#695 Oral delivery of a microbial extracellular vesicle induces potent anti-tumor immunity in mice

Loise Francisco, Mary Abdou, Alicia Ballok, Erin Troy, Fabian Romano Chernac, Maria Sizova, Audrey McBride, Michael Goldberg, Shubhra Kashyap, Shannon Argueta, Jessica Tsang, Kristie Barth, Krutika Invally, Holly Ponichtera, Mark Carlson, Tyler Rommel, Kevin Huynh, Valeria Kravitz, Tanmoy Ganguly, and Mark Bodmer

Evelo Biosciences, Cambridge MA

EVELO

INTRODUCTION

Evelo Biosciences is developing a new class of oral medicines which engage the immune system in the small intestine with distal effects throughout the body.

The microbial content of the gut interacts with host cells in the intestine to control systemic immune responses and inflammation. Our approach bypasses the need to modify the microbiota by selecting orally delivered agents which interact directly with host immune cells in the small intestine.

The small intestinal axis (SINTAX™) is a network of anatomic and functional connections with the rest of the body. It acts as an immunosurveillance system, integrating signals from the environment that affect physiological processes throughout the body. It can be harnessed for pharmacological effects of orally delivered agents that are systemically effective without systemic distribution. This is a newly appreciated and potent control system for immunity and inflammation.

This suggests a control mechanism for systemic immunity centered in the small intestine. This mechanism has novel features which are of considerable interest for the development of a new class of immunomodulatory therapies.

The impact of events in the gut on the control of tumor immunity is beginning to be appreciated. We have previously shown that an orally delivered single strain of commensal bacteria induces anti-tumor immunity preclinically via pattern recognition receptor-mediated activation of innate and adaptive immunity.

Some bacteria produce extracellular vesicles (EVs) that share molecular content with the parent bacterium in a particle that is roughly 1/1000th the volume in a non-replicating form.

We report here an orally-delivered and gut-restricted bacterial EV which potently attenuates tumor growth to a greater extent than whole bacteria or checkpoint inhibition.

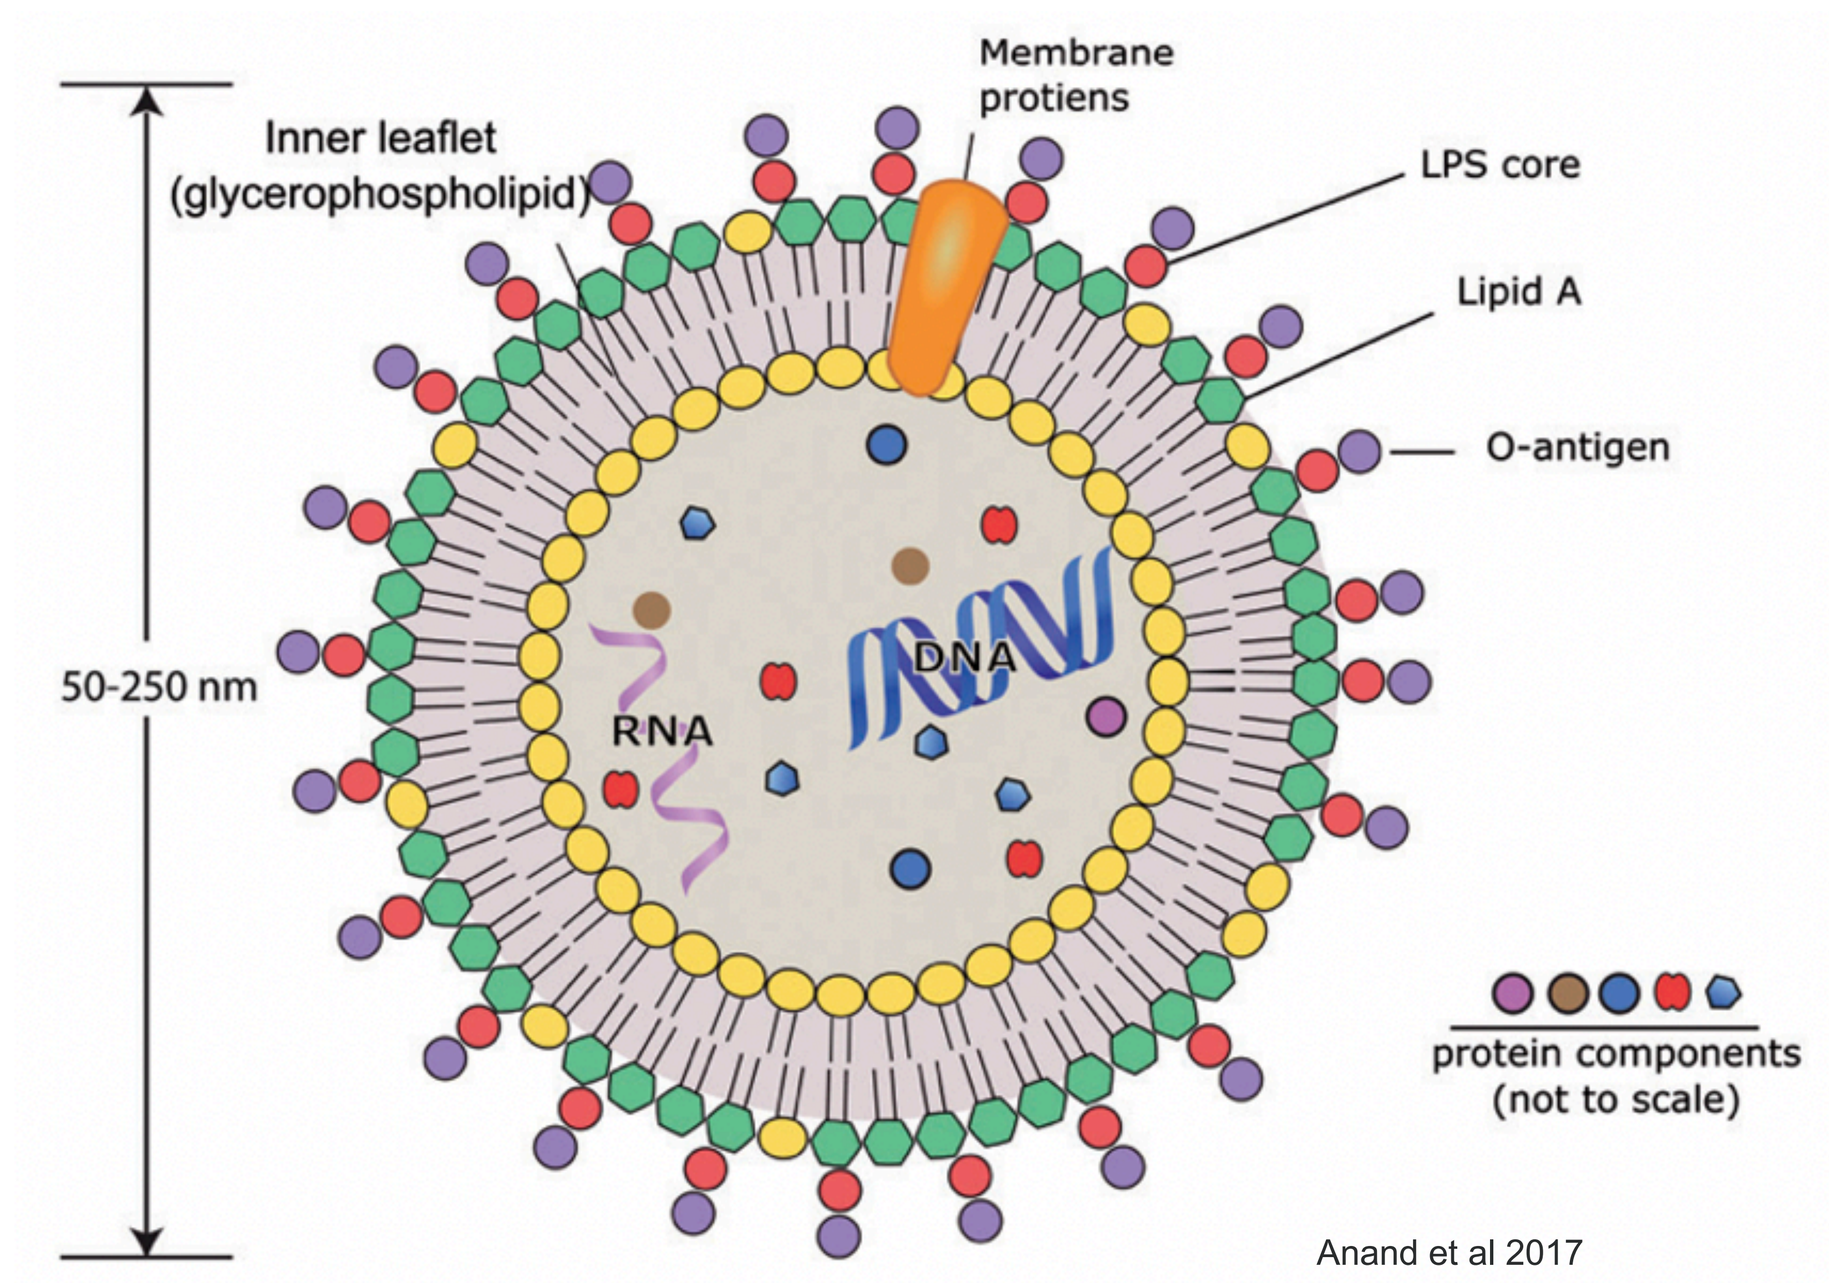
Inner leaflet
(glycerophospholipid)
Membrane
protiens
LPS core
Lipid A
O-antigen
50-250 nm
RNA
DNA
protein components
(not to scale)
Anand et al 2017

- Extracellular vesicles (EVs) are lipoprotein nanoparticles naturally produced by some species of bacteria.
- Their macromolecular content is a subset of the parent.
- EVs enable bacterial communication and survival during stress, host-immune modulation, material exchange, and cell-cell interactions.
- Compared to microbes, EVs are:
 - ~1/1000th volume of microbes enabling improved target engagement
 - Non-viable
 - No risk of infection and sepsis

EDP1908 is an extracellular vesicle derived from a strain of Oscillospiraceae

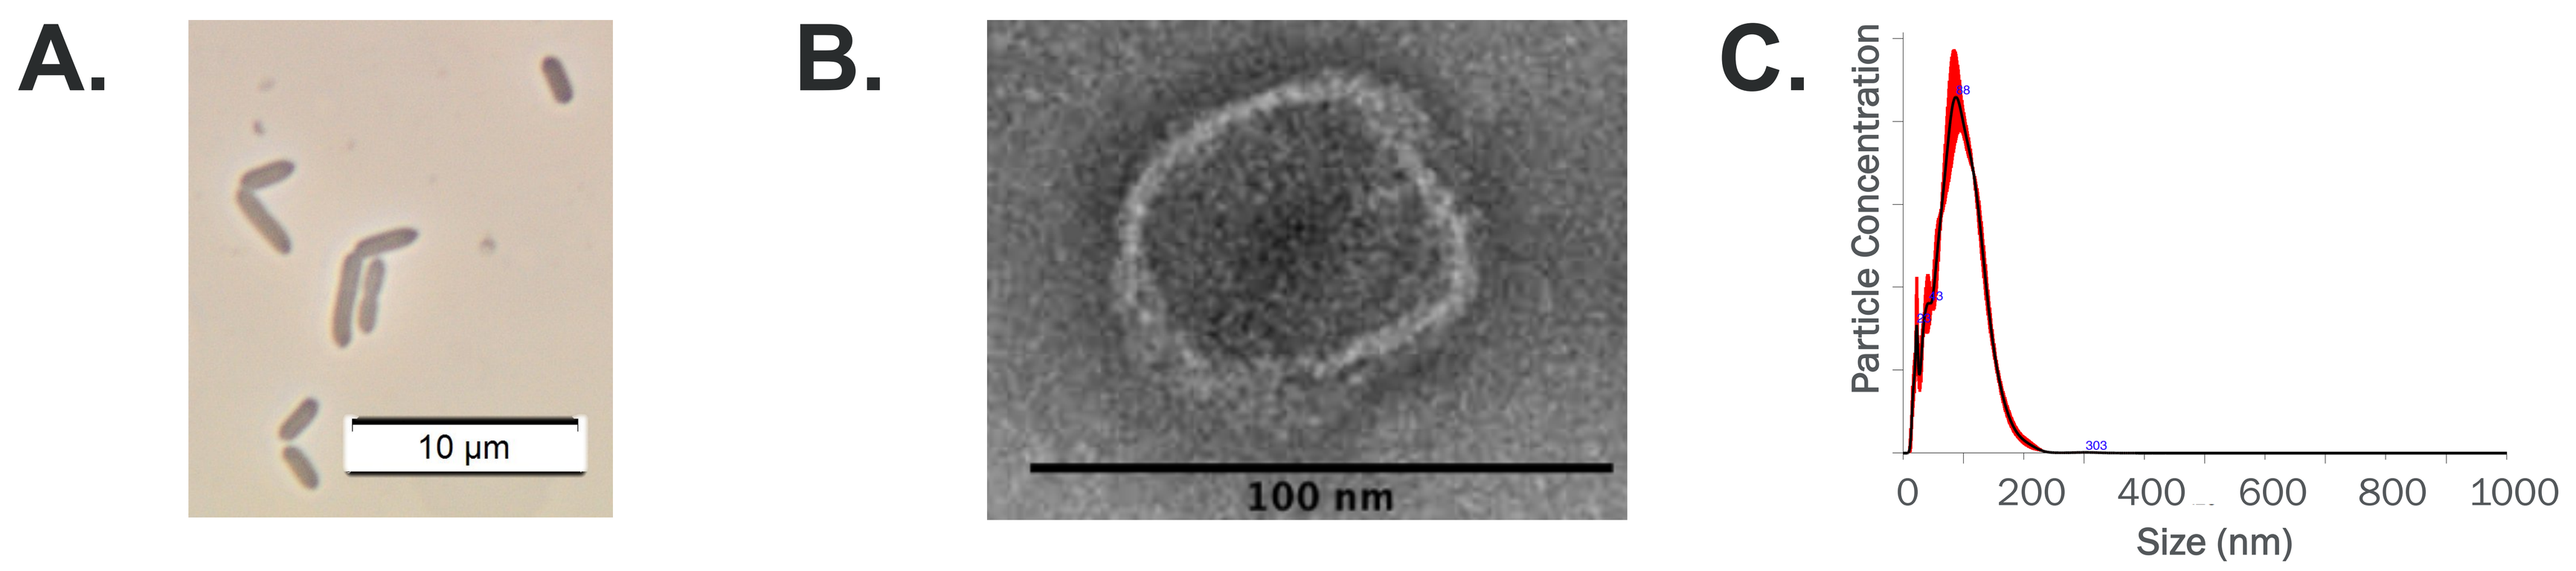
A.
10 µm
B.
100 nm
C.
Particle Concentration
0
200
400
600
800
1000
Size (nm)

Fig. 1. EDP1908 is an extracellular vesicle (EV) produced by a single bacterial species.

(A) Phase contrast image of the Oscillospiraceae species from which EDP1908 is derived.
(B) TEM image of EDP1908 isolated from the supernatant of a liquid culture of the parent microbe.
(C) EDP1908 comprises a population of EVs with an average diameter of 88nm determined by nanoparticle tracking analysis.

Orally administered EDP1908 has superior anti-tumor efficacy to anti-PD-1 or its parent microbe

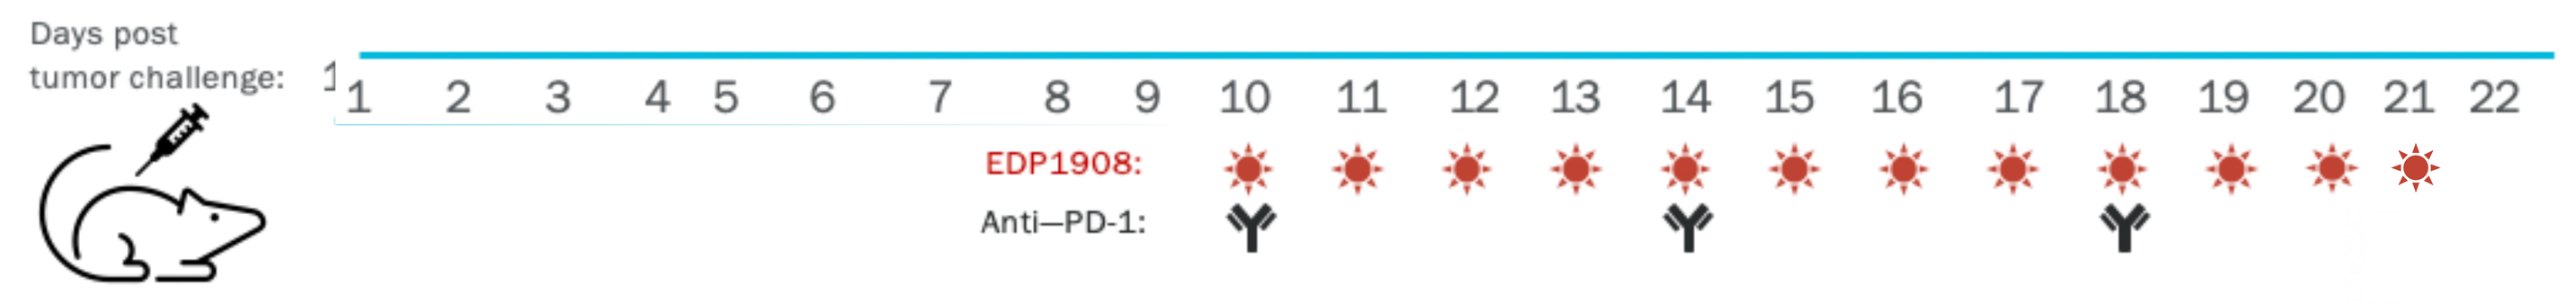
Days post
tumor challenge:
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22
EDP1908:
Anti–PD-1:

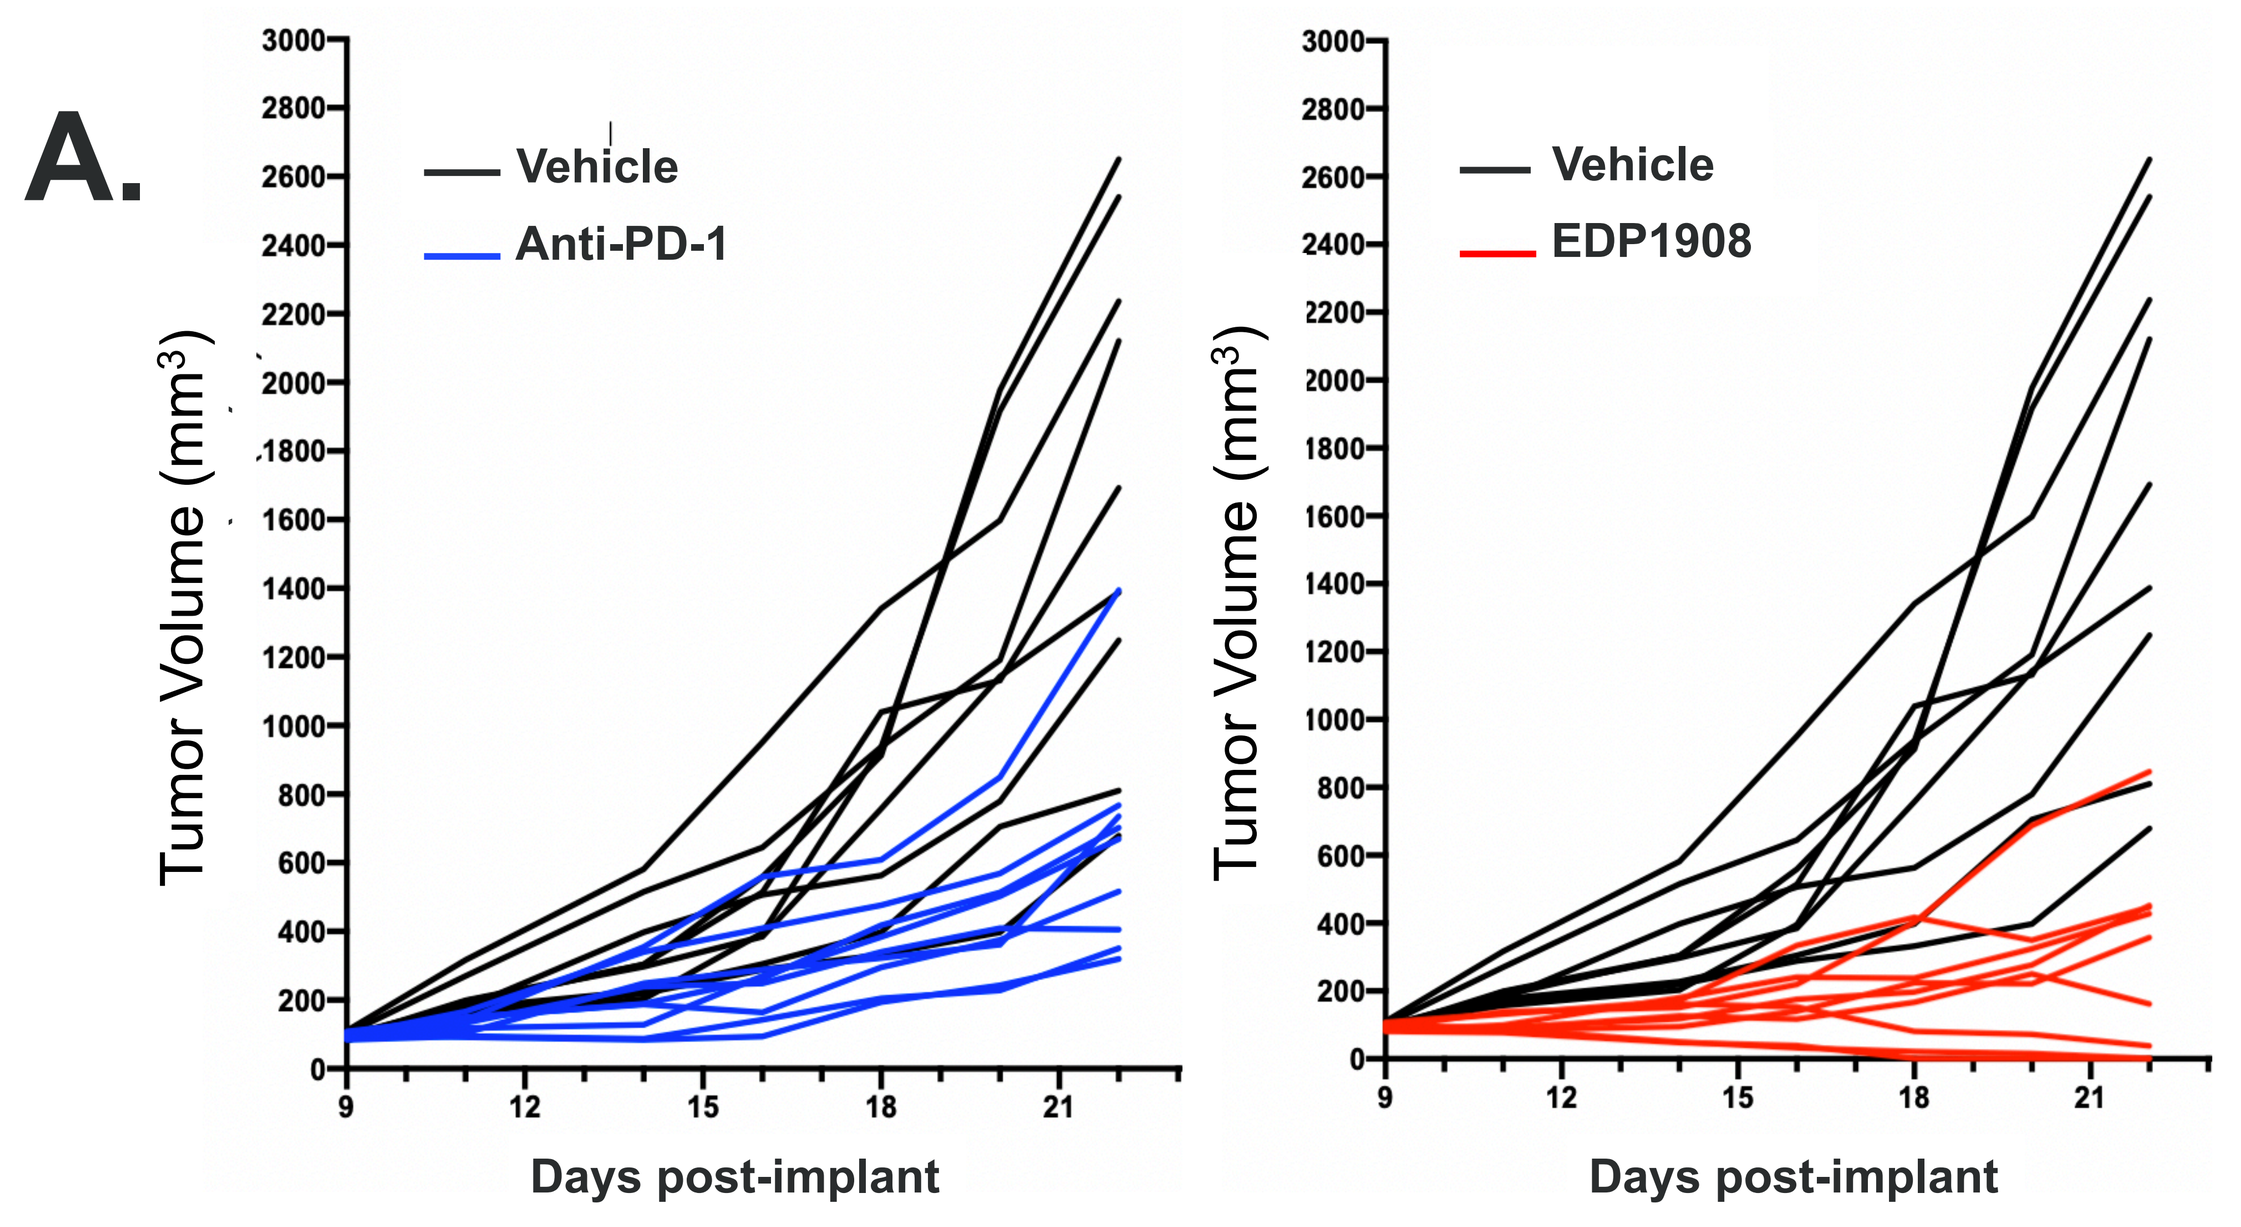
A.
Vehicle
Anti-PD-1
Tumor Volume (mm³)
Days post-implant
Vehicle
EDP1908
Tumor Volume (mm³)
Days post-implant

Fig 2. Therapeutic treatment of CT26 tumor-bearing mice with either oral EDP1908 b.i.d., anti-PD-1 i.p., or whole microbe b.i.d.

A) EDP1908 was administered orally at 2e11 particles beginning at day 10 for 12 days vs anti-PD-1. Spider plots show tumor growth for individual mice.
B) Dose-dependent control of tumor growth by EDP1908 doses from 2e5 – 2e11 particles shown at day 21. Data are median and range.
C) The parental Oscillospiraceae strain of EDP1908 was administered at its highest deliverable concentration of 2e9 cells. The 2e11 and 2e9 doses of EVs are shown for reference. Data are median and range.

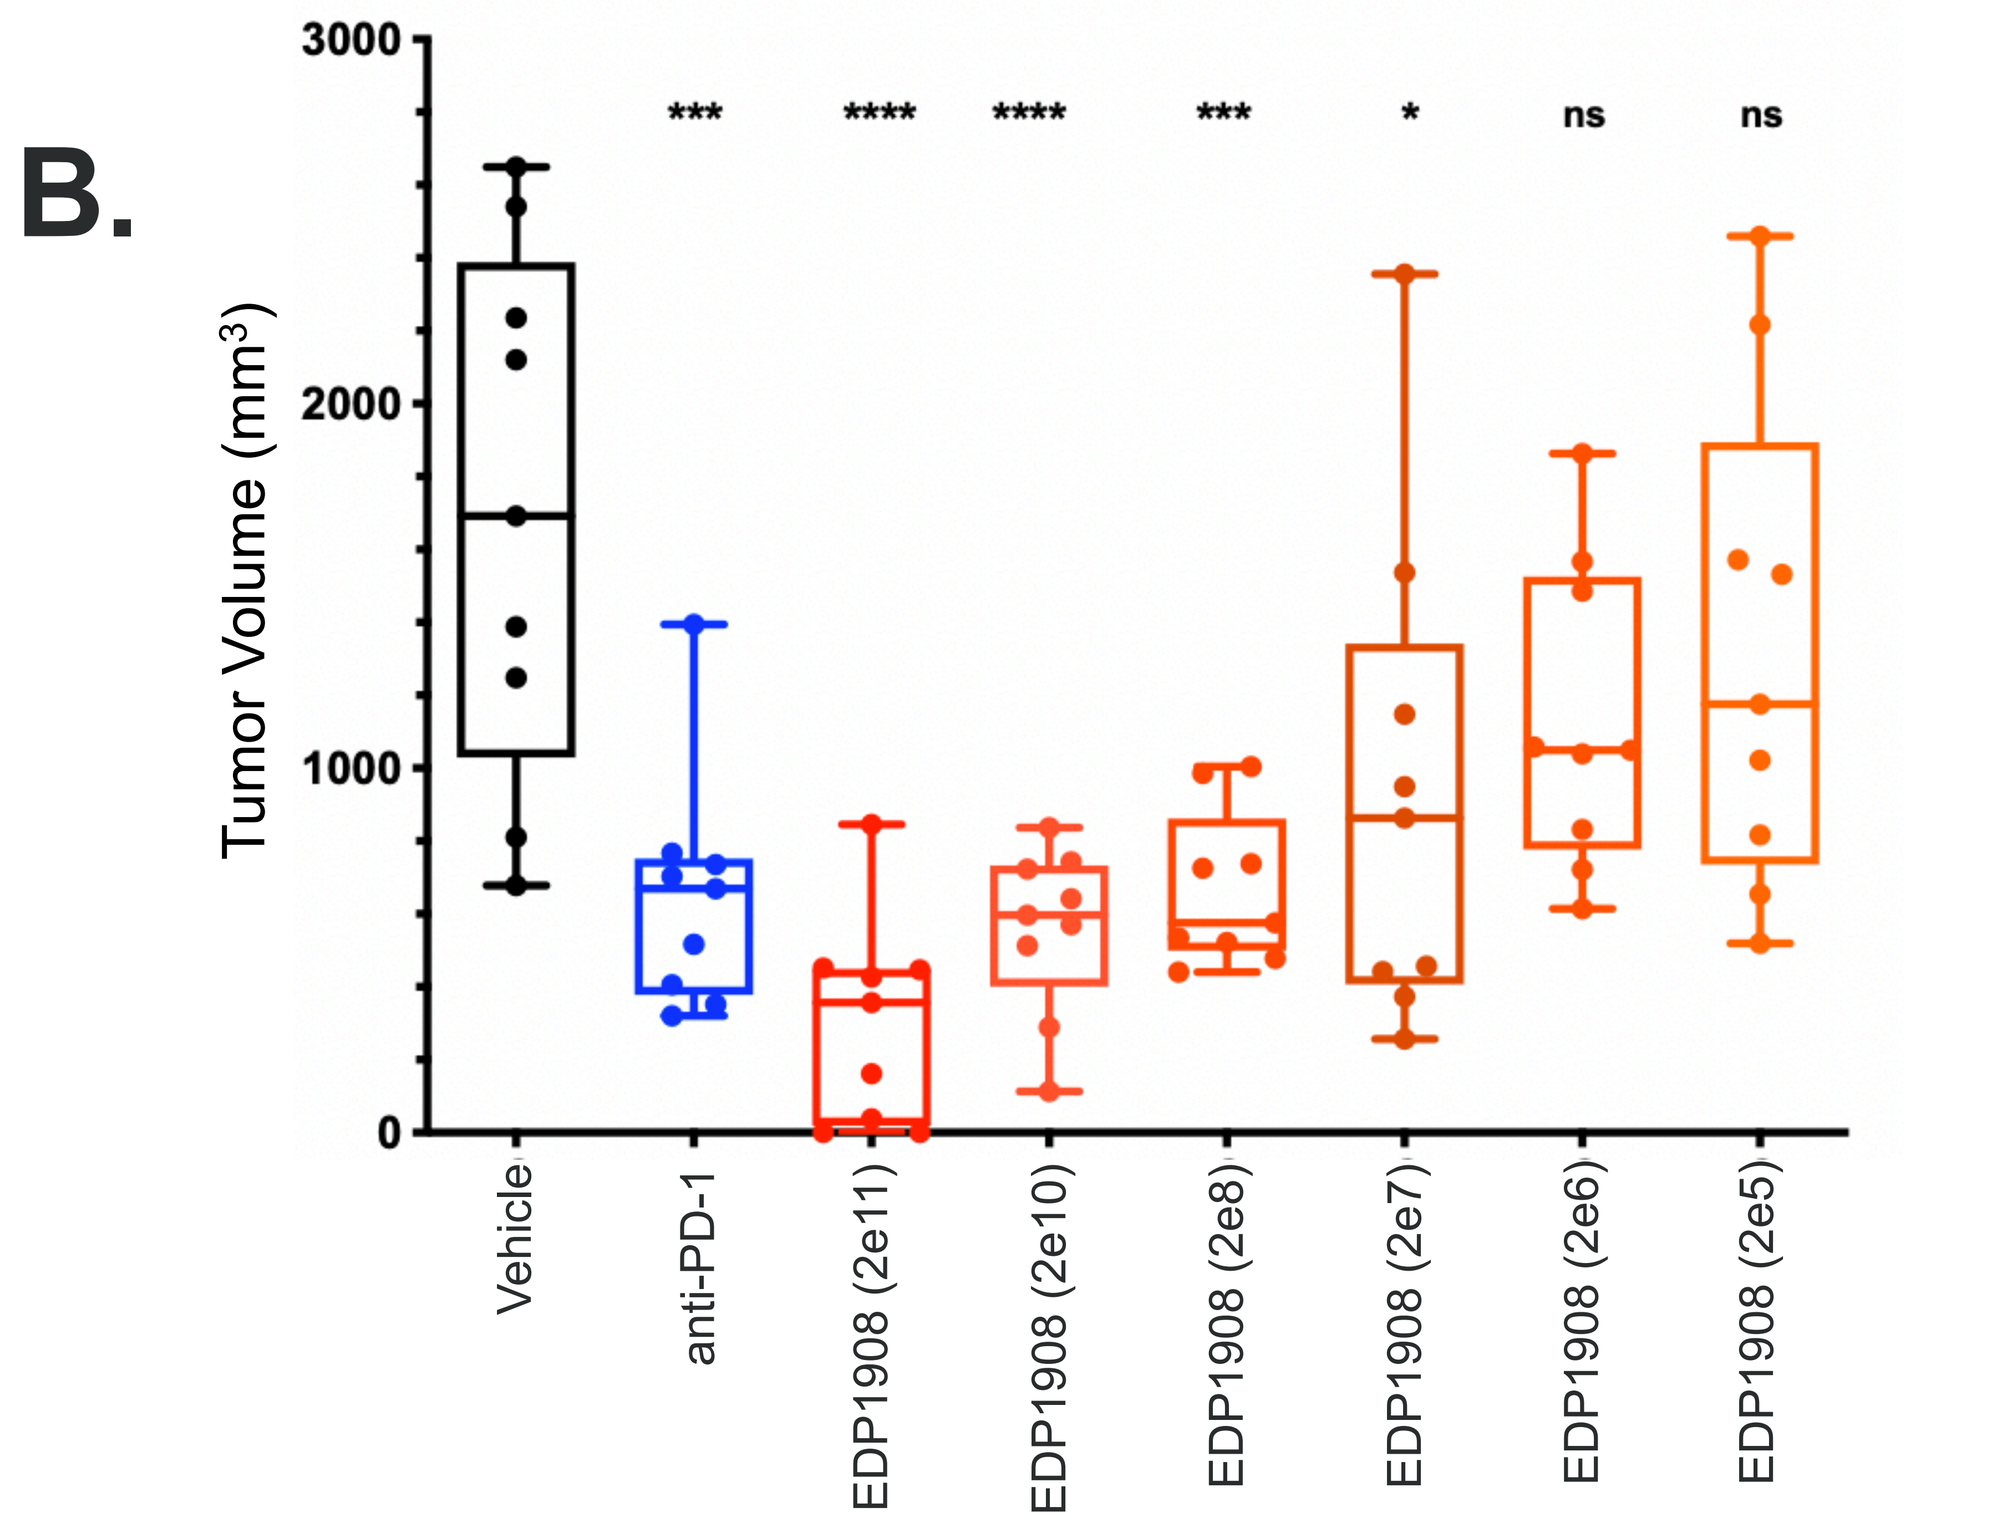
B.
Tumor Volume (mm³)
*** **** **** *** * ns ns
Vehicle
anti-PD-1
EDP1908 (2e11)
EDP1908 (2e10)
EDP1908 (2e8)
EDP1908 (2e7)
EDP1908 (2e6)
EDP1908 (2e5)

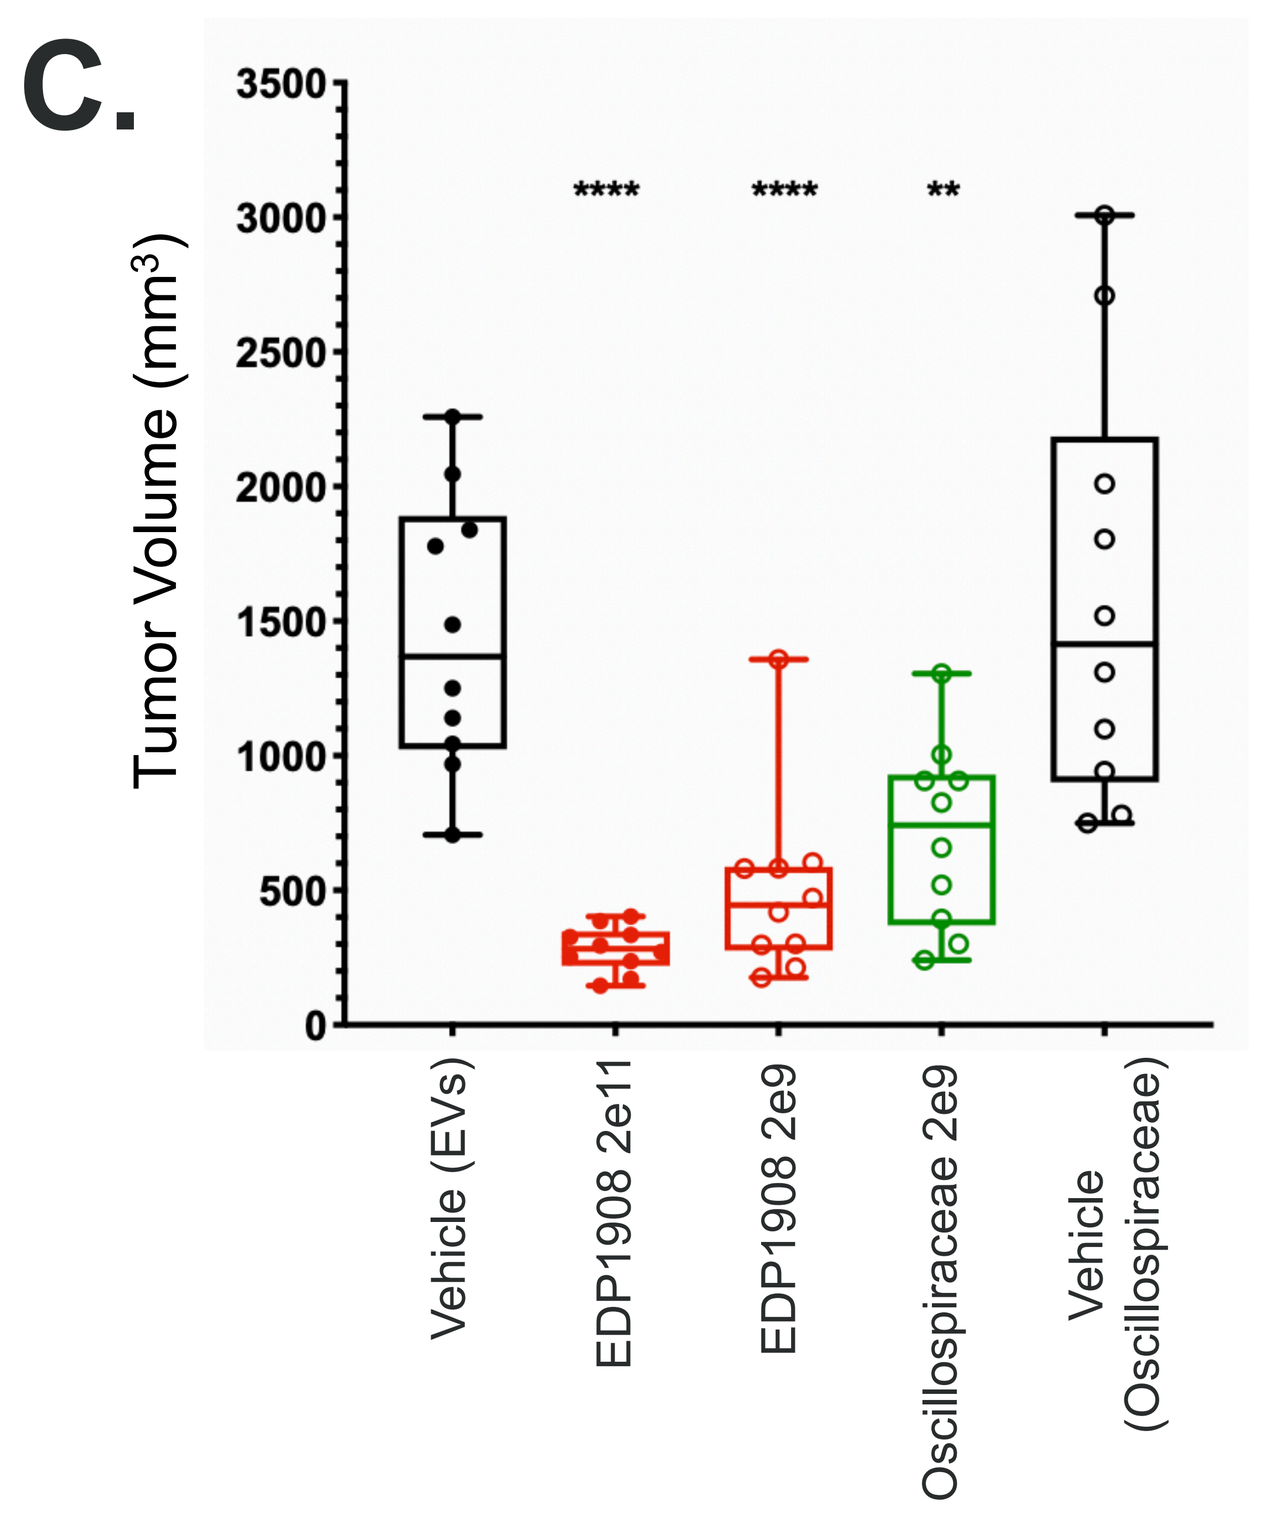
C.
Tumor Volume (mm³)
**** **** **
Vehicle (EVs)
EDP1908 2e11
EDP1908 2e9
Oscillospiraceae 2e9
Vehicle
(Oscillospiraceae)

Oral treatment with EDP1908 distally activates effector lymphocyte populations in the TME

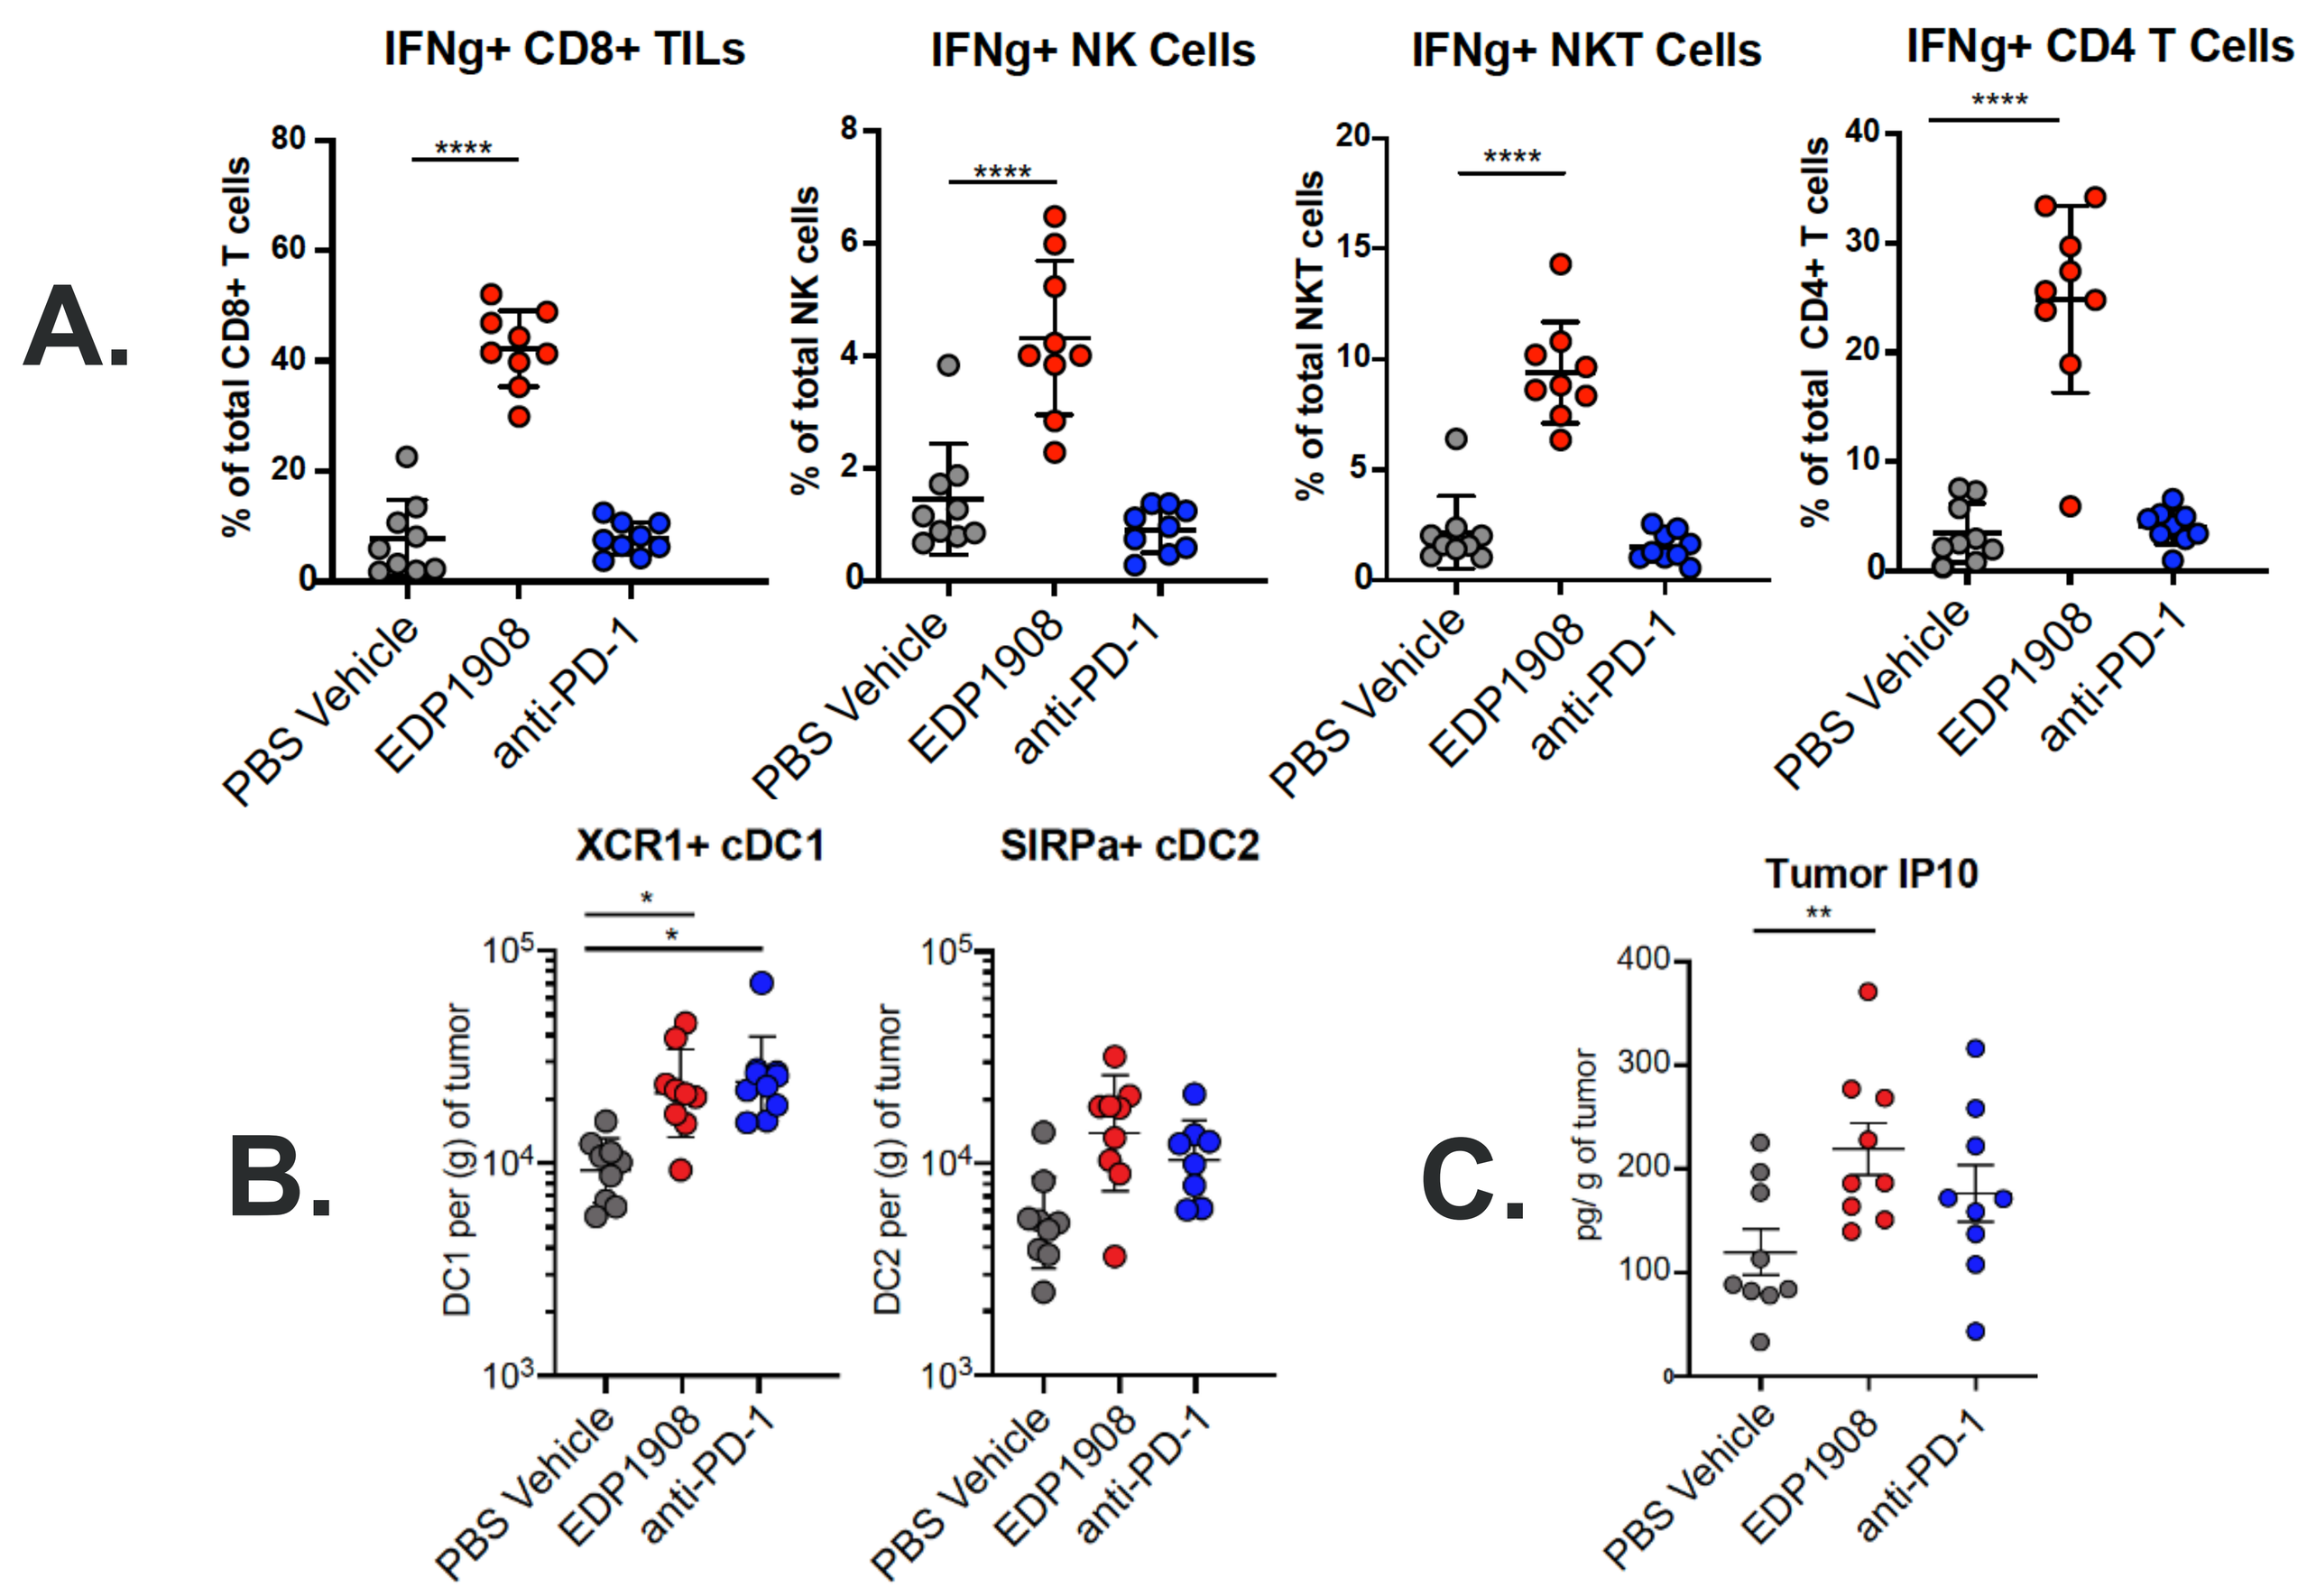
A.
IFNg+ CD8+ TILs
IFNg+ NK Cells
IFNg+ NKT Cells
IFNg+ CD4 T Cells
% of total CD8+ T cells
% of total NK cells
% of total NKT cells
% of total CD4+ T cells
PBS Vehicle
EDP1908
anti-PD-1
B.
XCR1+ cDC1
SIRPa+ cDC2
DC1 per (g) of tumor
DC2 per (g) of tumor
C.
Tumor IP10
pg/ g of tumor

Fig 3. EDP1908 activates and sustains IFNγ+ cytolytic and helper lymphocytes, DCs and IP-10 in the TME.

At study termination on day 21, tumors from mice treated with PBS, EDP1908 or anti-PD-1 were digested.

Flow cytometry was used to assess:
A) IFNγ production from CD8, NK, NKT and CD4 T cells.
B) Infiltrating DC subsets.

MSD assay was used to detect:
C) IP-10 production within the TME.

EDP1908 is not detected outside the GI tract by fluorescent biodistribution

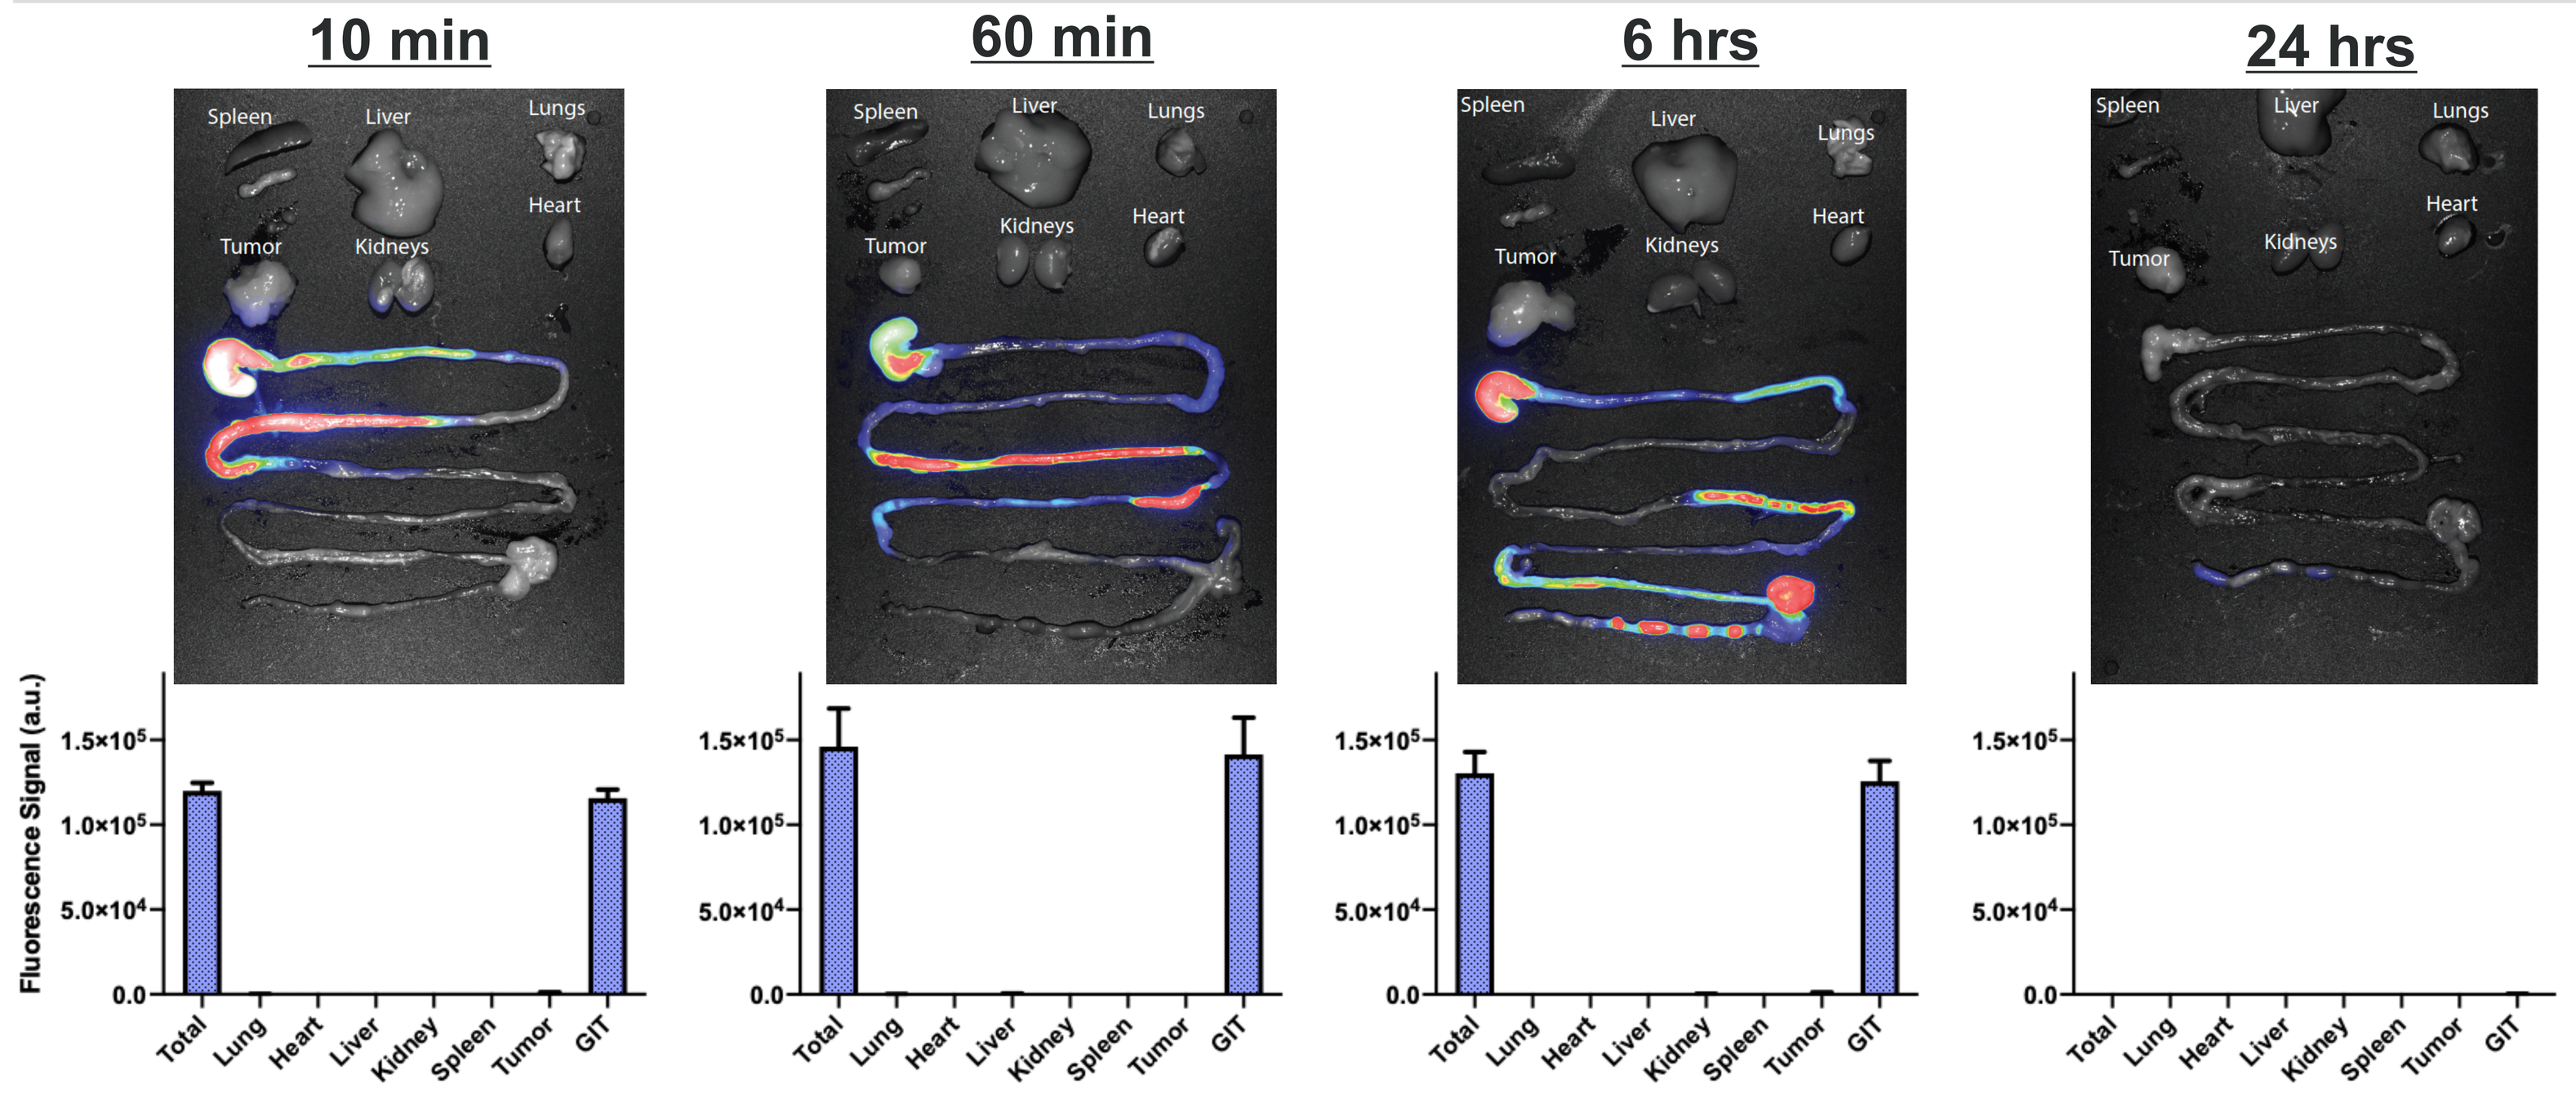
10 min
60 min
6 hrs
24 hrs
Spleen
Liver
Lungs
Heart
Tumor
Kidneys
Fluorescence Signal (a.u.)
Total
Lung
Heart
Liver
Kidney
Spleen
Tumor
GIT

Fig 4. EDP1908 is restricted to the gastrointestinal tract and not detected in systemic organs.

Mice were orally dosed with 2e11 EDP1908 EVs covalently labeled with IRDye800. Organ fluorescence was quantified using a small animal imaging system (Licor Pearl®).

CONCLUSIONS

This is the first report of striking anti-tumor effects of an orally delivered microbial extracellular vesicle. The magnitude of these effects in preclinical models appears to match the reported activity of injected intra-tumoral immunostimulators. The observation that this efficacy can be achieved with an oral agent which does not itself distribute to the tumor is evidence for the level of control that can be exerted via the small intestinal axis, with no apparent safety or tolerability issues in the animal models.

These data point to oral EVs as a new class of immunotherapeutic drugs. They are particularly effective at engaging the small intestinal axis, acting locally on host cells in the gut to activate distal immune responses within the tumor microenvironment. EDP1908 is in preclinical development for the treatment of cancer.