San Antonio Breast Cancer Symposium® - December 8-11, 2020

PS11-27

A phase I/II clinical trial of EDP1503 with pembrolizumab for triple-negative breast cancer

Loise Francisco-Anderson PhD[1*], Shamira Shariffudin[1*], Humphrey Gardner MD[1], Michael Goldberg PhD[1], Shubhra Kashyap MS[1], Mary Abdou[1], Chris Davitt PhD[1], Shannon Argueta PhD[1], Pooja Parameswaran MS[1], Peter Sandy PhD[1], Holly Ponichtera PhD[1], Mark Carlson[1], Maria Sizova PhD[1], Valeria Kravitz[1], Erin Troy PhD[1], Sam Andrewes MS[1], Michael Chisamore PhD[2], Mark Bodmer PhD[1], and Duncan McHale MD[1]

[1]*Evelo Biosciences, Cambridge MA; *contributed equally to this work; [2]Oncology Clinical Research, Merck & Co., Inc, Kenilworth, NJ, USA*

INTRODUCTION

ABSTRACT:
The small intestinal axis (SINTAX™) is a network of anatomical and functional connections with the rest of the body. Systemic immunity is regulated by the small intestine via pharmacological non-colonizing interactions of bacteria and immune cells. The potential to harness these interactions for IO treatment was first described by Sivan et al in 2015. EDP1503 is a non-colonizing preparation of a single strain of *Bifidobacterium animalis lactis.* In preclinical models, orally-administered EDP1503 invokes an anti-tumor immune response by activation of innate and adaptive immune mechanisms. The pleiotropic effects of EDP1503 are initiated in the small intestine and driven by interactions with multiple pattern-recognition receptors inducing a proinflammatory signature in human PBMCs, activating CD8 and NK cell IFNγ production and cytolytic activity. Preclinically, EDP1503 inhibits tumor growth as a monotherapy and in combination with anti-PD-1/L1. Single agent anti-PD-1/L1 therapies have shown limited clinical benefit in previously treated triple-negative breast cancer (TNBC) patients. Here, we report initial data on the safety, tolerability, and efficacy of EDP1503 in combination with pembrolizumab in patients with TNBC (NCT03775850).

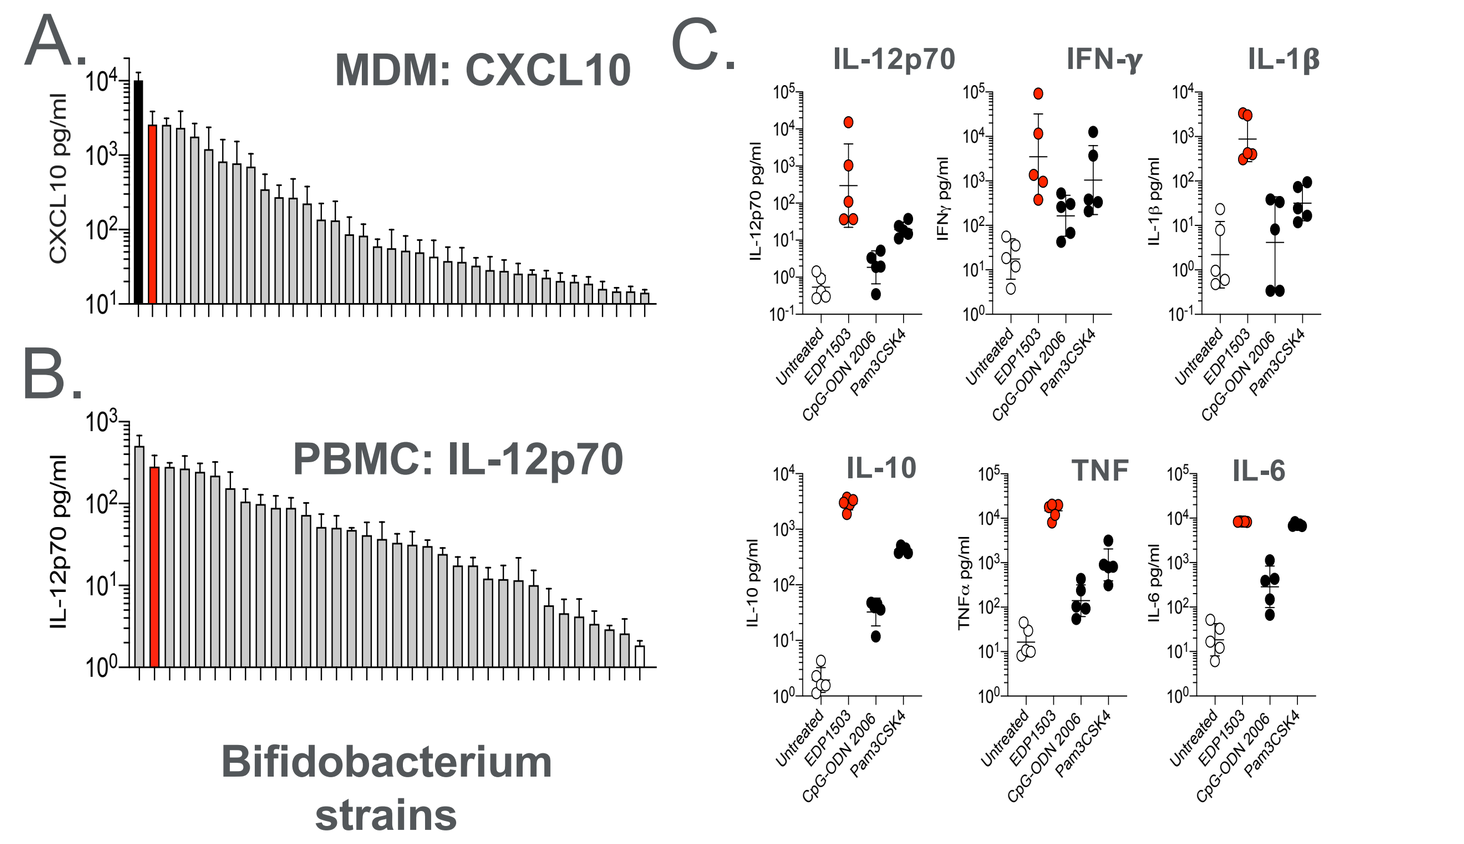
A.
MDM: CXCL10
CXCL10 pg/ml
B.
PBMC: IL-12p70
IL-12p70 pg/ml
Bifidobacterium strains
C.
IL-12p70
IFN-γ
IL-1β
IL-10
TNF
IL-6

Fig 1. EDP1503 Stimulates Type I Immune Responses
(A) Human monocyte-derived macrophages stimulated with EDP1503 (shown in red) *in vitro* lead to the strong induction of CXCL10 compared to other strains of *Bifidobacterium* (M1 polarized macrophages shown in black). **(B)** Similarly, human PBMCs stimulated with EDP1503 induced significant IL-12p70 in comparison to other *Bifidobacterium.* **(C)** EDP1503 stimulates the PBMC production of a pro-inflammatory cytokine signature significantly greater than individual TLR2 or TLR9 agonists (N = 5 donors).

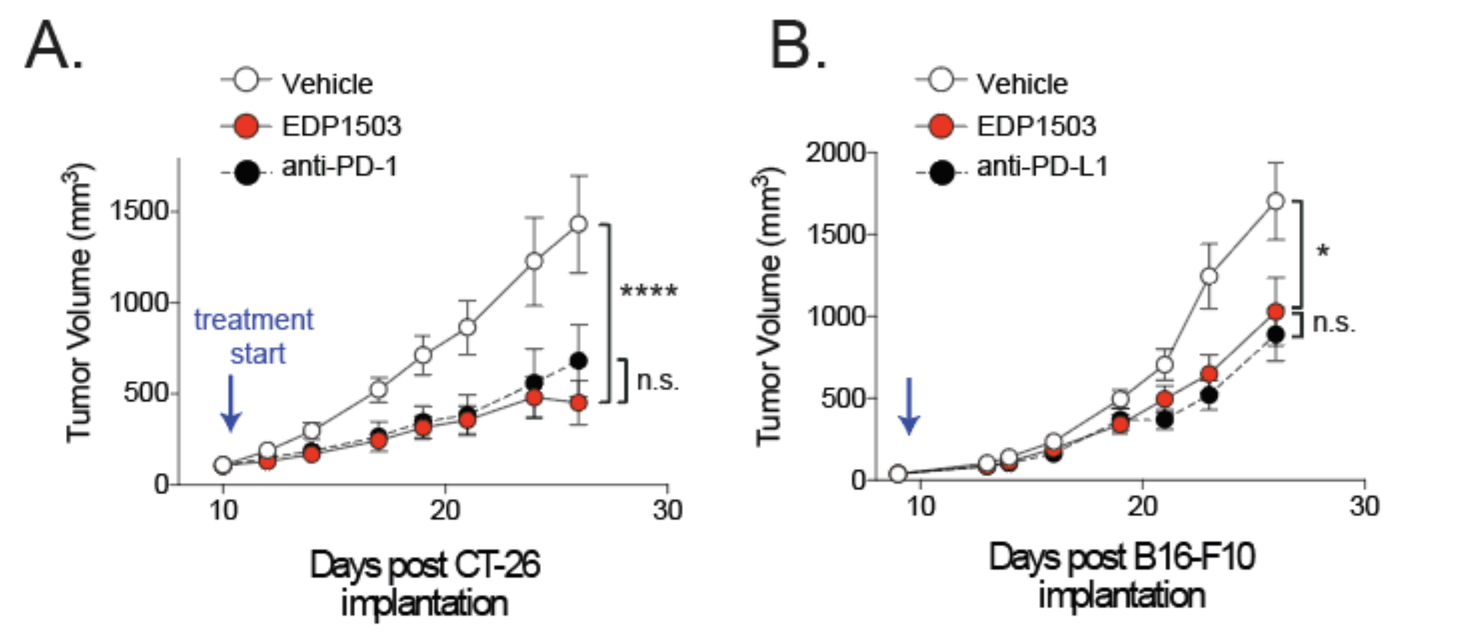
A.
Vehicle
EDP1503
anti-PD-1
Tumor Volume (mm³)
treatment start

n.s.
Days post CT-26 implantation
B.
Vehicle
EDP1503
anti-PD-L1
*
n.s.
Days post B16-F10 implantation

Fig 2. Oral delivery of EDP1503 is as effective as anti-PD-1 in multiple tumor models *in vivo*
Oral delivery of EDP1503 monotherapy leads to tumor control comparable to checkpoint inhibition in multiple subcutaneous syngeneic tumor models. **(A)** Syngeneic mice were inoculated with CT26 or **(B)** B16-F10 tumor cells at 10^5 subcutaneously. Tumors were established for 10 days (~100mm³) prior to initiation of treatment (indicated by blue arrow). Mice were treated with either sucrose vehicle or EDP1503 via oral gavage daily or treated with anti-PD-1/ anti-PD-L1 every four days for the duration of the study.

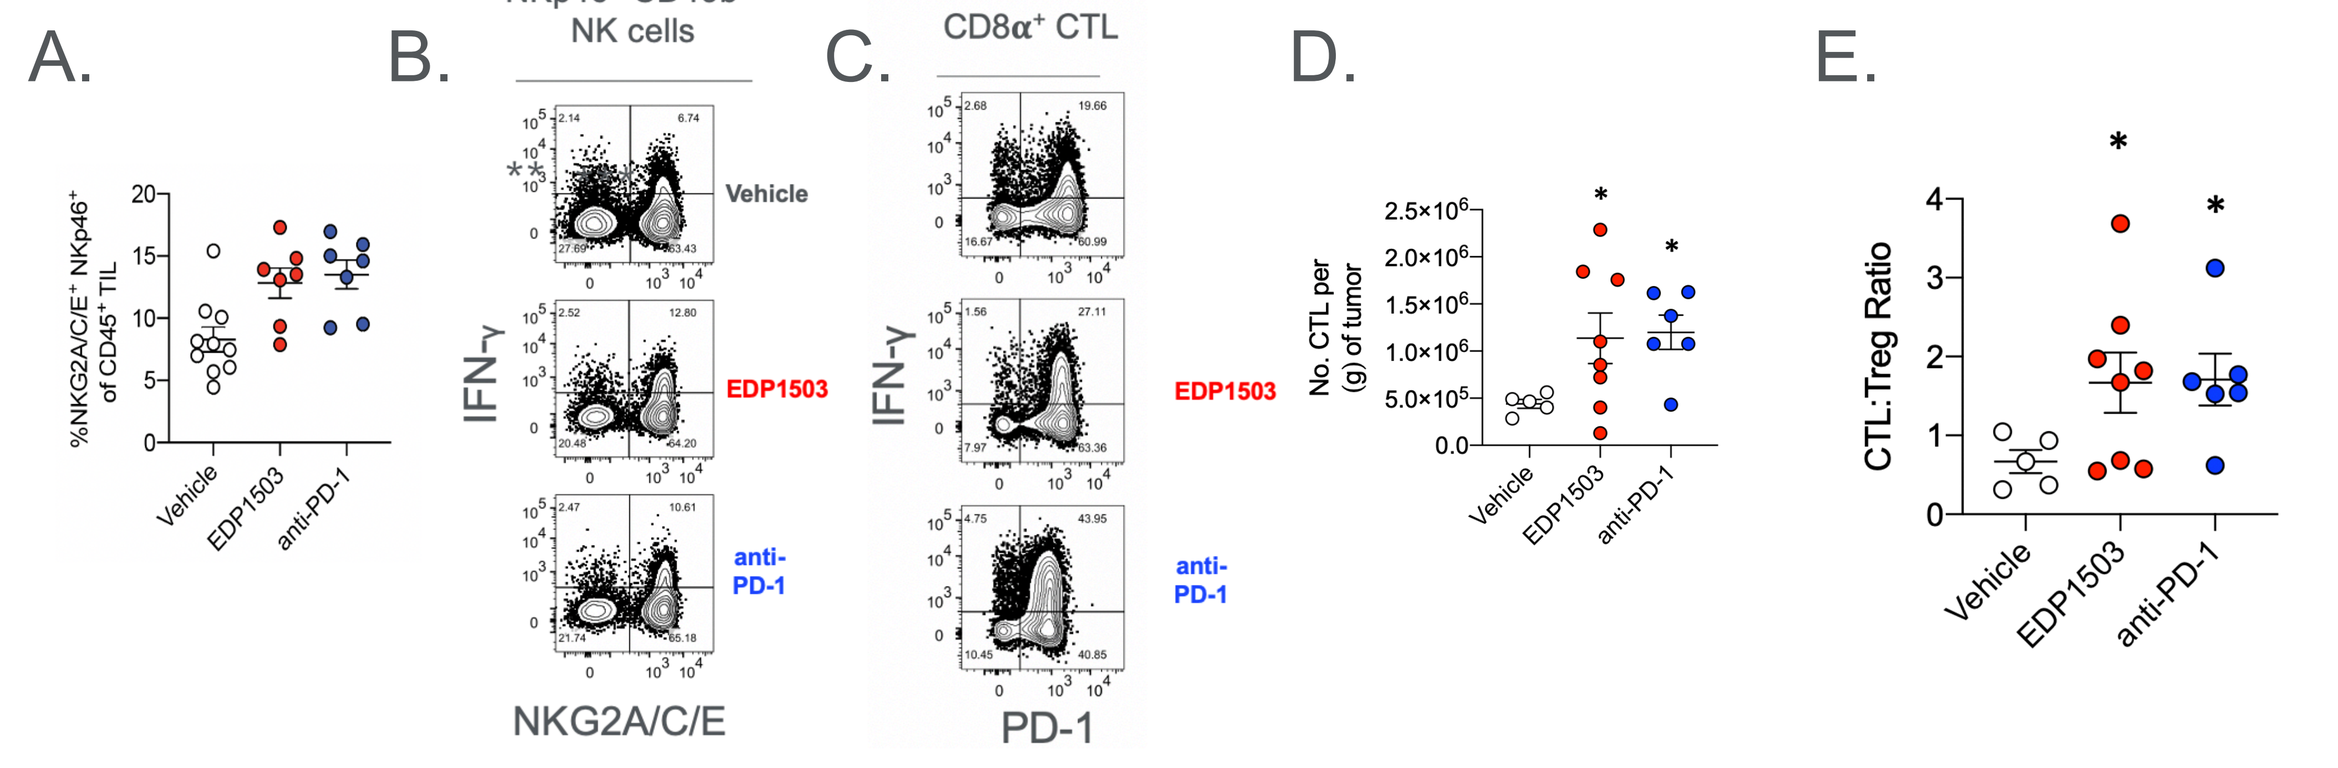
A.
B.
NKp46⁺ CD49b⁺ NK cells
C.
CD44^Hi CD3e⁺ CD8α⁺ CTL
D.
E.
Vehicle
EDP1503
anti-PD-1
IFN-γ
NKG2A/C/E
PD-1
CTL:Treg Ratio

Fig 3. EDP1503 activates effector NK and CD8 TILs **(A)** EDP1503 treatment of CT26 bearing mice increased NKp46⁺NKG2A/C/E⁺ NK cells within the TME comparable to anti-PD-1 treatment. **(B)** EDP1503 induces IFNγ production by NKp46⁺NKG2A/C/E⁺ NK cells. **(C)** EDP1503 induces elevated IFNγ production and concomitant upregulation of PD-1 in CD44⁺CD8 CTL within the TME. **(D)** EDP1503 treatment results in increased tumor-infiltrating CTLs in responding tumors, comparable to anti-PD-1; reported as counts of CTL normalized per gram of tumor. **(E)** EDP1503 increases the CD8 CTL to Treg ratio within the TME of responding tumors, comparable to anti-PD-1. Responding tumors were identified as below the median tumor weight for each respective group. Tumors were analyzed following 5 days of EDP1503 treatment by flow cytometry and statistics were obtained using Brown-Forsythe and Welch ANOVA and Holm-Sidak post-hoc analysis.

OBJECTIVES: This study is being conducted to assess the safety, tolerability, and efficacy of EDP1503 in combination with pembrolizumab in patients with advanced metastatic triple-negative breast cancer, and other tumor types#.

#Initial data from all cohorts was previously reported at the World Congress on Gastrointestinal Cancer 2020. Data reported here is an update of the TNBC cohort.

METHODS

CLINICAL TRIAL: Open-label Study of EDP1503 in Combination With Pembrolizumab in Patients With Advanced Metastatic Colorectal Carcinoma, Triple-negative Breast Cancer, and Checkpoint Inhibitor Relapsed Tumors (NCT03775850)

Primary Outcome Measures:
1. Safety and tolerability of EDP1503 in combination with pembrolizumab as assessed per CTCAE v5.0. Number of participants with EDP1503 related adverse events as assessed per CTCAE v5.0
2. Safety and tolerability of EDP1503 alone and in combination with pembrolizumab assessed via clinical laboratory evaluations
3. Evidence of anti-tumor activity of EDP1503 based on ORR per RECIST 1.1 and iRECIST

Endpoints
- Safety and tolerability
- Overall response rates
- Biomarkers of immune response in paired tumor biopsies taken before and after 2-week EDP1503 monotherapy

Dosing
- Patients receive a 14-day run-in of EDP1503 monotherapy, followed by combination of EDP1503 and pembrolizumab
- EDP1503 is given as 2-4 capsules taken by mouth twice daily; pembrolizumab (200 mg) is administered by intravenous (IV) infusion once every 3 weeks

Triple-negative Breast Cancer (N=15)

15 patients across all EDP1503 doses, recruited as of 7 Oct 2020
- Low dose: 2 capsules BID (n=3)
- High dose: 4 capsules BID (n=12)

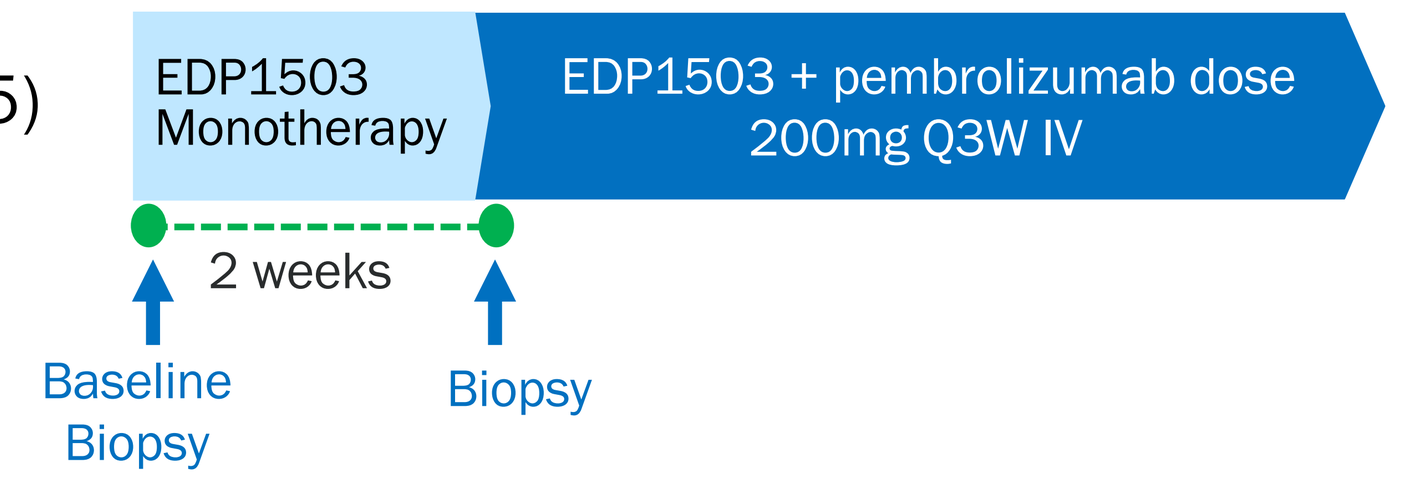
EDP1503 Monotherapy
EDP1503 + pembrolizumab dose 200mg Q3W IV
2 weeks
Baseline Biopsy
Biopsy

RESULTS

Table 1: Baseline Characteristics

Characteristics	n (%)
Gender	
Female	15 (100)
Median age	52
≥ 65 years	4 (27)

Characteristics	n (%)
ECOG	
0	8 (53)
1	7 (47)
Prior anti-PD-(L)1 therapy	4 (27)

Table 2: Treatment-related Adverse Events (TRAE)

Adverse event, n (%)	Related to study treatment		
N=15	All Gr.	Gr. 1-2	Gr. 3
≥ 1 any TRAE	8 (53)	8 (53)	1 (7)
TRAE led to discontinuation	1 (7)	0	1 (7)
TRAE led to death	0	0	0
Abdominal distension	3 (20)	3 (20)	0
Decreased appetite	3 (20)	3 (20)	0
Diarrhea	2 (13)	2 (13)	1 (7)
Flatulence	2 (13)	2 (13)	0
Nausea	2 (13)	2 (13)	0
Pruritus	2 (13)	2 (13)	0
Rash maculo-papular	2 (13)	2 (13)	0
Abdominal pain	1 (7)	1 (7)	0

Adverse event, n (%)	Related to study treatment		
	All Gr.	Gr. 1-2	Gr. 3
Constipation	1 (7)	1 (7)	0
Dizziness	1 (7)	1 (7)	0
Eructation	1 (7)	1 (7)	0
Fatigue	1 (7)	1 (7)	0
Gastroesophageal reflux disease	1 (7)	1 (7)	0
Headache	1 (7)	1 (7)	0
Hypothyroidism	1 (7)	1 (7)	0
Myalgia	1 (7)	1 (7)	0
Skin hyperpigmentation	1 (7)	1 (7)	0
Vomiting	1 (7)	1 (7)	0
Weight decreased	1 (7)	1 (7)	0

Note: No treatment-related Grade 4-5 AEs or treatment-related serious AEs have been reported.

Fig 4A. Duration on Study Treatment

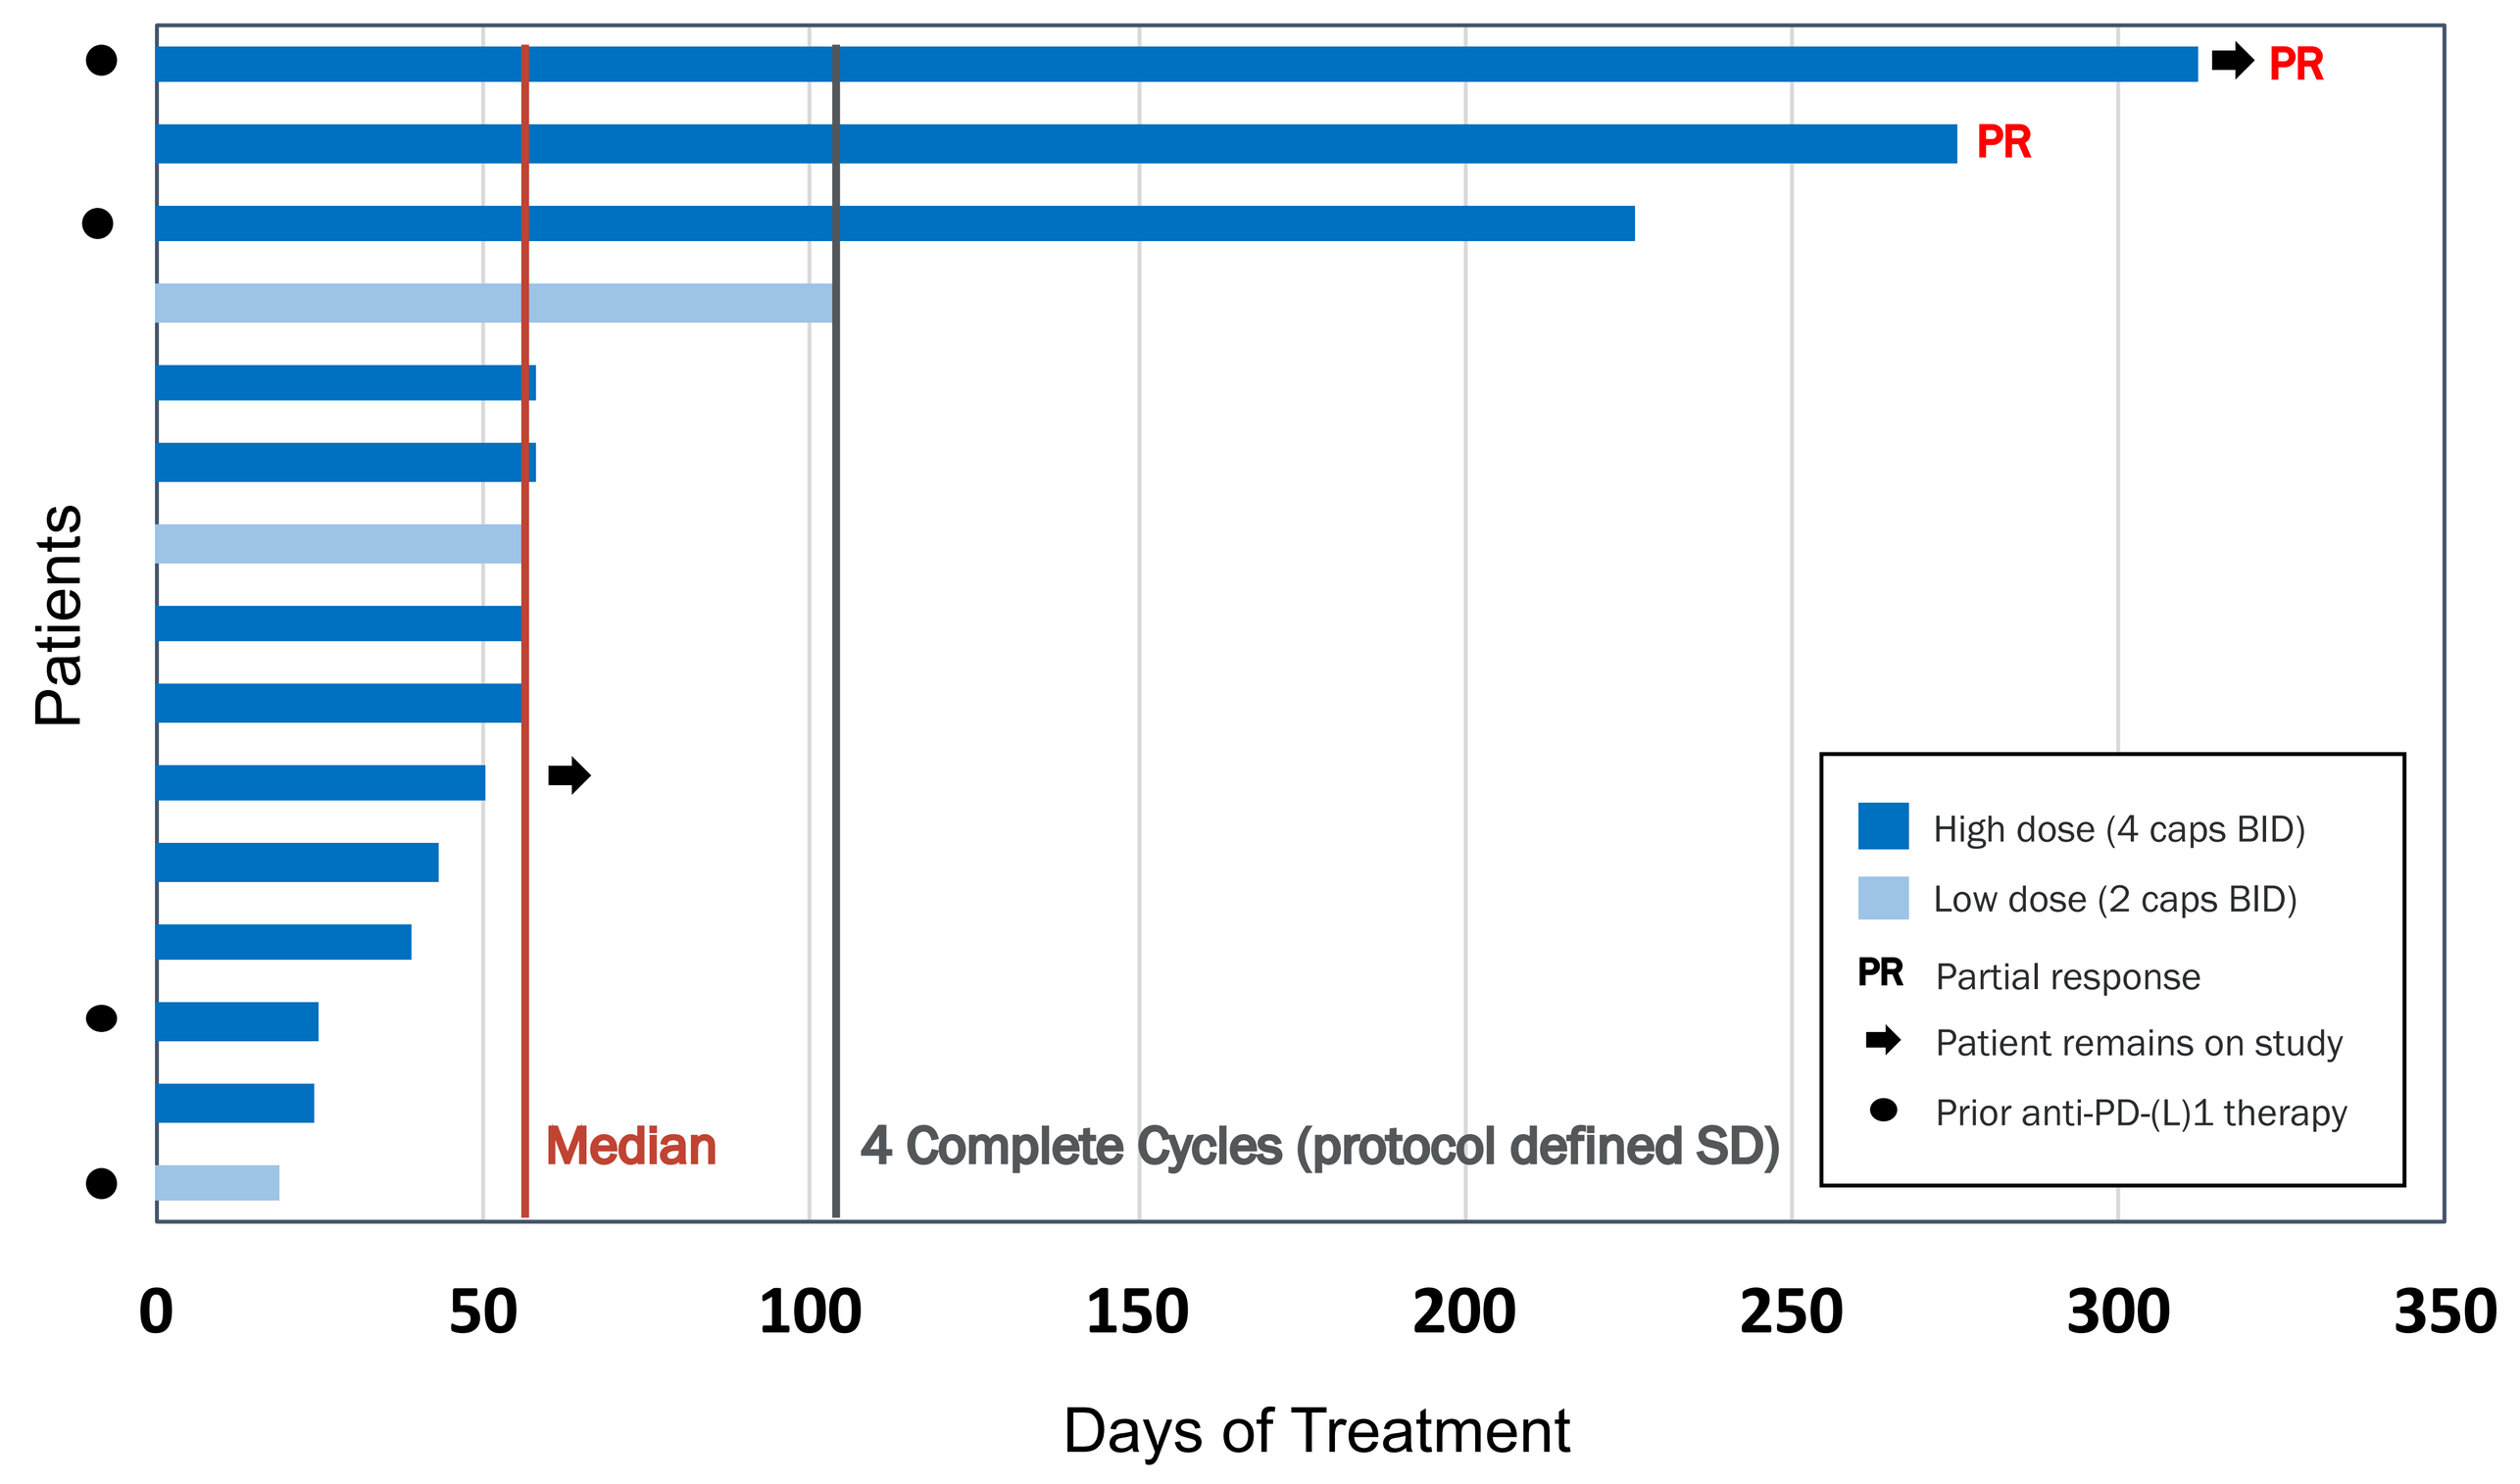
PR
PR
Patients
Median
4 Complete Cycles (protocol defined SD)
High dose (4 caps BID)
Low dose (2 caps BID)
PR Partial response
Patient remains on study
Prior anti-PD-(L)1 therapy
0
50
100
150
200
250
300
350
Days of Treatment

Fig 4B. Percent Change from Baseline Over Time for Target Lesions

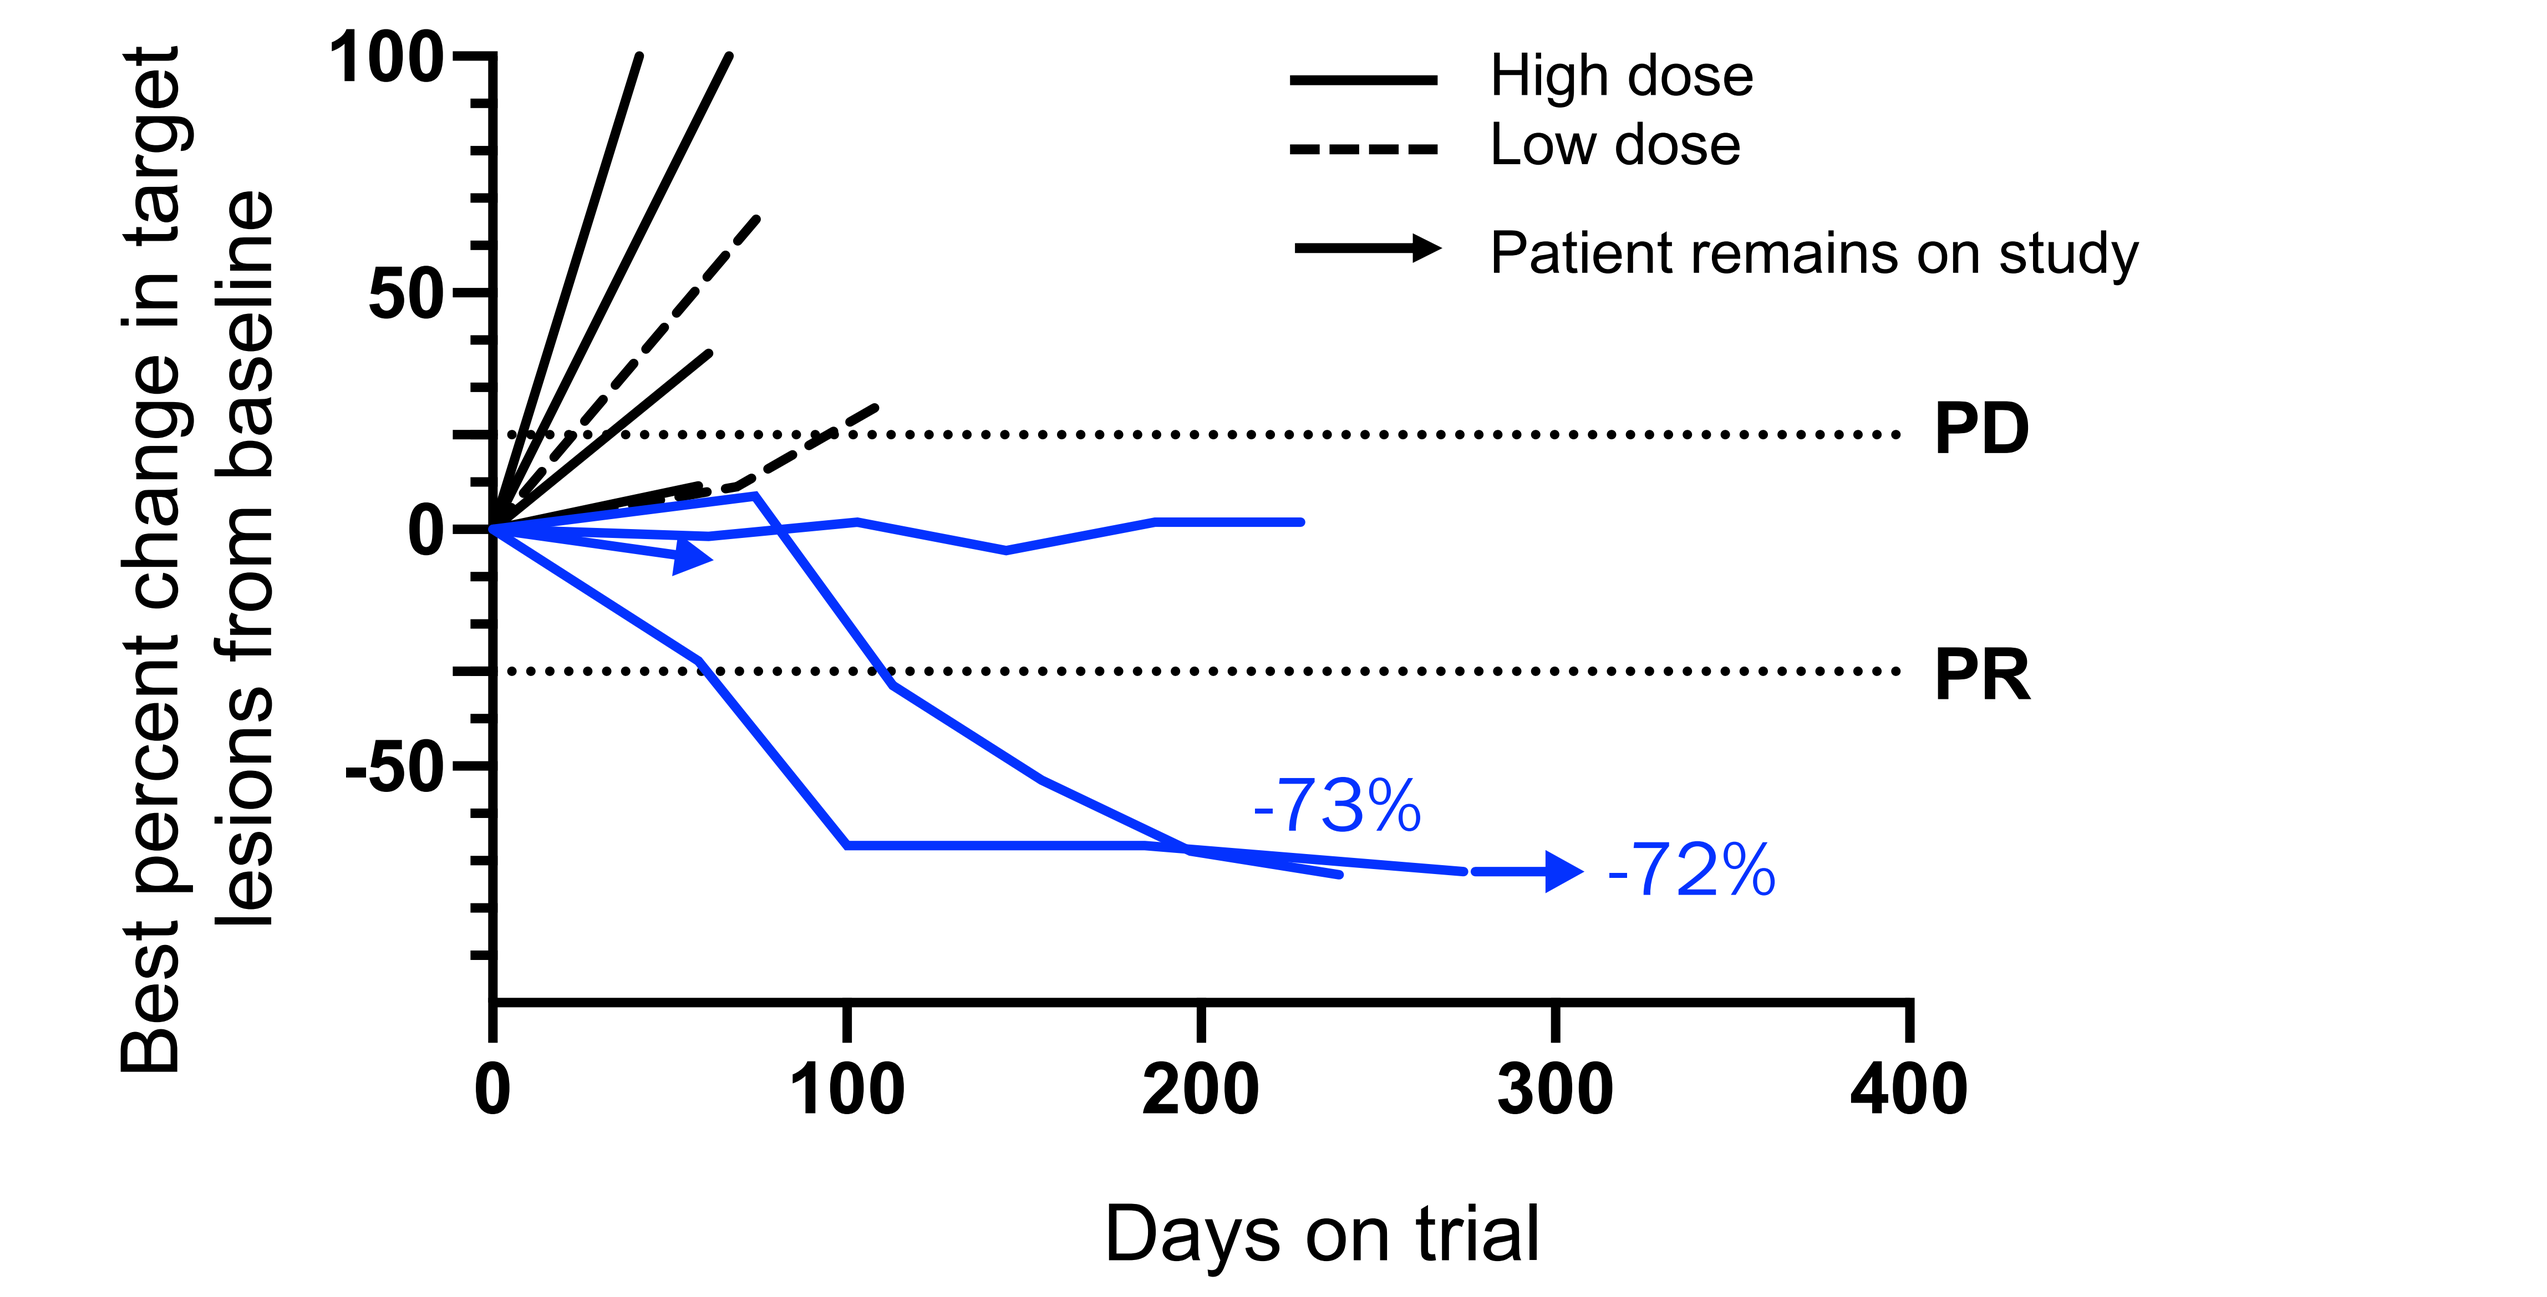
High dose
Low dose
Patient remains on study
PD
PR
-73%
-72%
Best percent change in target lesions from baseline
100
50
0
-50
0
100
200
300
400
Days on trial

Fig 4. TNBC patients (N=15) were treated with EDP1503 for 2 weeks prior to combination with pembrolizumab. Patients received EDP1503 at either a low dose of 2 enteric capsules twice daily (N=3) or a high dose of 4 enteric capsules twice daily (N=12). 2 patients on high dose treatment had partial responses by RECIST and 1 had best response of stable disease. (A) Duration on study treatment, as of 30 Oct 2020, and (B) Best percent change in target lesions of subjects who received tumor evaluation by radiographic scan.

Preliminary data, investigator assessment of best percentage change in target lesions by RECIST. Data cutoff 30 Oct 2020. Fig 4B does not include patients who were not evaluable due to early disease progression or termination prior to first response assessment.

CASE REPORT

Near complete response in TNBC patient who had relapsed on prior anti-PD-L1 therapy

Patient baseline characteristics and treatment history
- 48 year old Black female
- ECOG status of 1
- PD-L1 CPS score of 30
- 4 prior lines of therapy for metastatic disease:
 1) carboplatin and gemcitabine combination, 2) nab-paclitaxel. 3) PD-L1 and IDO inhibitor combination, 4) PI3K/mTOR and CHK1 inhibitor combination
- Patient was on a previous anti-PD-L1 combination regimen for 15 months with a partial response prior to progression.

Fig 5. Tumor reduction in responding patient previously relapsed on anti-PD-L1 therapy

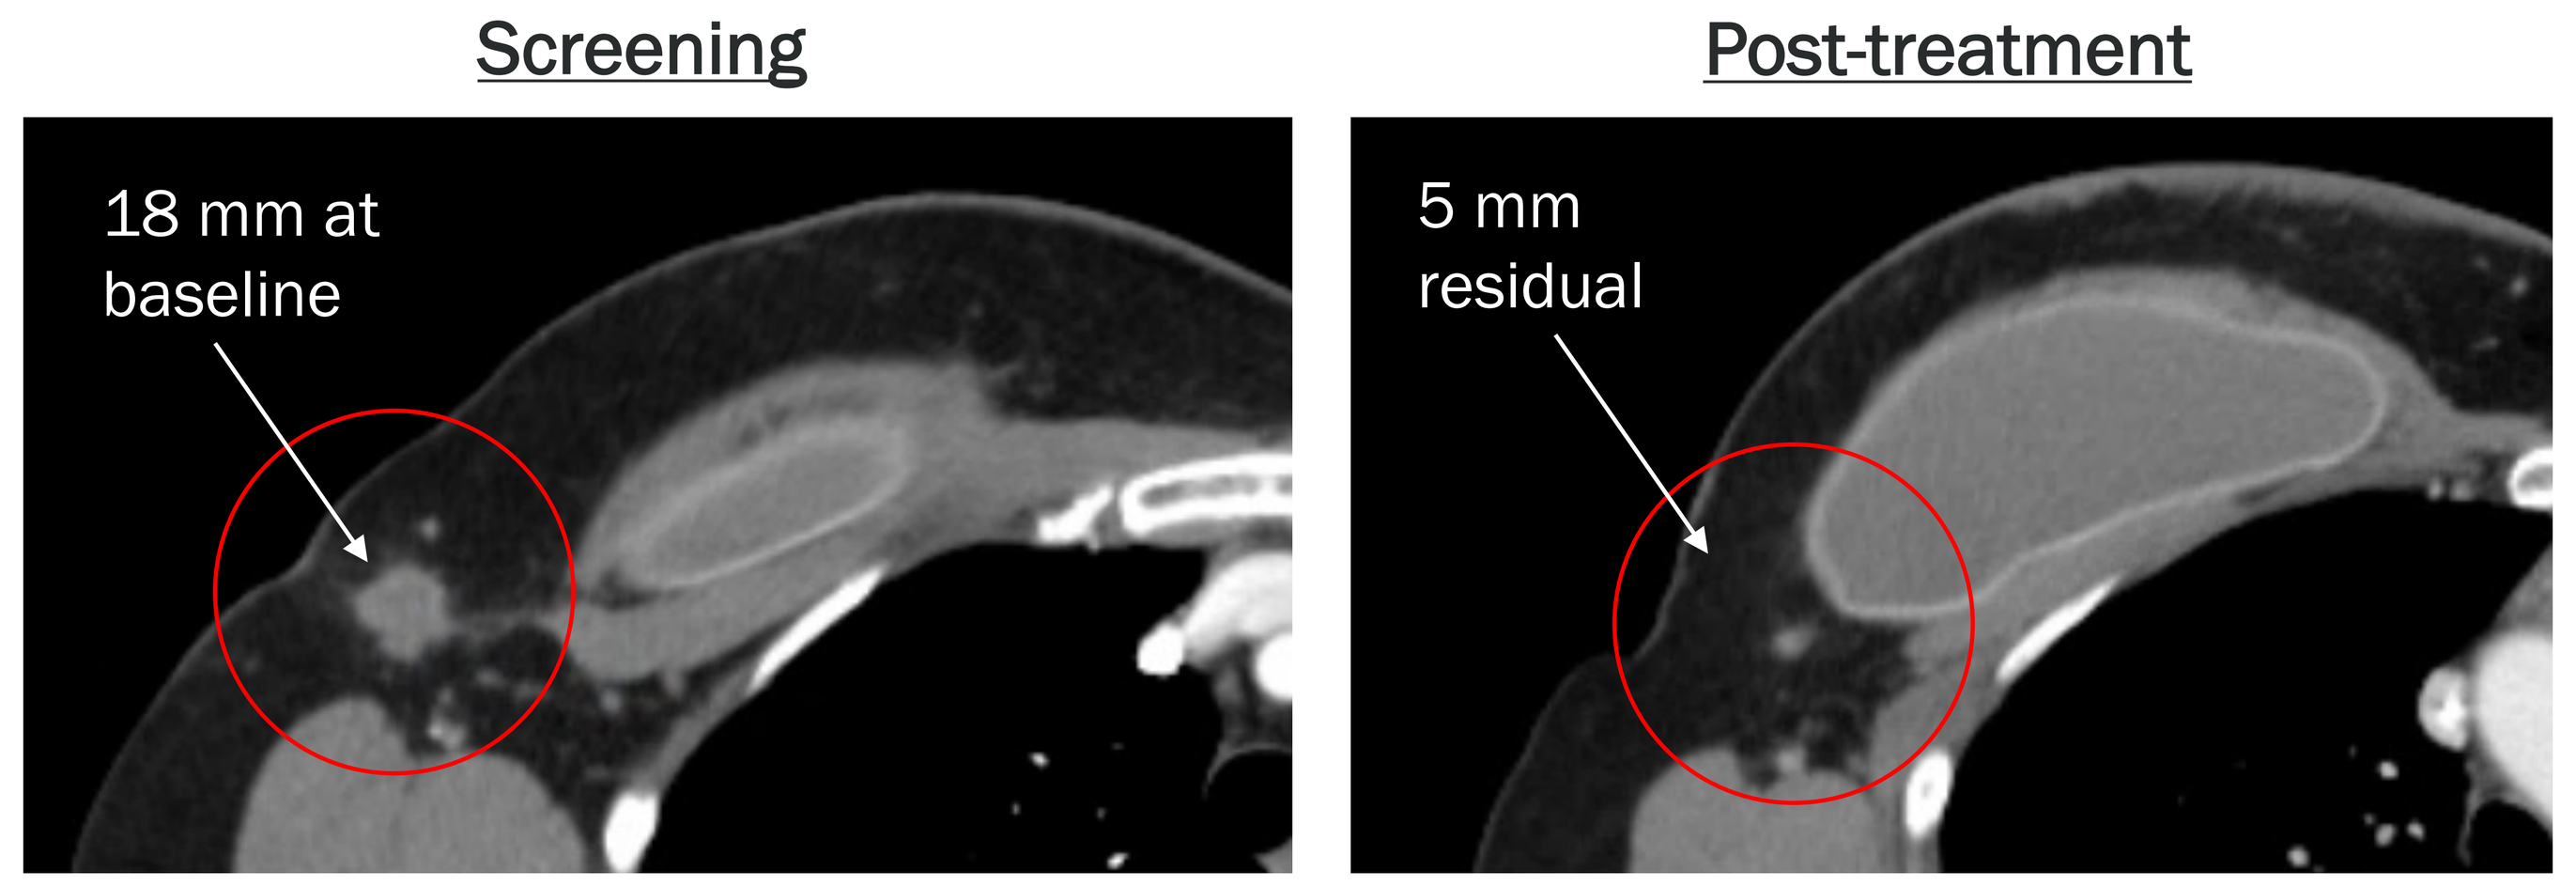
Screening
Post-treatment
18 mm at baseline
5 mm residual

Fig 5. CT scans showing tumor reduction from baseline in patient with ongoing partial response for 36 weeks, by RECIST criteria. Post-treatment scan performed after 10 months (14 cycles) of combination treatment.

Patient outcome on EDP1503 and pembrolizumab combination
- On treatment for 10.5 months on higher EDP1503 dose of 4 caps BID
- Patient has had an ongoing PR by RECIST criteria (72% tumor reduction)
- No measurable disease by PET scan but has a small residual lesion measuring 5 mm in diameter (right axillary soft tissue nodule)

CONCLUSIONS

- Single strains of microbes can alter systemic immunology without systemic exposure.
- The small intestine is the key site of action of EDP1503.
- Colonization is not required for pharmacological activity.
- Preclinically, orally delivered EDP1503 has multiple effects on the systemic immune system:
 - Induces a proinflammatory gene signature (CXCL10, IL12p70, IFNγ, IL-1β, TNF, and IL-6)
 - Activates effector lymphocyte populations (NK, NKT, CD4⁺, CD8⁺)
 - Induces a proinflammatory chemotactic signature within the TME (data not shown)
- The combination of EDP1503 and pembrolizumab is safe and well-tolerated with no treatment-related Grade 4/5 AEs or SAEs.
- Across all TNBC patients, ORR of 13% (2/15) and DCR of 20% (3/15).
- An ORR of 17% (2/12) and DCR of 25% (3/12) was observed in patients receiving the higher EDP1503 dose of 4 capsules BID.
- 1 TNBC patient, who had previously relapsed on an anti-PD-L1 combination therapy, has an ongoing near-complete response and has no measurable disease by PET scan.
- Partial responses were observed in 2 patients with greater than 2 lines of previous therapy; 1 patient had a best response of stable disease for 9 months. All 3 patients had received the higher dose treatment of EDP1503.
- The objective responses observed in 2 patients with TNBC, combined with excellent tolerability and preclinical data, support the potential of harnessing the small intestinal axis as a new and promising area in delivering oral medicines to activate systemic immunity and drive therapeutic responses.

REFERENCES:
1. Sivan A., *et al*., **Commensal Bifidobacterium promotes antitumor immunity and facilitates anti-PD-L1 efficacy.** Science. 2015 Nov 27;350(6264):1084-9.
2. Routy B., *et al*., **Gut microbiome influences efficacy of PD-1-based immunotherapy against epithelial tumors.** Science. 2018 Jan 5;359(6371):91-97.
3. Gopalakrishnan V., *et al*., **Gut microbiome modulates response to anti-PD-1 immunotherapy in melanoma patients.** Science. 2018 Jan 5;359(6371):97-103.
4. Matson V., *et al*., **The commensal microbiome is associated with anti-PD-1 efficacy in metastatic melanoma patients.** Science. 2018 Jan 5;359(6371):104-108.

ACKNOWLEDGEMENTS: This study was sponsored by Evelo Biosciences in collaboration with Merck & Co., Inc., Kenilworth, NJ USA. We thank the patients and their families, as well as all investigators and site personnel for participating in this study.

This presentation is the intellectual property of the author/presenter (Evelo Biosciences). Contact them at duncan@evelobio.com for permission to reprint and/or distribute.