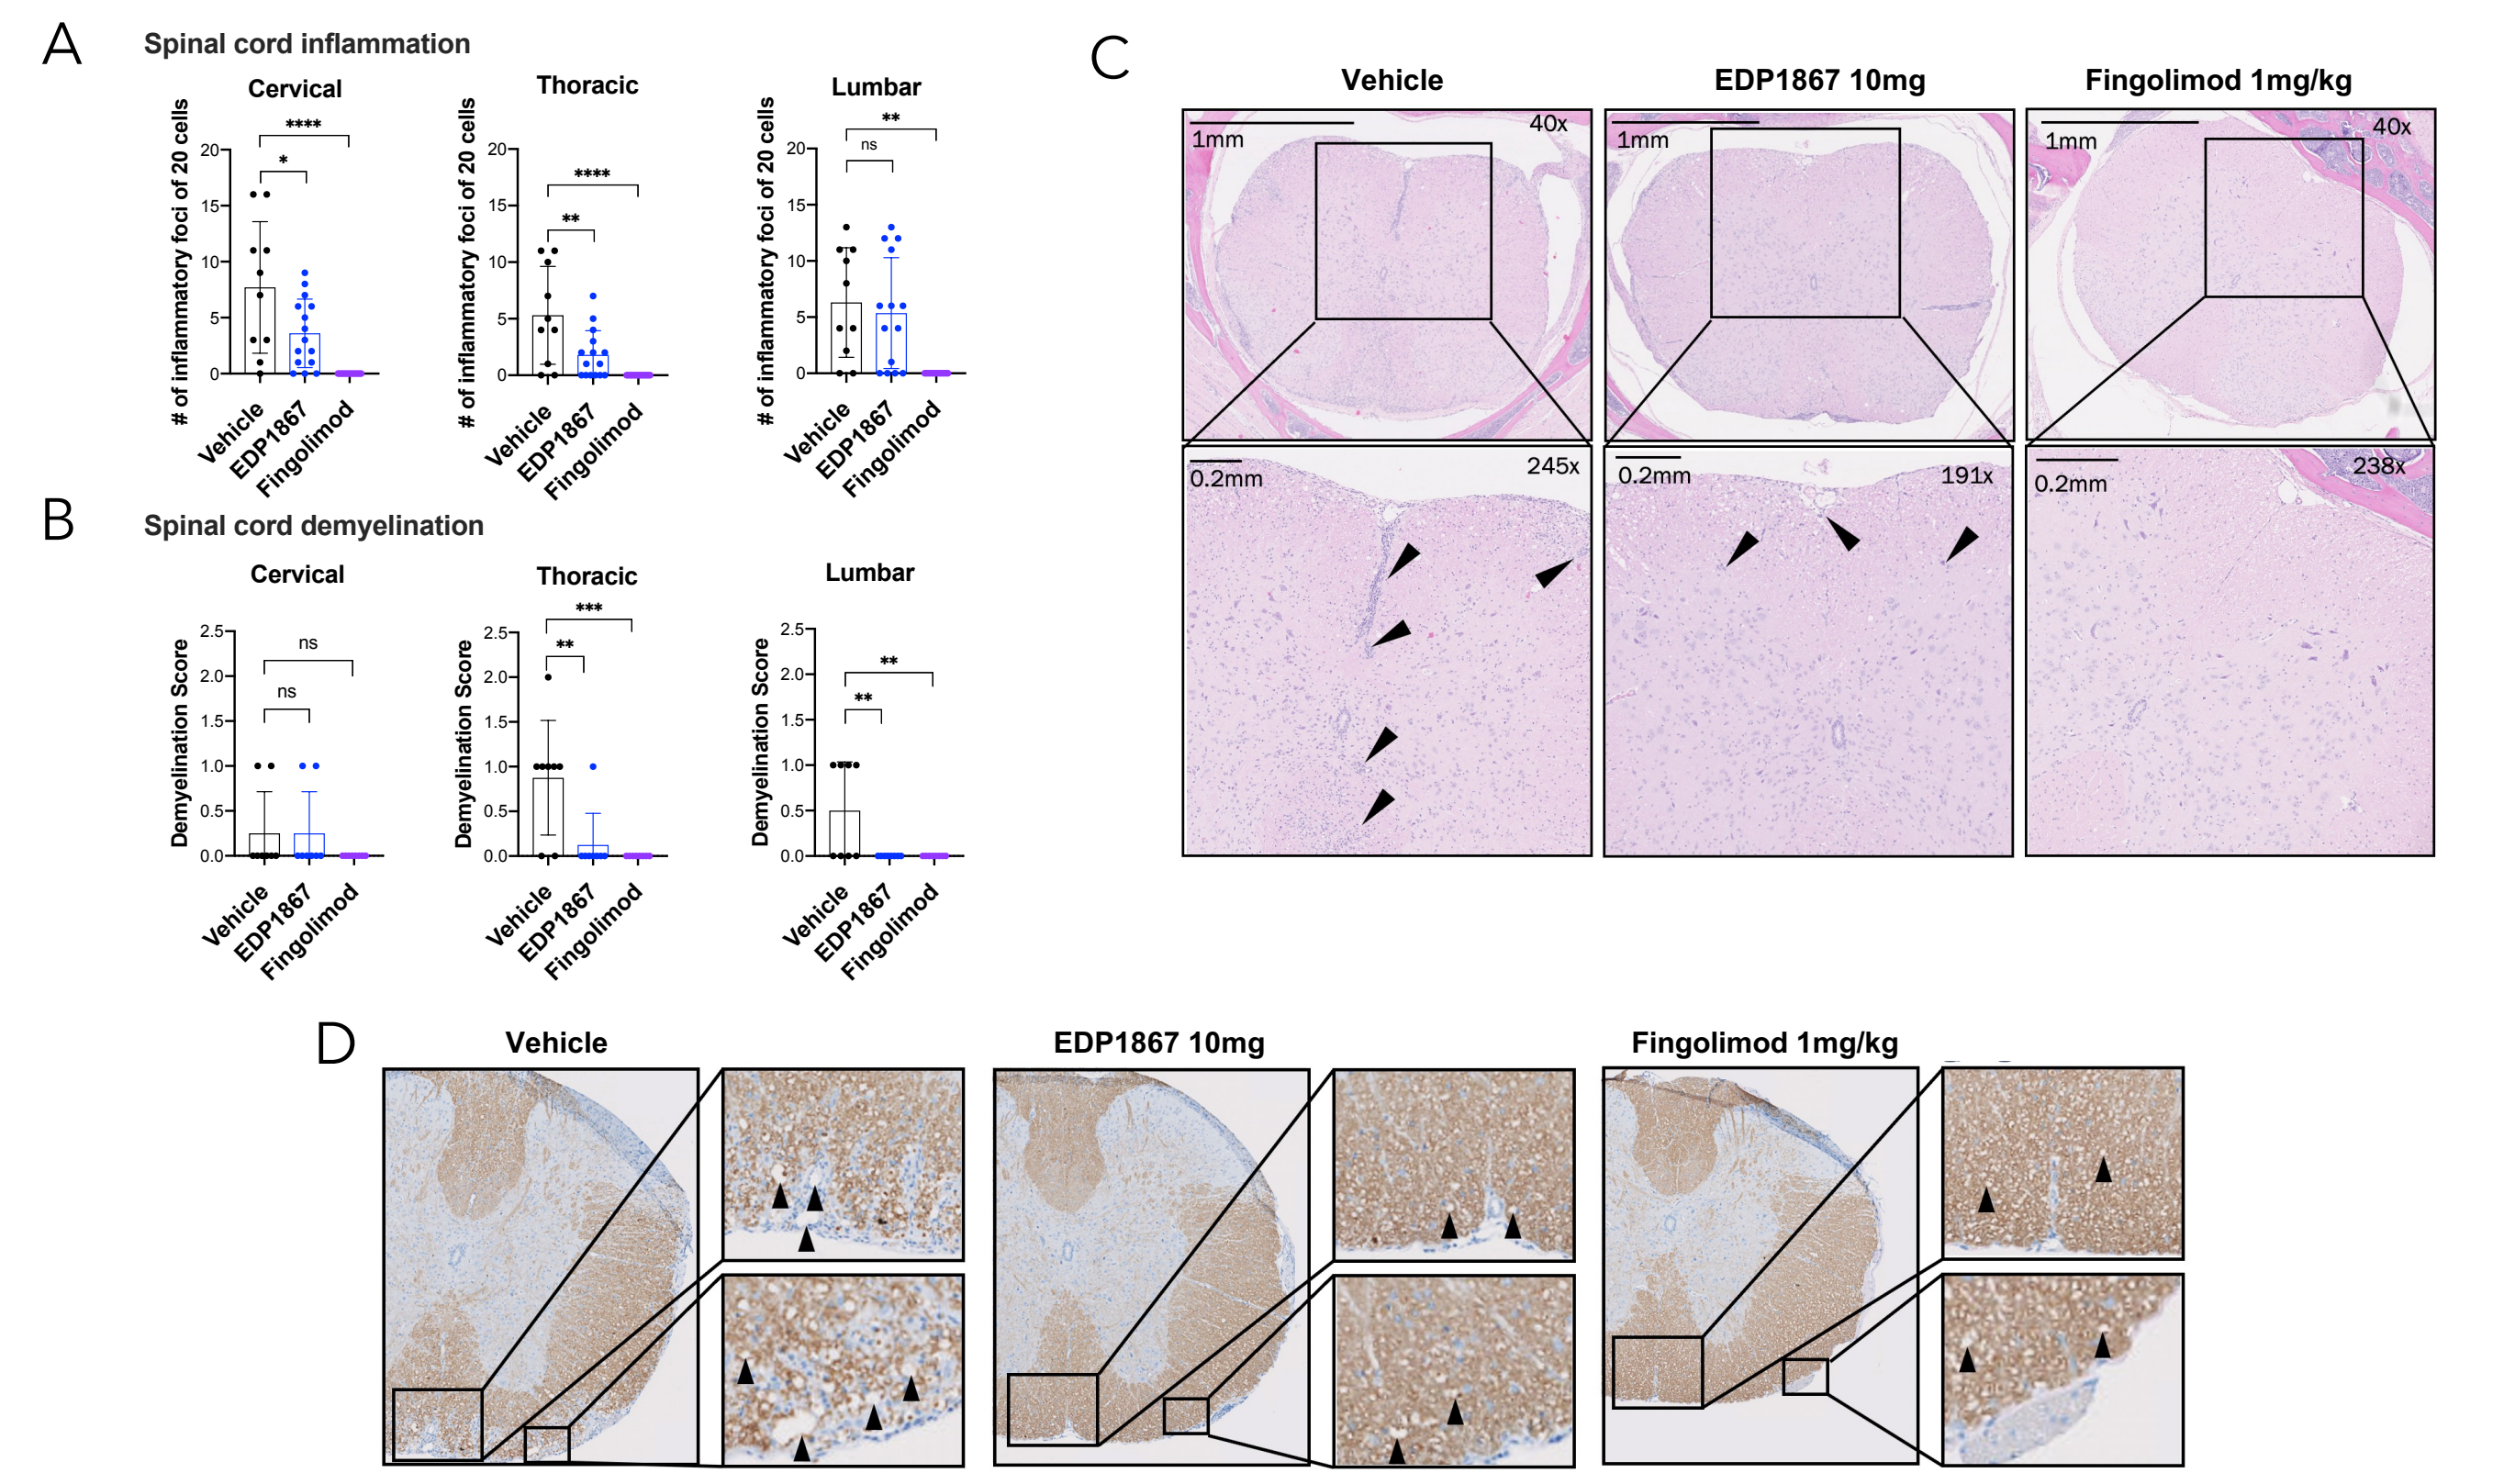
A
Spinal cord inflammation
Cervical
Thoracic
Lumbar
of inflammatory foci of 20 cells
Vehicle
EDP1867
Fingolimod
B
Spinal cord demyelination
Demyelination Score
C
Vehicle
EDP1867 10mg
Fingolimod 1mg/kg
1mm
40x
0.2mm
245x
191x
238x
D

Figure 3. Histopathological analysis of spinal cord tissue sections from EDP1867-treated mice showed reduced infiltration of inflammatory cells and reduced demyelination
A) Prophylactic treatment with EDP1867 reduced the number of inflammatory cells in the cervical and thoracic spinal cord regions as shown by histopathology scoring of inflammatory foci in H&E-stained tissue sections taken from EAE diseased mice on day 42. Statistics display ordinary one-way ANOVA of treatment vs. vehicle (*p≤0.05, **p≤0.01, ***p≤0.001, p, ****p≤0.0001).
B) Therapeutic treatment with EDP1867 reduced demyelination in the thoracic and lumbar spinal cord regions as shown by histopathology scoring of anti-myelin basic protein (MBP)-stained tissue sections taken from EAE diseased mice on day 42. Scoring: 0, <5% demyelinated area; 1, 5-20% demyelinated area; 2, 20-40% demyelinated area; 3, 40-60% demyelinated area.
C) Representative images of H&E-stained tissue sections from the cervical spinal cord region with arrows indicating accumulations of immune cells.
D) Representative images of anti-MBP-stained tissue sections from the cervical spinal cord region with arrows indicating areas of demyelination and loss of structure.

EDP1867 acts on cells of the small intestine to resolve peripheral inflammation

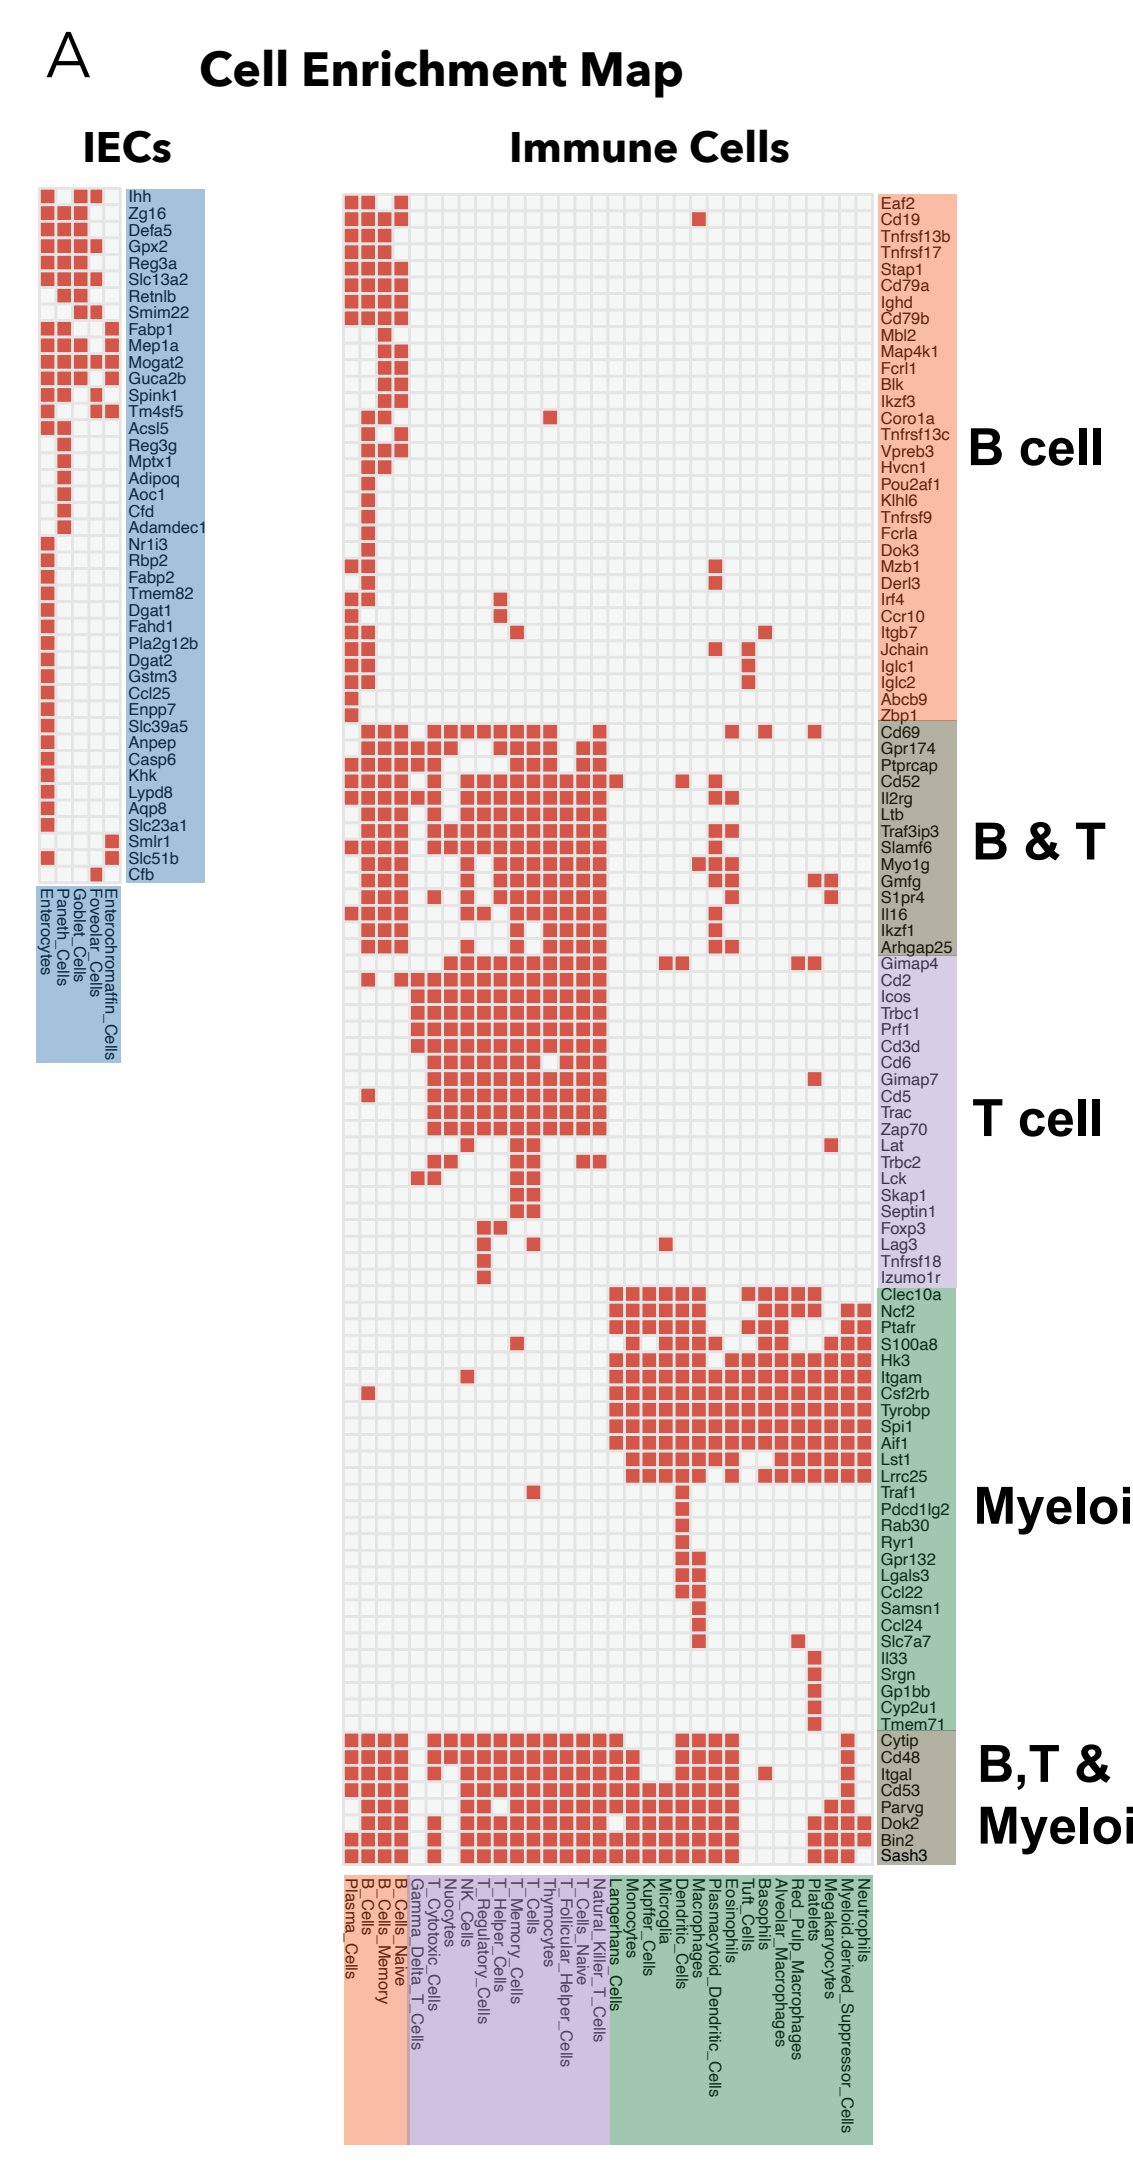
A
Cell Enrichment Map
IECs
Immune Cells
B cell
B & T
T cell
Myeloid
B,T & Myeloid

Table 3. Summary of Gene Set Enrichment Analysis For EDP1867 vs. vehicle

Small Intestine Cell Type	Biological process	Genes elevated for EDP1867 vs. vehicle (Fold change ≥1.5, BH-adjusted p≤0.05)
Cells of the immune system	Lymphocyte migration, gut-homing, and adhesion	*Ccl25, Ccr10, Ccl22, Ccl24, Itgb7, Itgal, Itgam*
	T cell lineage maturation and activation	*Cd69, Icos, Il2rg, Cd3d, Trbc1, Trbc2, Trac, Lat, Zap70, Lck, Cd2*
	B cell lineage maturation and activation	*CD19, CD79a, CD79b, CD69, Ighd, Fcrl1, Blk, Ikzf3, Tnfrsf13c, Jchain, Iglc1, Iglc2*
	Immune modulation	*Foxp3, Lag3, Traf3ip3, Slamf6, Il33, Cd5, Adamdec1, Nr1i3*
Cells of the intestinal epithelium	Intestinal epithelial cell homeostasis (barrier, metabolic, absorption)	*Gpx2, Gstm3, Aqp8, Guca2b, Adipoq, Dgat1, Dgat2, Slc23a1, Slc51b*
	Host-protective pathways	*Zg16, Def5a, Reg3a, Retnlb, Reg3g, Cfd, Lypd8, Casp6*

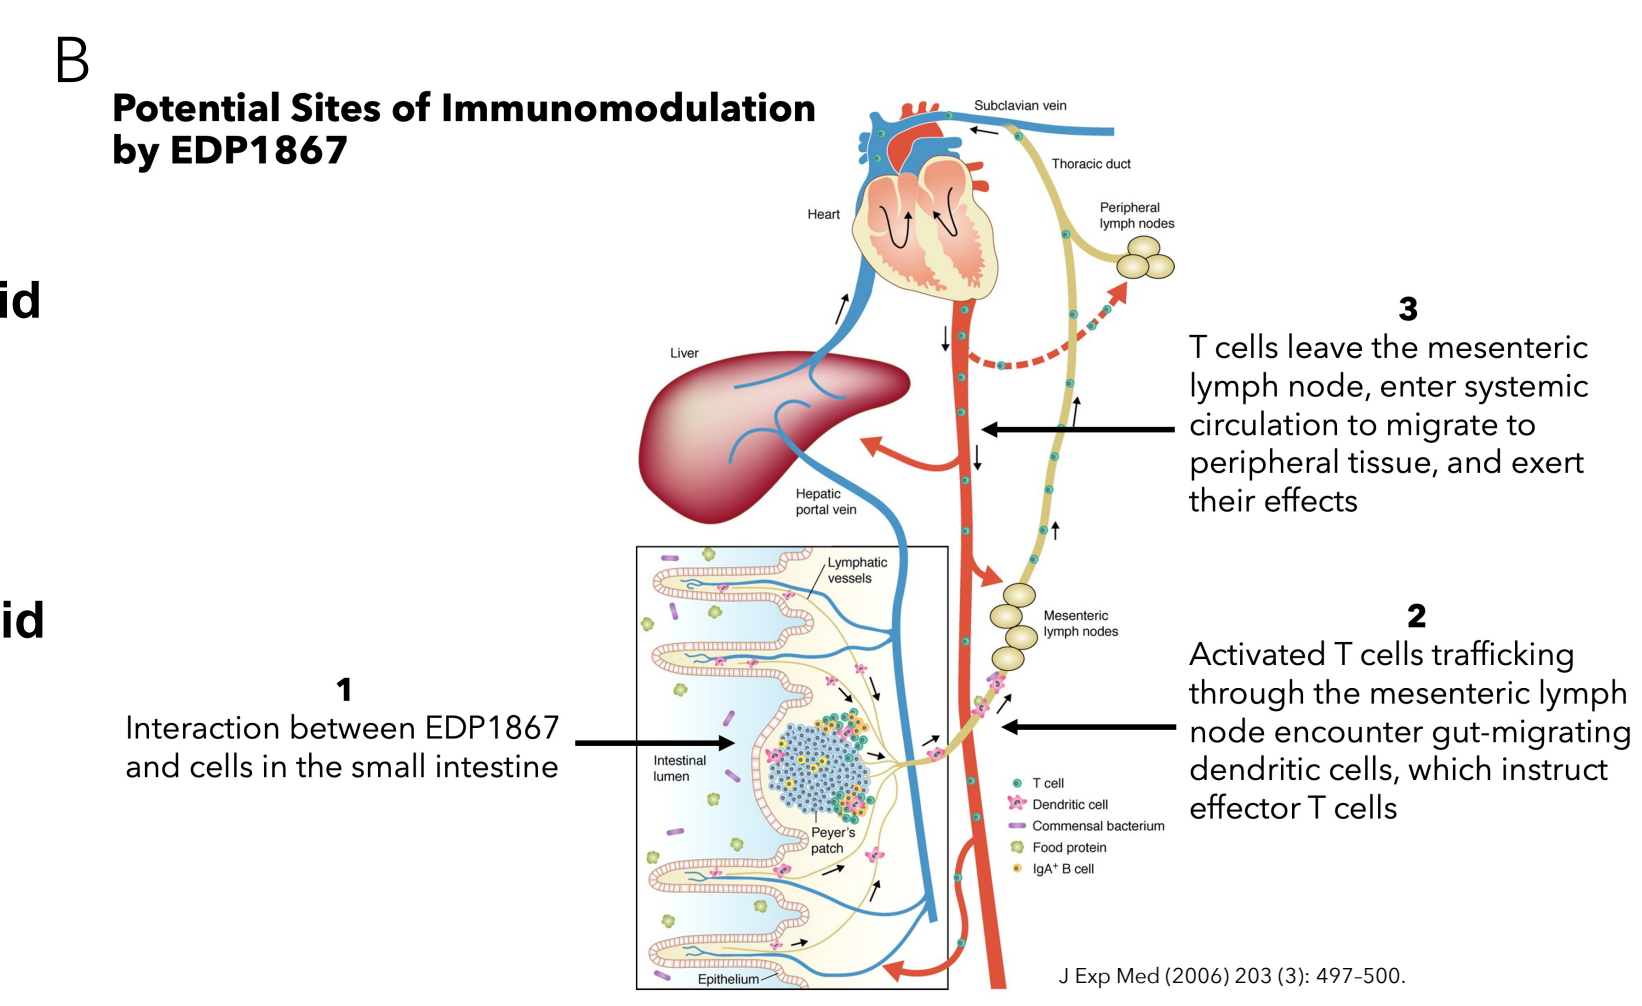
B
Potential Sites of Immunomodulation by EDP1867
3
T cells leave the mesenteric lymph node, enter systemic circulation to migrate to peripheral tissue, and exert their effects
2
Activated T cells trafficking through the mesenteric lymph node encounter gut-migrating dendritic cells, which instruct effector T cells
1
Interaction between EDP1867 and cells in the small intestine
J Exp Med (2006) 203 (3): 497-500.

Figure 4. Transcriptional profiling of duodenal tissue revealed that EDP1867 upregulated genes in lymphocyte pathways that resolve inflammation as well as genes associated with intestinal homeostasis
RNA from day 42 duodenal tissue was sequenced to a target depth of 20 million paired-end reads with Illumina 2x150 technology and processed using STAR and RSEM. Data normalization used trimmed mean of M-values (TMM), with generalized linear models for variance estimation using functions from empirical analysis of gene expression. P-values were adjusted to control false discovery rate (FDR) in multiple testing (Benjamini and Hochberg method). Differential gene expression was defined as a fold change of ≥1.5 and FDR-adj. p≤0.05. Enrichment and over-representation analyses were performed on datasets available through KEGG, gene ontology, EnrichR and MSigDB.
A) Cell types found enriched amongst genes showing elevated expression with EDP1867 vs. vehicle. IEC, intestinal epithelial cell. EDP1867 elevated genes with known function in immune cell migration to the gut, immune modulation, and intestinal homeostasis (**Table 3**).
B) Immunomodulation by EDP1867 occurs at various potential sites. 1. Orally-delivered EDP1867 interacts with innate immune cells in the small intestine. 2. Innate immune cells from the small intestine encounter activated T cells trafficking through the mesenteric lymph nodes and instruct effector T cell phenotype. 3. T cells enter systemic circulation and resolve inflammation in peripheral tissues.

Conclusions

We show that oral treatment with EDP1867 administered prophylactically or therapeutically reduces MMS and incidence of relapse in relapsing-remitting EAE. Spinal cord inflammation and demyelination were reduced in EDP1867-treated mice. Transcriptional profiling of small intestine tissue revealed that EDP1867 modulates immune cell responses through the gut, whilst reducing pathology in the CNS. These findings support EDP1867 development for the treatment of neuroinflammatory disease such as MS.

Our results indicate that SINTAX is an immune signaling portal to the CNS from the gut. This is a striking finding that suggests a new pathway of biological control of neuroinflammation and type of medicine for its treatment. The efficacy of this non-live biotherapeutic product is evidence that this is a result of a direct pharmacological interaction with host cells in the gut and is not the result of modification of the gut microbiota. The transcriptional profiling results are an insight into the mechanisms of this novel biology and pharmacology. EDP1867 is in Phase I clinical development for the treatment of atopic dermatitis.

Acknowledgements and Disclosure

Acknowledgements: EAE animal studies and histological analysis were performed at Hooke Laboratories, Inc. (Lawrence, MA) under contract with Evelo Biosciences.
Disclosures: H.E.P., A.N.R.C., A.A., W.C., A.N., M.S., V.K., K.H., K.R., D.E., E.U.K, M.J., T.G., M.B., and A.I are employees of Evelo Biosciences and as such receive salary and stock options. S.R. N.O., C.D., and M.A. have nothing to disclose.