Monoclonal microbial EDP1503 induces antitumor responses via gut-mediated activation of both innate and adaptive immunity

Humphrey Gardner, Michael Goldberg, Shubhra Kashyap, Chris Davitt, Shannon Argueta, Pooja Parameswaran, Peter Sandy, Holly Ponichtera, Mary Abdou, Mark Carlson, Maria Sizova, Valeria Kravitz, Erin Troy, Sam Andrewes, Mark Bodmer, and Loise Francisco
Evelo Biosciences, Cambridge MA

INTRODUCTION

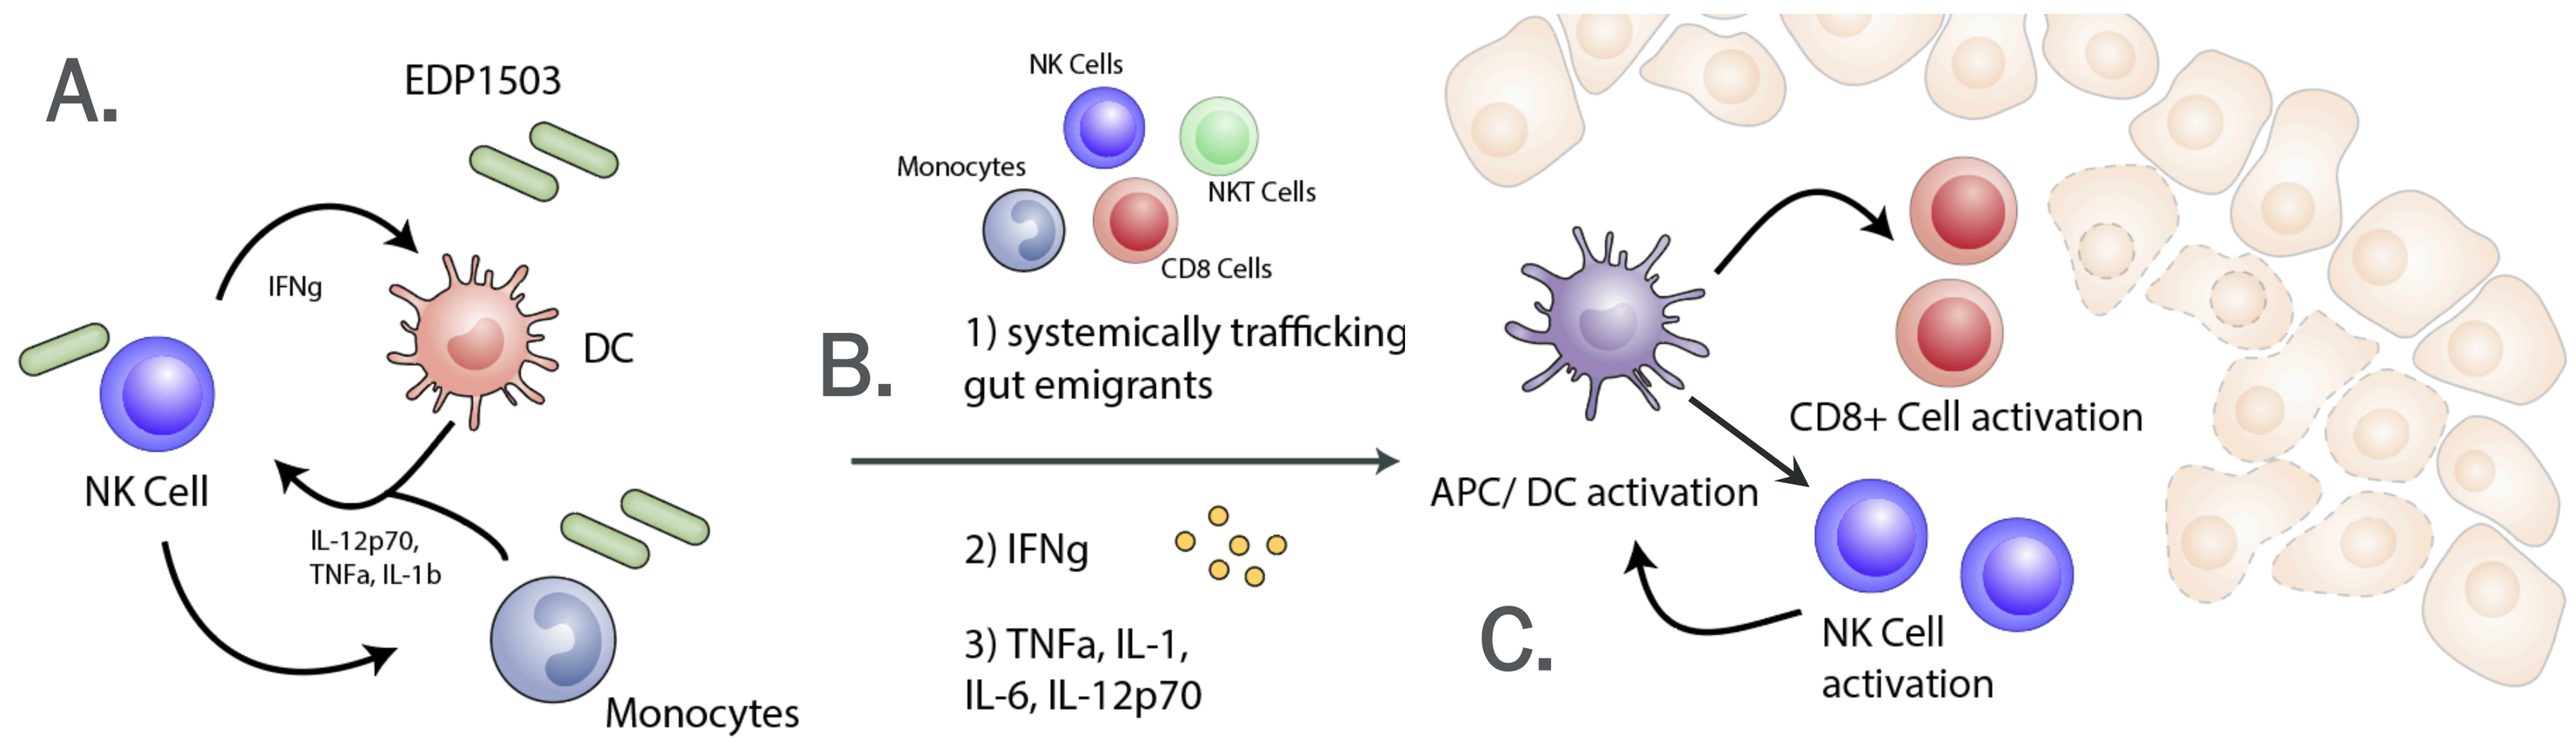
A.
EDP1503
IFNg
DC
NK Cell
IL-12p70, TNFa, IL-1b
Monocytes
B.
NK Cells
Monocytes
NKT Cells
CD8 Cells
1) systemically trafficking gut emigrants
2) IFNg
3) TNFa, IL-1, IL-6, IL-12p70
APC/ DC activation
CD8+ Cell activation
C.
NK Cell activation

EDP1503 is a human commensal strain of *Bifidobacterium animalis lactis with potent anti-tumor activity.* Treatment with EDP1503 *in vitro* promotes the production of type-I effector molecules by cells of the innate immune system, including the transactivation of human NK cells (**A**). Oral delivery of EDP1503 monotherapy leads to tumor control comparable to checkpoint inhibition in multiple subcutaneous syngeneic tumor models. This treatment promotes the immunogenic remodeling of the tumor microenvironment to favor the infiltration of protective effector cells (**B**). In the CT-26 colon carcinoma model, this protection is dependent on both NK and CD8⁺ T cells, however, NK cells are insufficient to mediate protection in the absence of CD8⁺ cytotoxic T lymphocytes (CTL). Our data suggests a model where EDP1503 stimulates NK cell transactivation which potentiates cross-priming of tumor-specific CTL by XCR1⁺ cDC1 to limit tumor growth (**C**).

RESULTS

EDP1503 Stimulates Type I Immune Responses

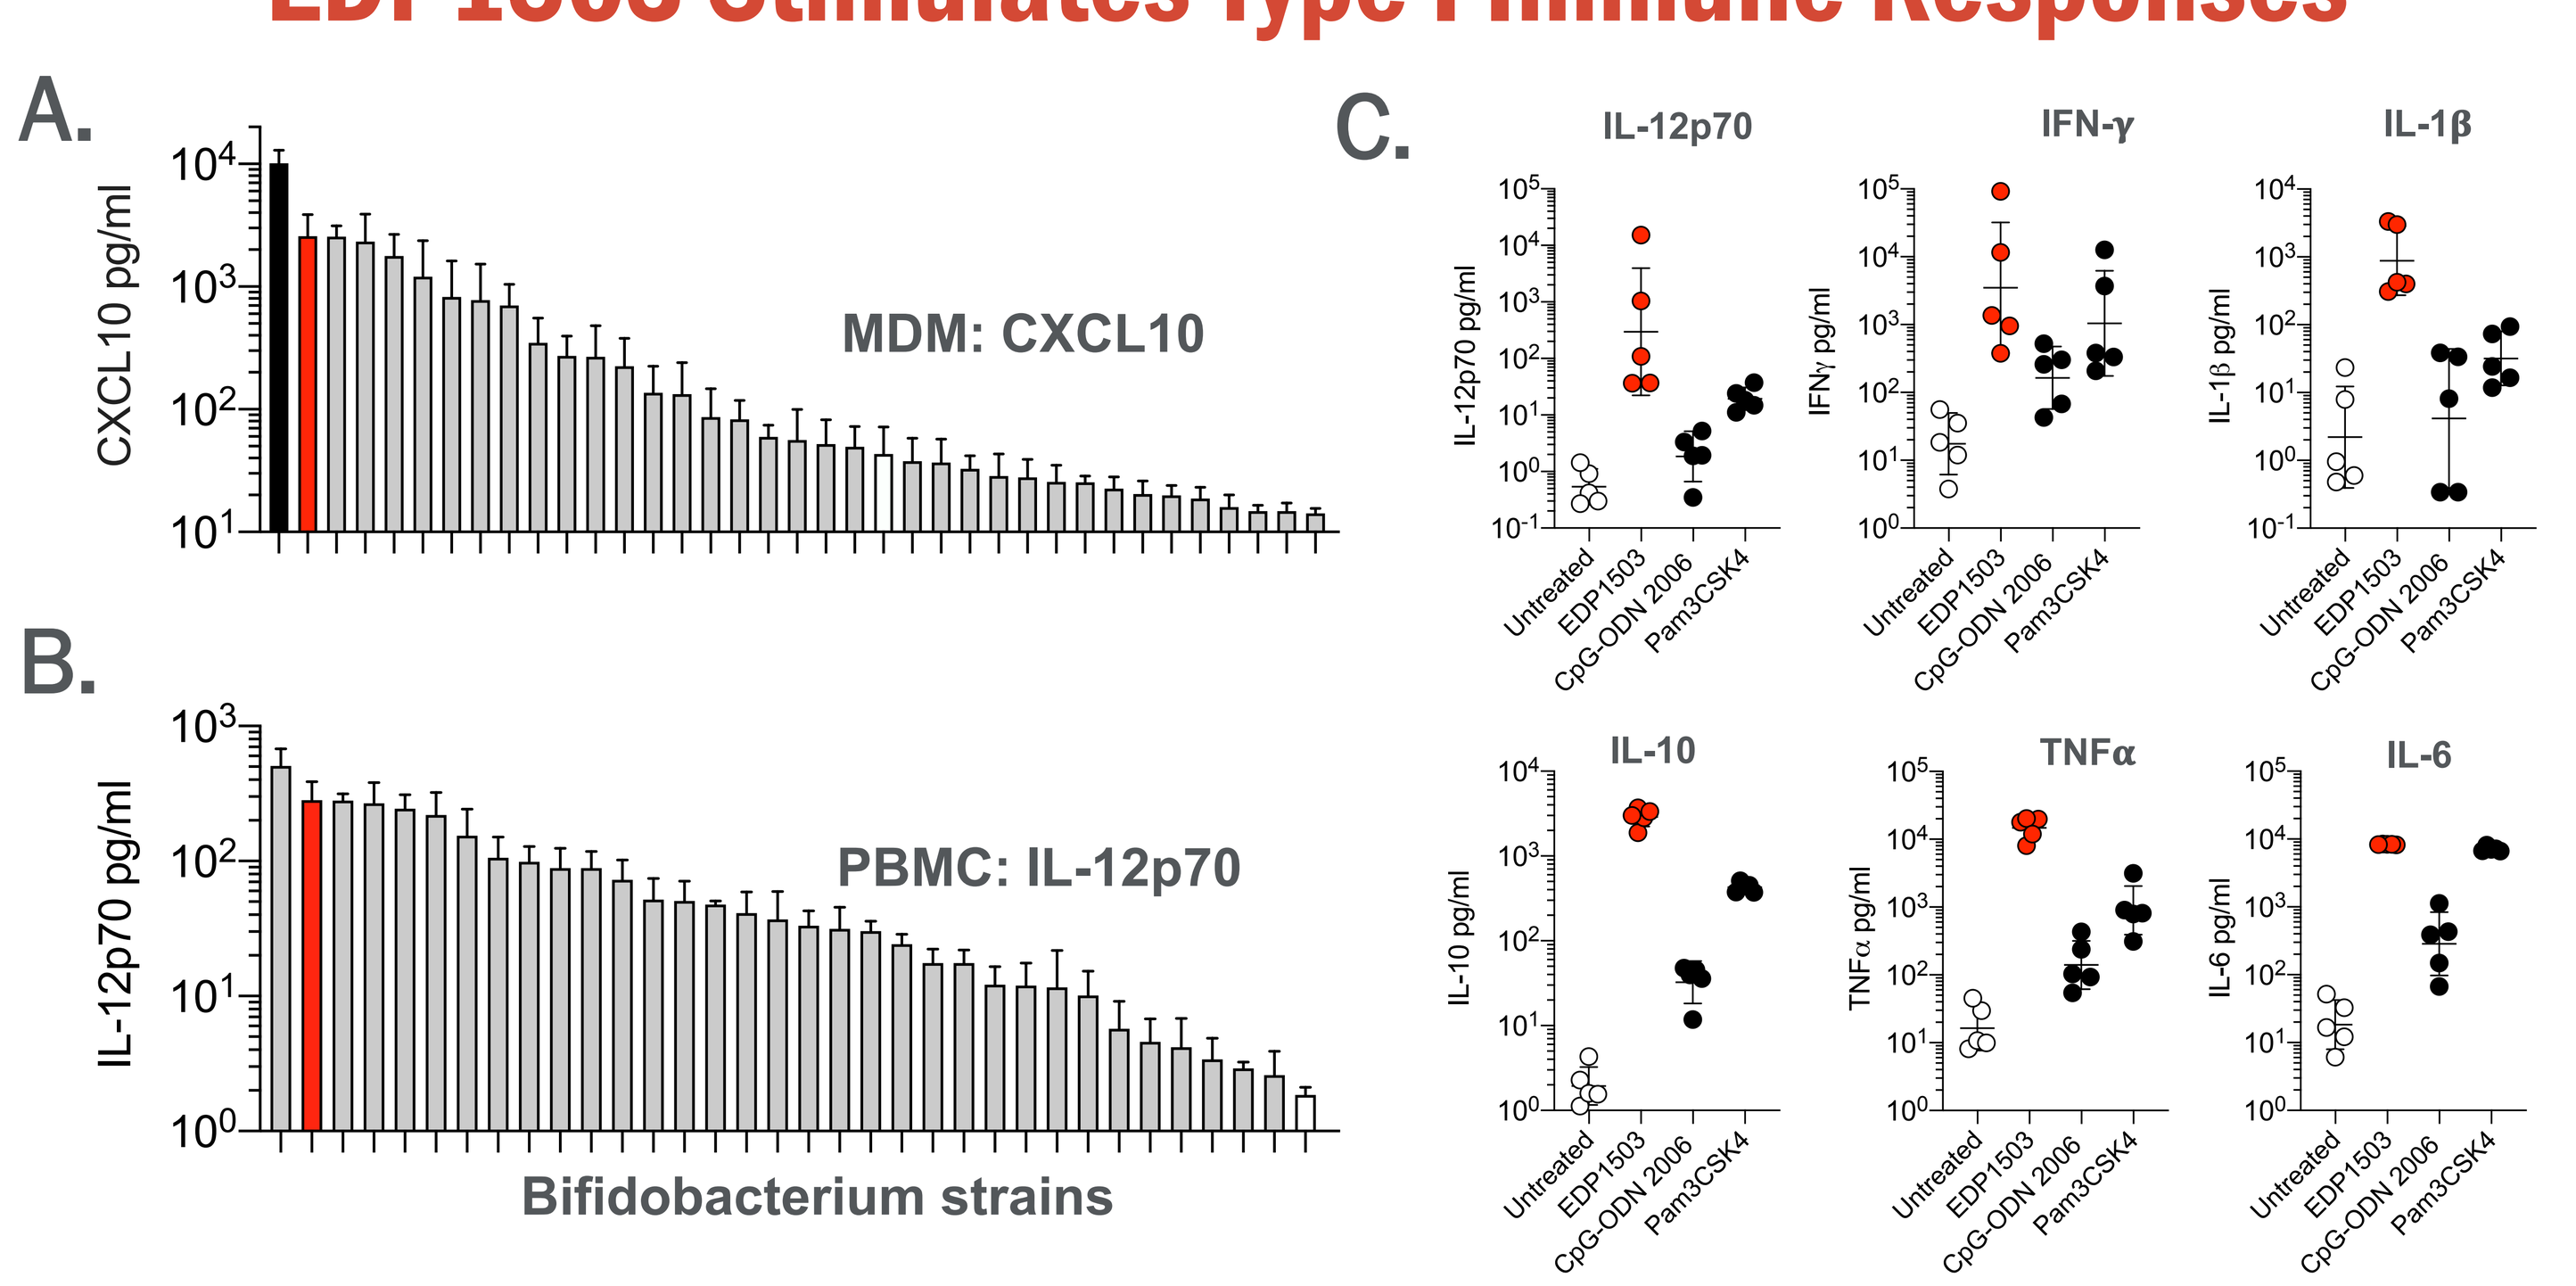
A.
CXCL10 pg/ml
MDM: CXCL10
B.
IL-12p70 pg/ml
PBMC: IL-12p70
Bifidobacterium strains
C.
IL-12p70
IFN-γ
IL-1β
IL-10
TNFα
IL-6
Untreated
EDP1503
CpG-ODN 2006
Pam3CSK4

Fig 1. (**A**) Human monocyte-derived macrophages stimulated with EDP1503 (shown in red) *in vitro* lead to the strong induction of CXCL10 compared to other strains of *Bifidobacterium* (M1 polarized macrophages shown in black; unstimulated control is shown in white). (**B**) Similarly, human PBMCs stimulated with EDP1503 induced significant IL-12p70 in comparison to other *Bifidobacterium.* (**C**) EDP1503 stimulates the PBMC production of a pro-inflammatory cytokine signature significantly greater than individual TLR2 or TLR9 agonists (N = 5 donors).

EDP1503 transactivates human NK cell cytotoxic activity

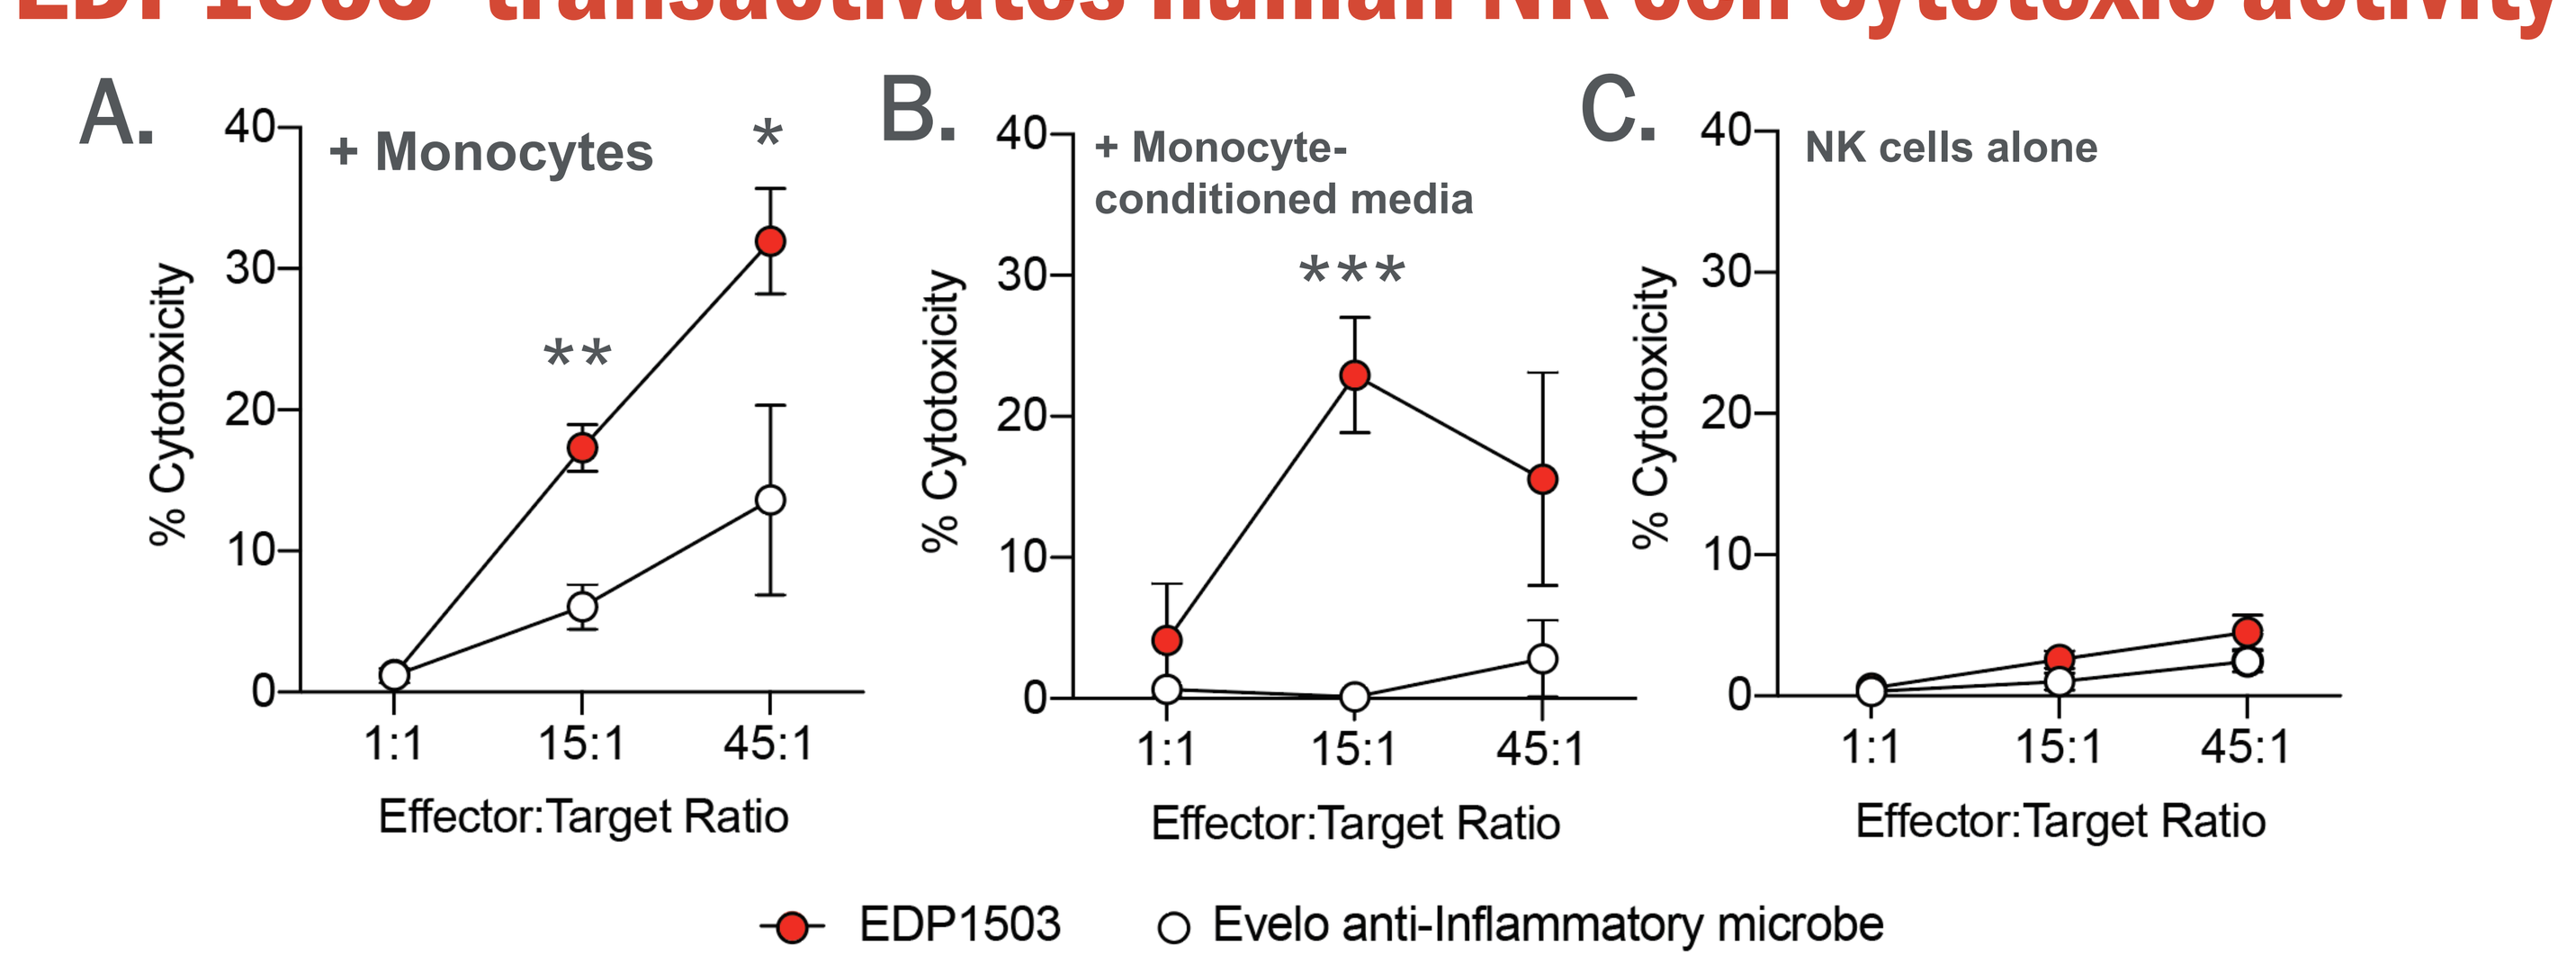
A.
+ Monocytes
B.
+ Monocyte-conditioned media
C.
NK cells alone
% Cytotoxicity
1:1
15:1
45:1
Effector:Target Ratio
EDP1503
Evelo anti-Inflammatory microbe

Fig 2. (**A**) Human monocytes were pulsed with EDP1503 or an Evelo anti-inflammatory microbe at an MOI of 1 for 18 hours and then cultured with purified autologous NK cells. NK cells were then transferred to wells containing CFSE-labeled K-562 cells for 4 hours and % cytotoxicity was measured by flow cytometry. (**B**) NK cells were cultured with microbe-treated monocyte-conditioned media and subsequently transferred to plates containing CFSE-labeled K-562 cells to assess cytotoxic activity. (**C**) NK cells alone were incubated with EDP1503 or an Evelo anti-inflammatory microbe prior to culture with CFSE-labeled K-562 cells. (* *p* > 0.05, ** *p* > 0.01, *** *p* > 0.001; Two-Way ANOVA)

EDP1503 stimulates anti-tumor effector activity in the MLN

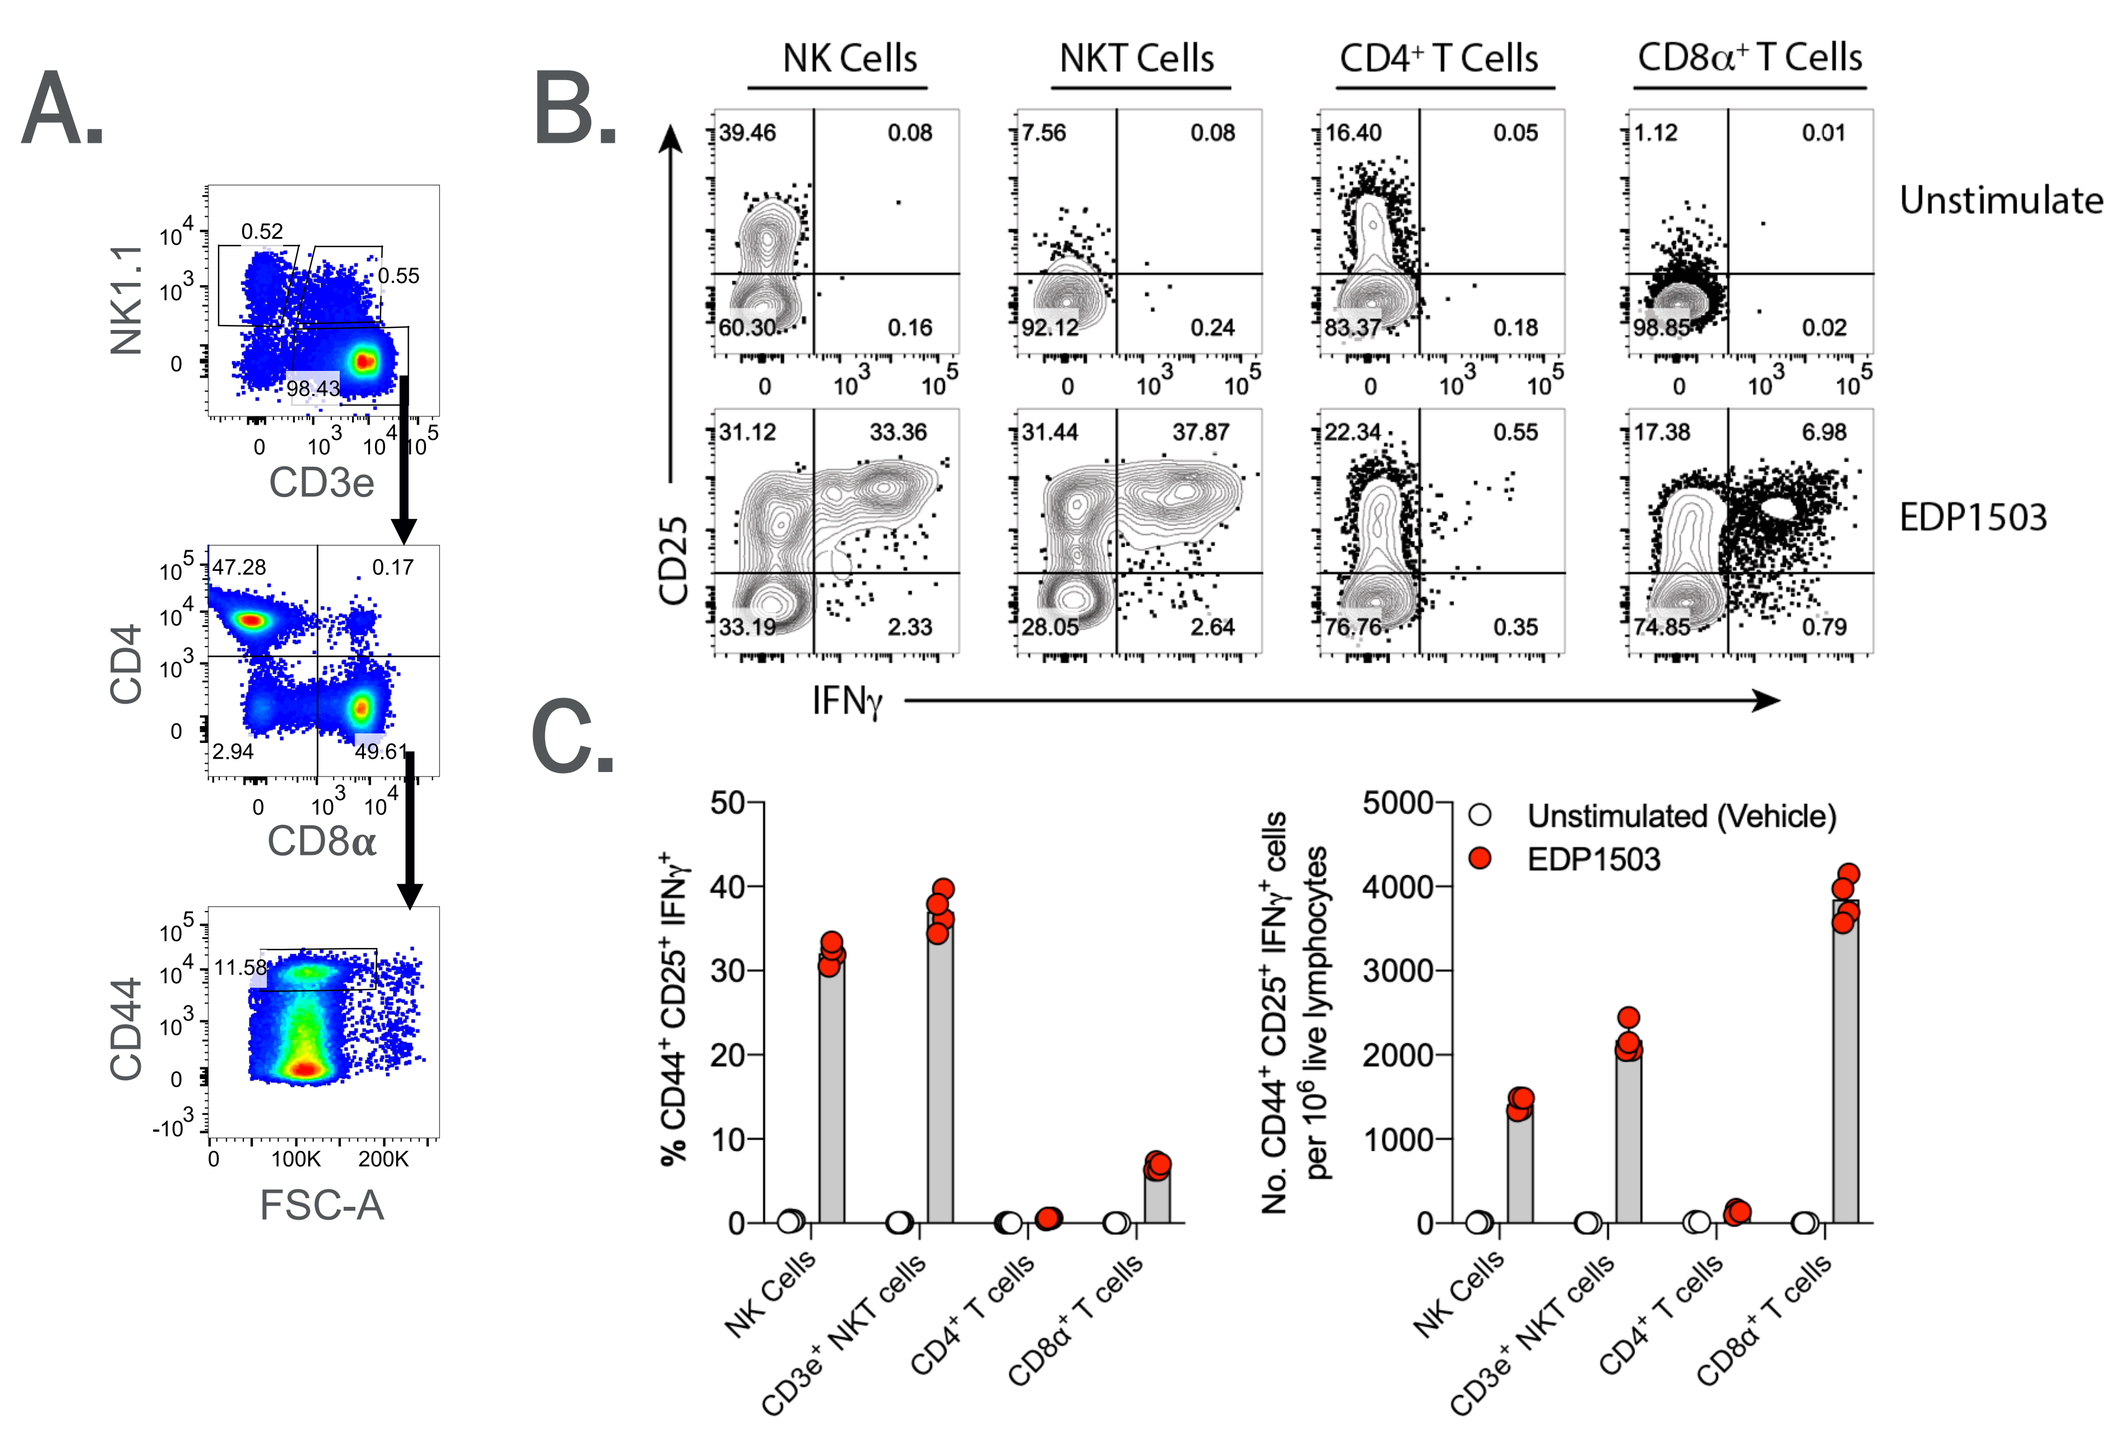
A.
NK1.1
CD3e
CD4
CD8α
CD44
FSC-A
B.
NK Cells
NKT Cells
CD4⁺ T Cells
CD8α⁺ T Cells
Unstimulated
EDP1503
CD25
IFNγ
C.
% CD44⁺ CD25⁺ IFNγ⁺
No. CD44⁺ CD25⁺ IFNγ⁺ cells per 10⁶ live lymphocytes
Unstimulated (Vehicle)
EDP1503
NK Cells
CD3e⁺ NKT cells
CD4⁺ T cells
CD8α⁺ T cells

Fig 3. (**A**) Single-cell suspensions from mesenteric lymph nodes (MLN) were treated with EDP1503 overnight. (**B**) Cellular activation of CD44⁺ effector lymphocyte populations (NK, NKT, CD4, CD8) was assessed via expression of CD25 and IFN-γ. (**C**) Frequency and number of CD44⁺CD25⁺ IFN-γ⁺ effector cells in the MLN following treatment with EDP1503 *ex vivo*.

Oral delivery of EDP1503 is as effective as anti-PD-1 in multiple tumor models *in vivo*

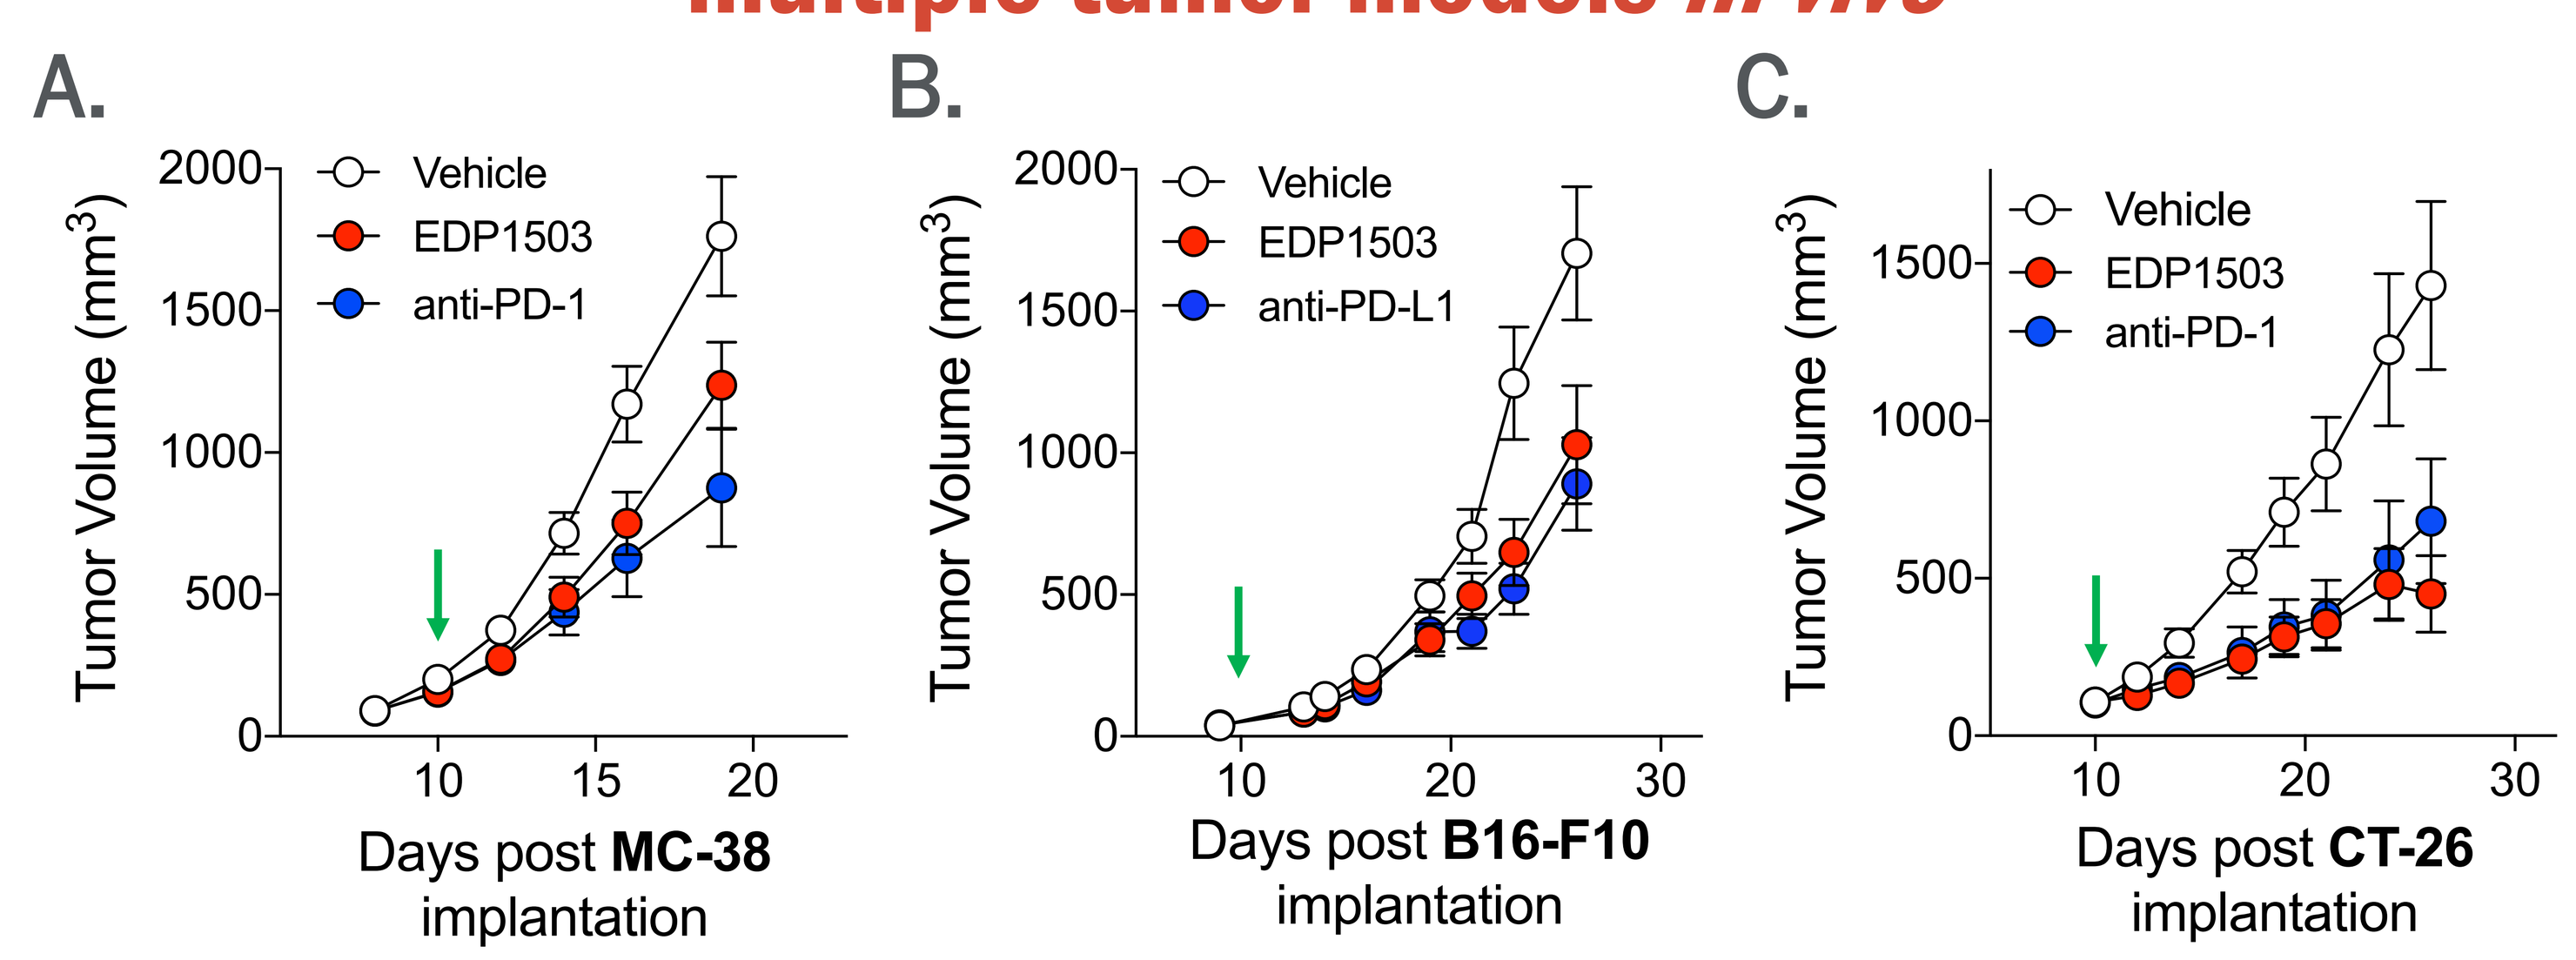
A.
Vehicle
EDP1503
anti-PD-1
Tumor Volume (mm³)
Days post **MC-38** implantation
B.
Vehicle
EDP1503
anti-PD-L1
Days post **B16-F10** implantation
C.
Vehicle
EDP1503
anti-PD-1
Days post **CT-26** implantation

Fig 4. Syngeneic mice were inoculated with MC-38 (**A**), B16-F10 (**B**), or CT-26 (**C**) tumor cells at 10^5 subcutaneously. Tumors were established for 10 days (~100mm³) prior to initiation of treatment (indicated by green arrow). Mice were treated with either sucrose vehicle or EDP1503 via oral gavage daily or treated with anti-PD-1 (A, C) or anti-PD-L1 (B) every four days for the duration of the study.

Oral treatment with EDP1503 distally programs the TME toward immune cell promotion

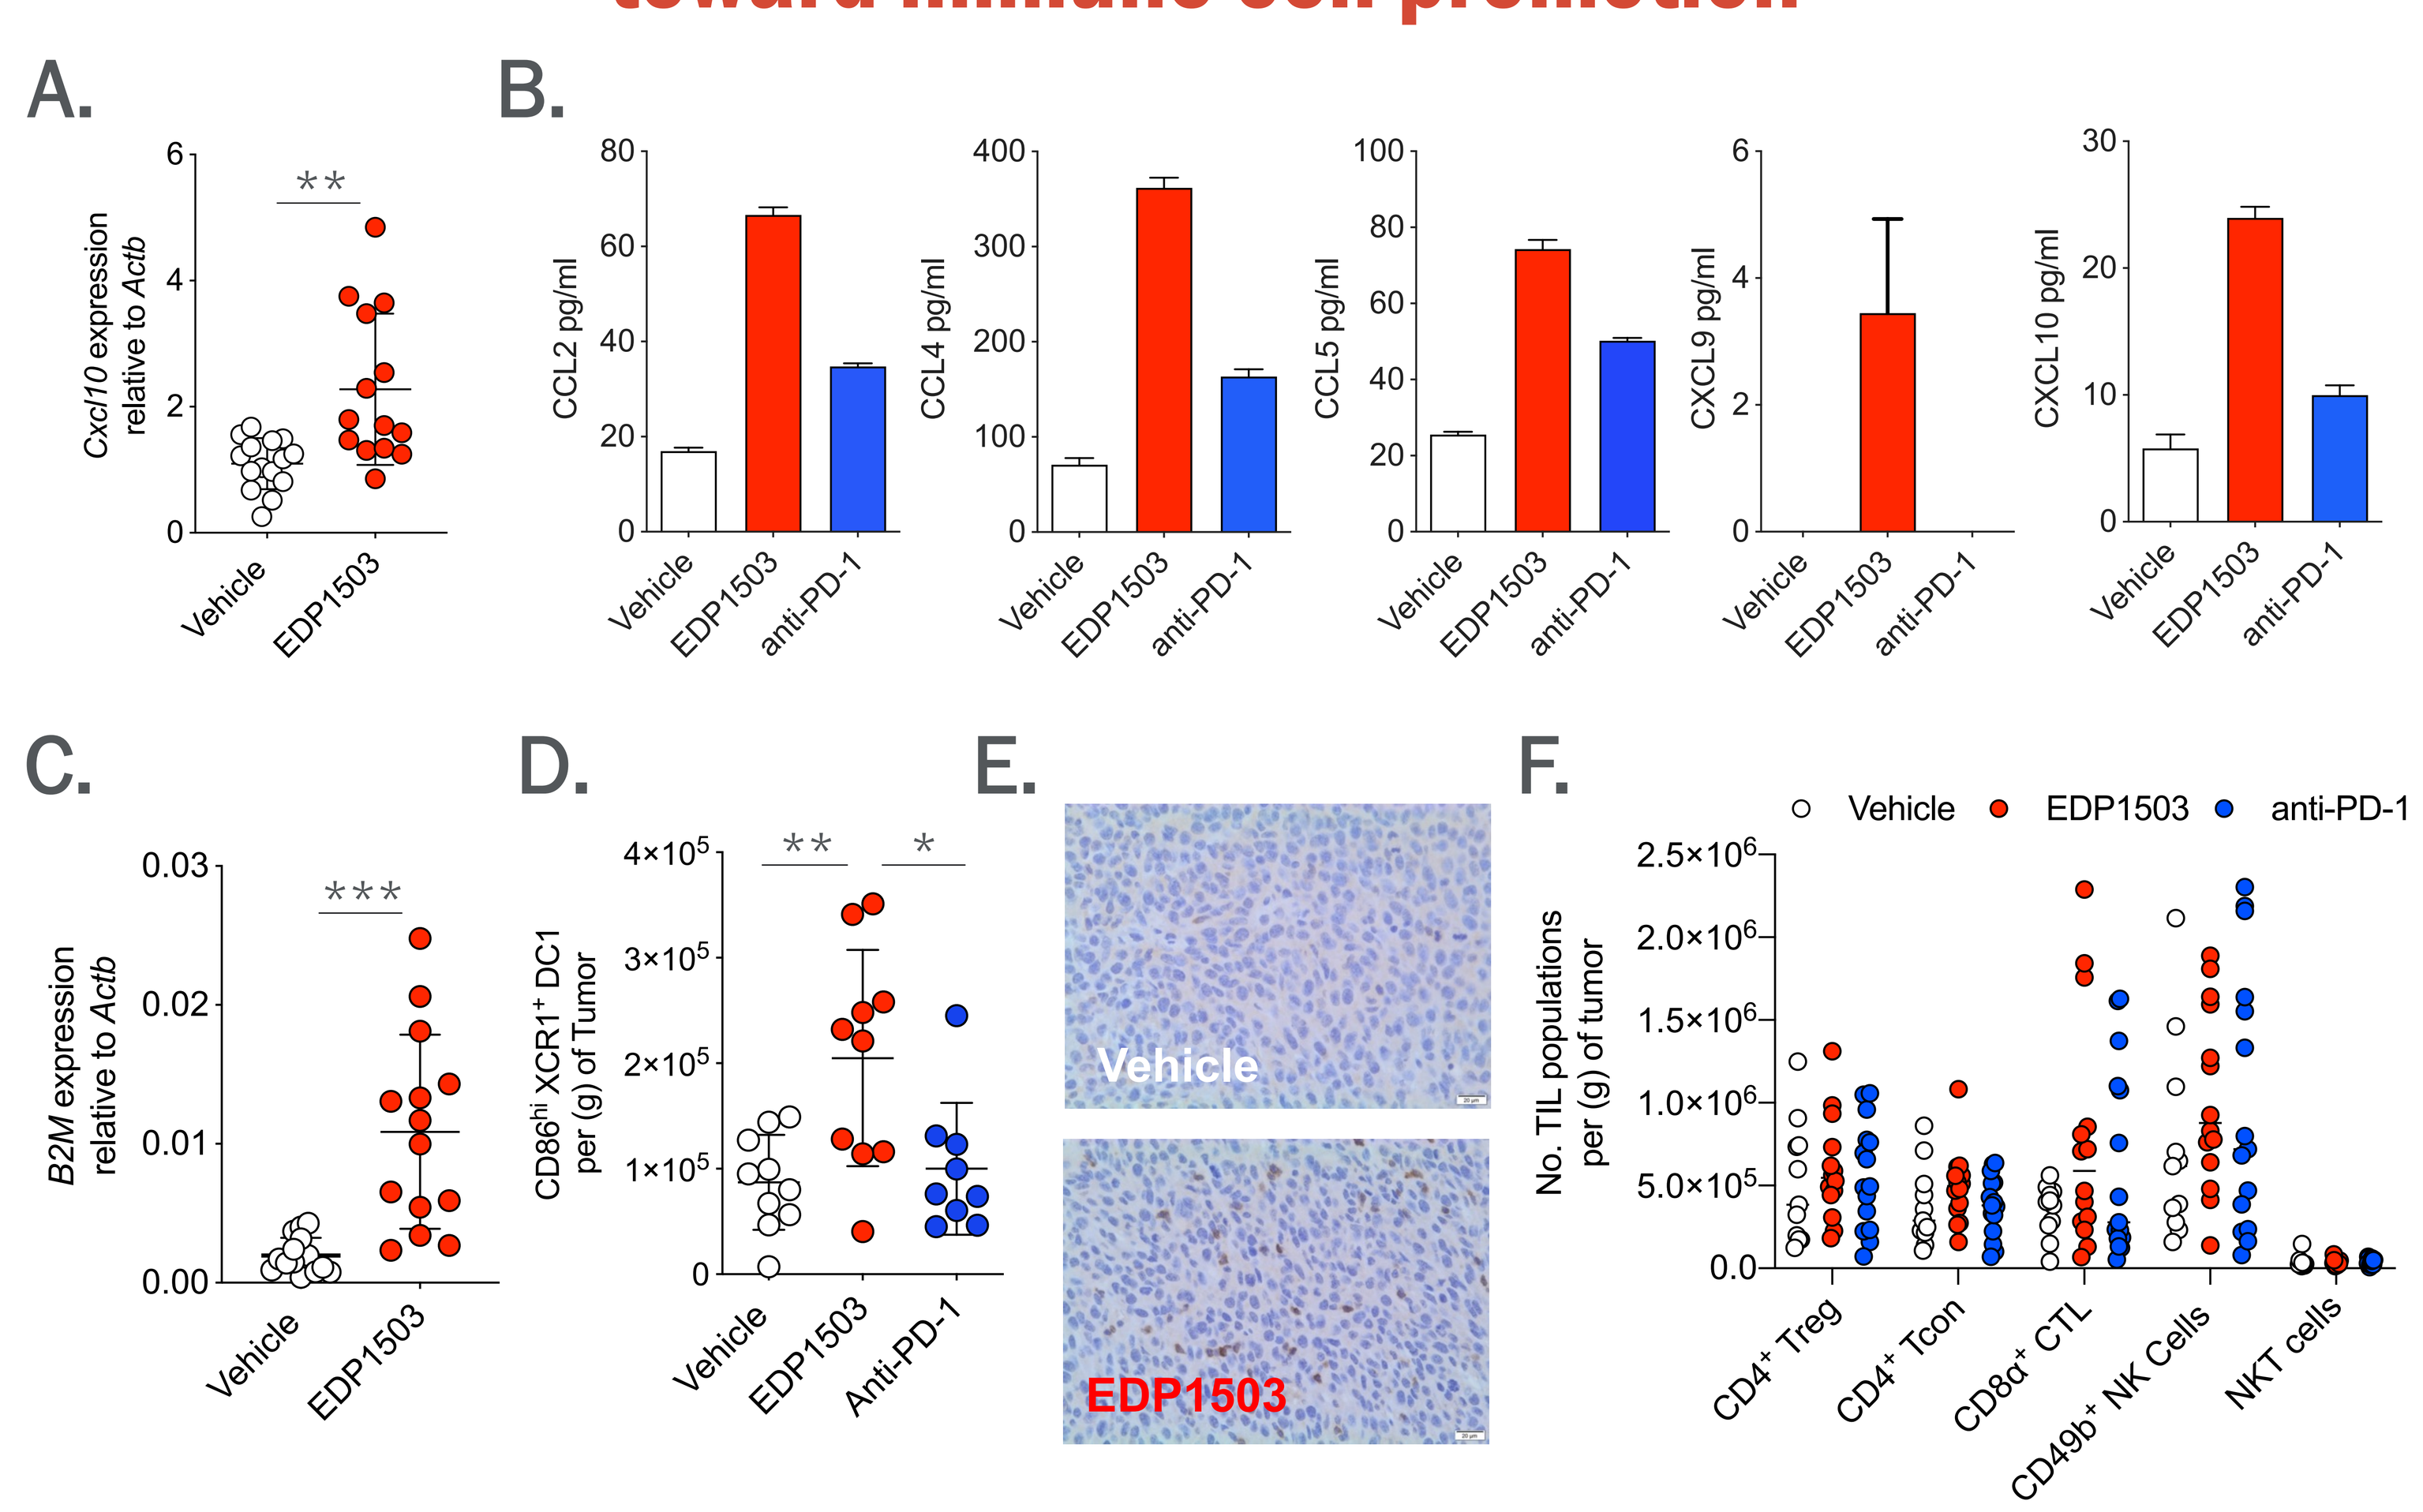
A.
Cxcl10 expression relative to *Actb*
B.
CCL2 pg/ml
CCL4 pg/ml
CCL5 pg/ml
CXCL9 pg/ml
CXCL10 pg/ml
Vehicle
EDP1503
anti-PD-1
C.
B2M expression relative to *Actb*
D.
CD86ʰⁱ XCR1⁺ DC1 per (g) of Tumor
Anti-PD-1
E.
Vehicle
EDP1503
F.
No. TIL populations per (g) of tumor
CD4⁺ Treg
CD4⁺ Tcon
CD8α⁺ CTL
CD49b⁺ NK Cells
NKT cells

Fig 5. (**A**) RT-PCR analysis of mRNA derived from CT-26 tumor lysate indicates an increase in the relative expression of the pro-inflammatory chemokine CXCL10 and the antigen presenting molecule, **β**2M (**C**). (**B**) Similarly, *ex vivo* analysis of purified CD45⁺ cells from CT-26 tumors revealed strong induction of chemokines consistent with immunogenic remodeling of the TME. (**D**) CT-26 tumors from EDP1503-treated mice are infiltrated with greater numbers of activated XCR1⁺ cDC1 than control mice. (**E**) Histological examination of CT26 tumors reveals increase CD3⁺ TILs in mice treated with EDP1503. (**F**) Quantification of various TIL populations following EDP1503 treatment.

EDP1503 activates effector lymphocyte populations in the TME

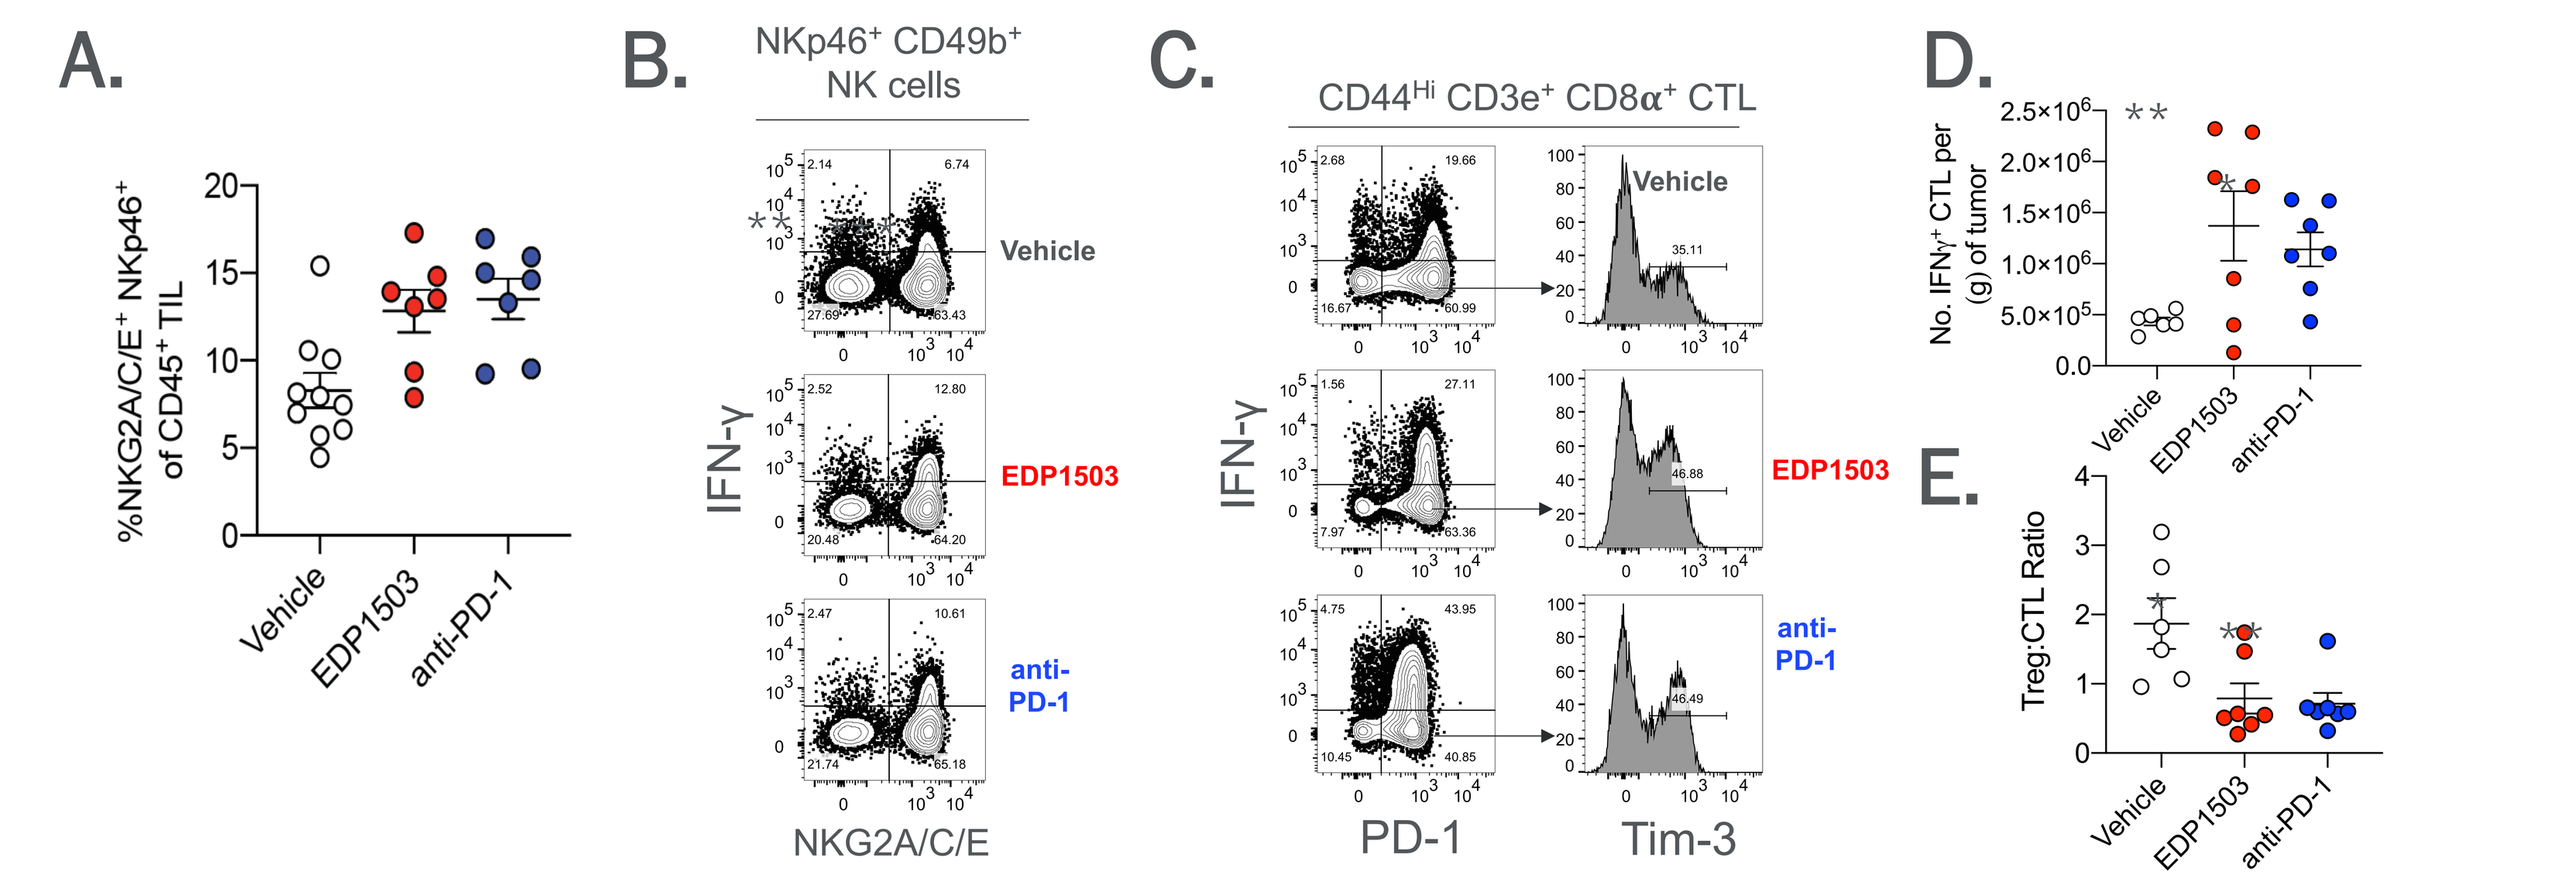
A.
%NKG2A/C/E⁺ NKp46⁺ of CD45⁺ TIL
Vehicle
EDP1503
anti-PD-1
B.
NKp46⁺ CD49b⁺ NK cells
IFN-γ
NKG2A/C/E
C.
CD44ᴴⁱ CD3e⁺ CD8α⁺ CTL
PD-1
Tim-3
D.
No. IFNγ⁺ CTL per (g) of tumor
E.
Treg:CTL Ratio

Fig 6. (**A**) EDP1503 treatment of CT26 bearing mice have increased NKp46⁺NKG2A/C/E⁺ NK cells within the TME comparable to anti-PD-1 treatment. (**B**) EDP1503 induces IFNγ production by NKp46⁺NKG2A/C/E⁺ NK cells. (**C**) EDP1503 results in elevated IFNγ production and concomitant upregulation of TIM-3 by CD44⁺CD8 CTL within the TME, (quantified in (**D**)). (**E**) Similar to anti-PD-1, EDP1503 treatment results in decreased Treg to CTL ratio within the TME. Tumors were analyzed following 5 days of EDP1503 treatment.

NK cells and CD8α⁺ CTLs cooperate for optimal control of tumor growth

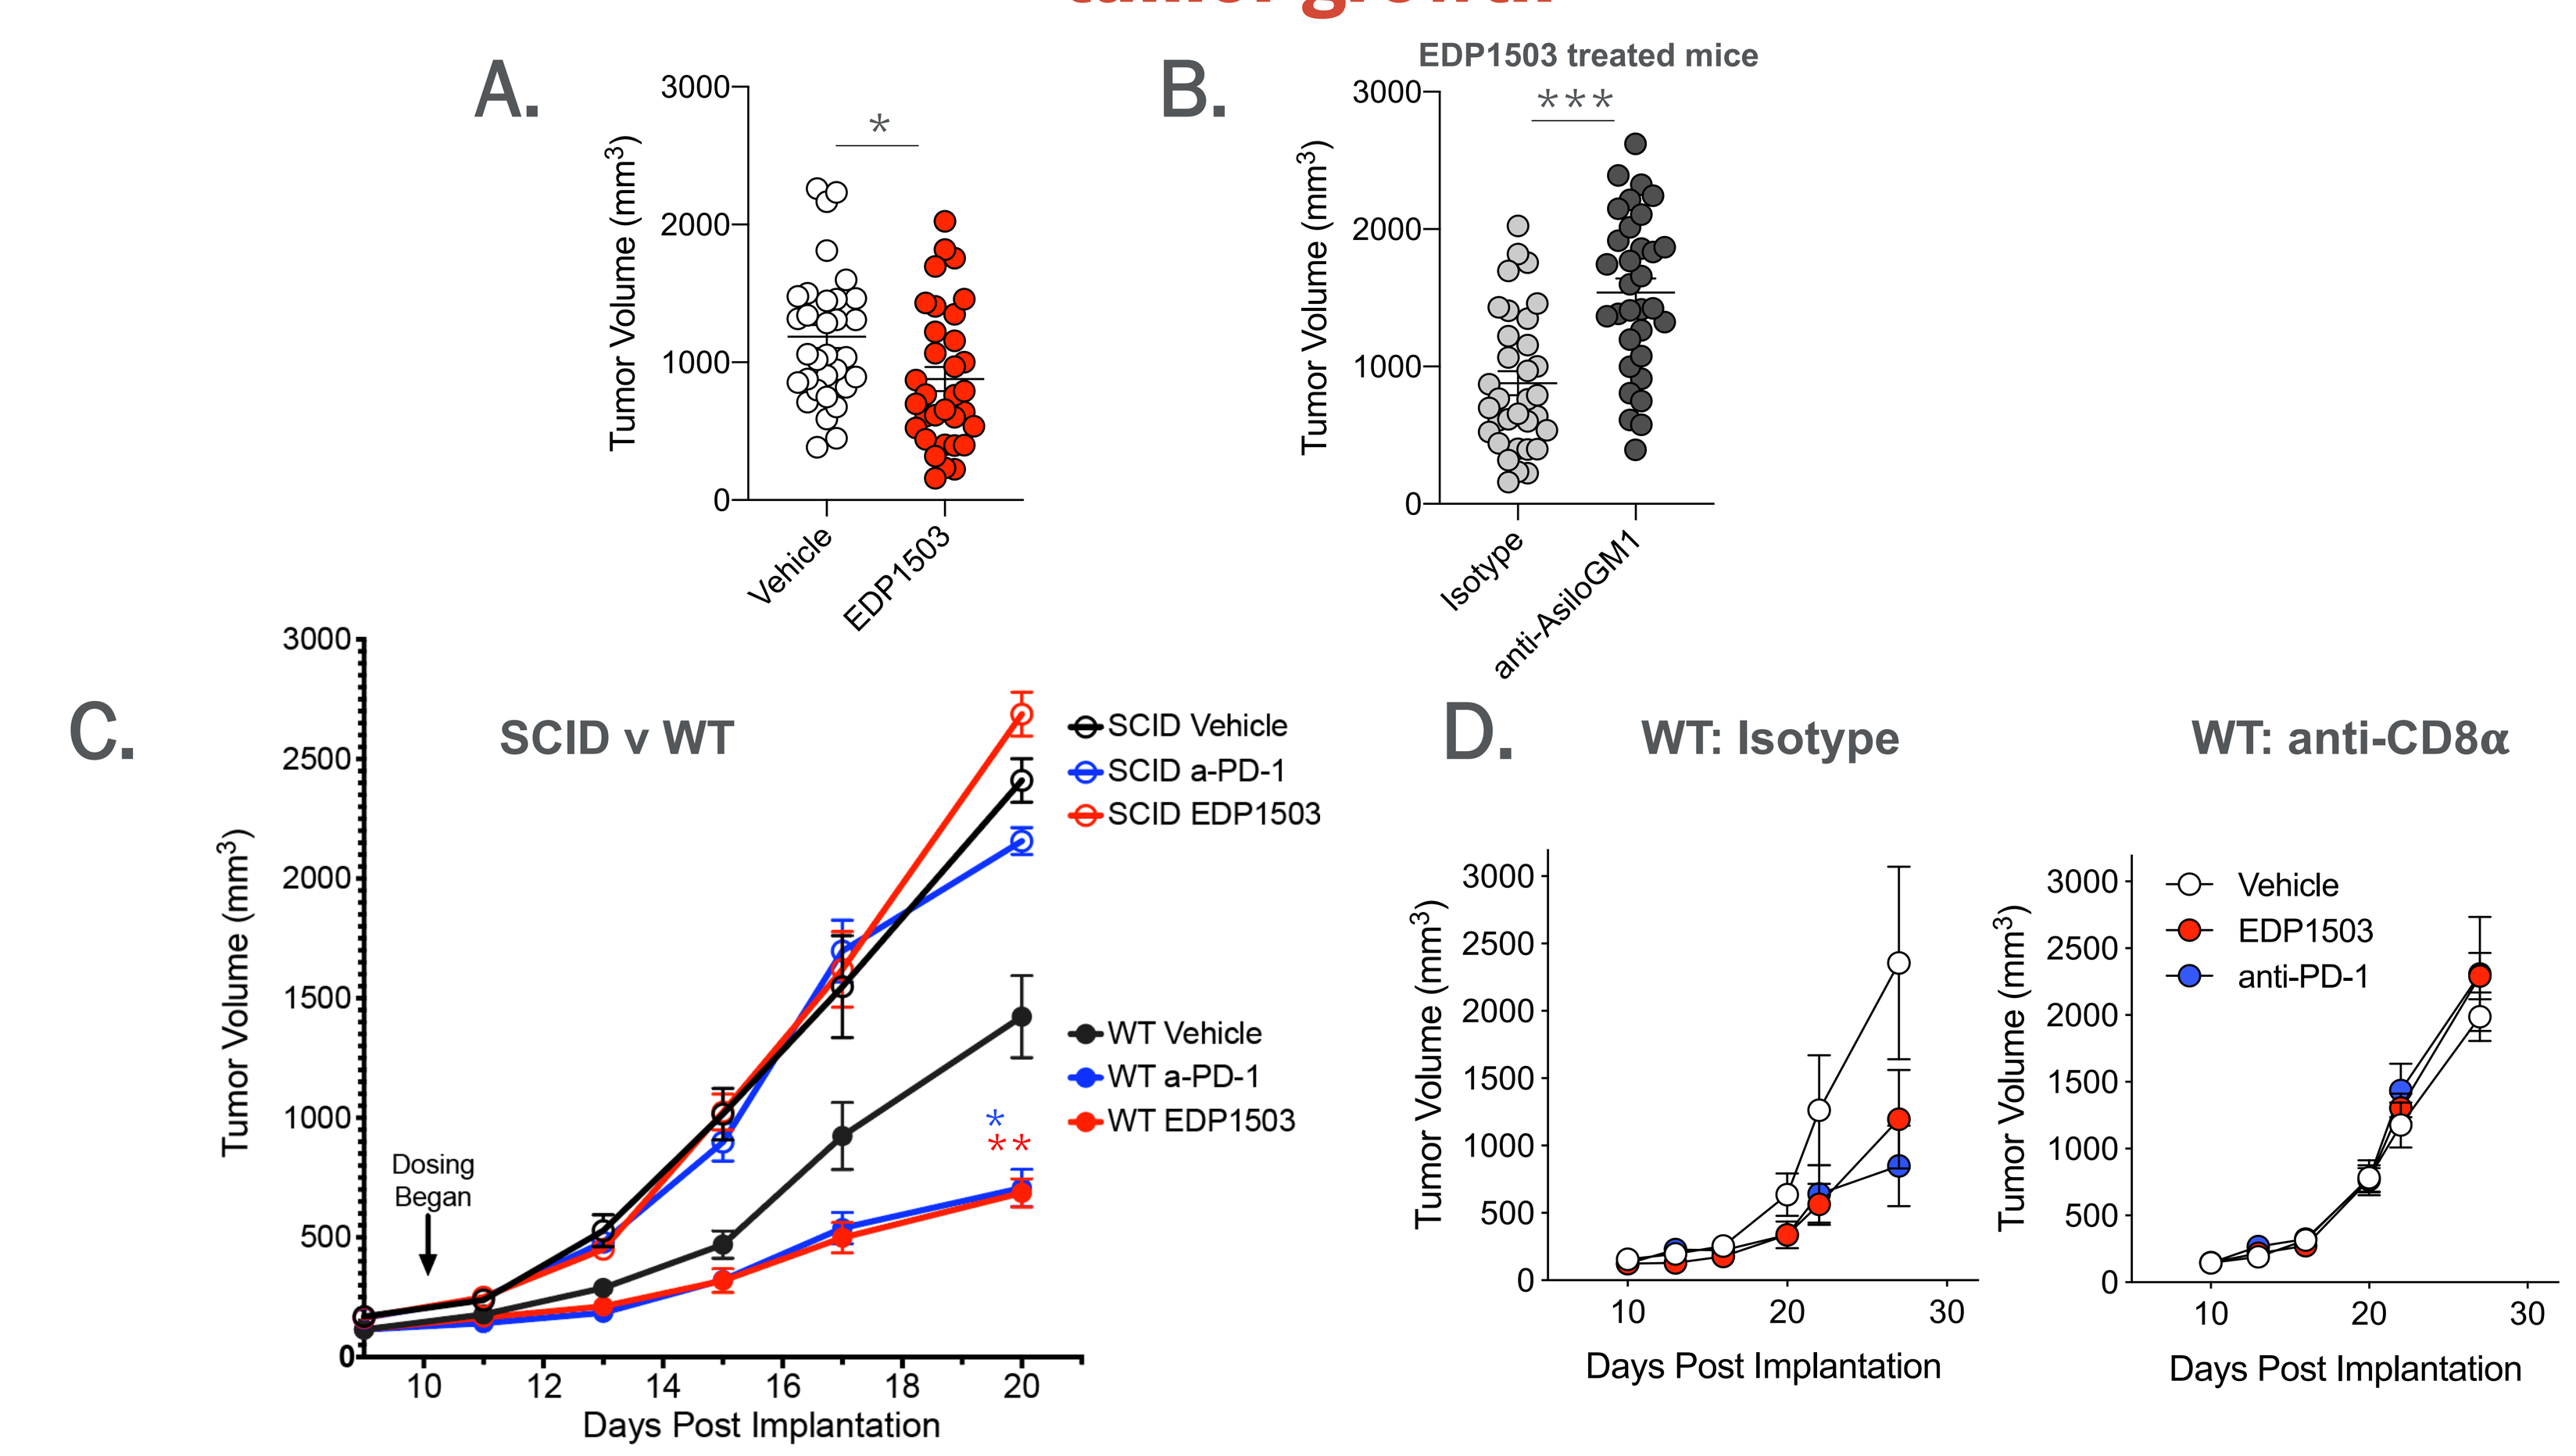
A.
Tumor Volume (mm³)
Vehicle
EDP1503
B.
EDP1503 treated mice
Isotype
anti-AsiloGM1
C.
SCID v WT
SCID Vehicle
SCID a-PD-1
SCID EDP1503
WT Vehicle
WT a-PD-1
WT EDP1503
Dosing Began
Days Post Implantation
D.
WT: Isotype
WT: anti-CD8α
Vehicle
EDP1503
anti-PD-1

Fig 7. (**A**) EDP1503 treated WT mice have decreased tumor burden relative to vehicle. (**B**) Depletion of NK cells with anti-asialoGM1 reveals an NK-mediated anti-tumor response induced by EDP1503. (**C**) Loss of efficacy of EDP1503 in SCID mice inoculated with CT26 tumor cells. (**D**) CD8⁺ T cells are required for EDP1503 efficacy as mice depleted of CD8⁺ T cells are unable to mount a productive anti-tumor response upon treatment with EDP1503.

Conclusions

- Oral delivery of EDP1503 is effective in multiple tumor models (CT26, B16-F10, and MC38).
- EDP1503 induces a proinflammatory gene signature (CXCL10, IL12p70, IFNγ, IL-1β, TNFα and IL-6).
- EDP1503-treated monocyte-conditioned sups induces NK cell cytolytic activity
- CD44⁺ effector lymphocyte populations (NK, NKT, CD4⁺, CD8⁺) upregulate CD25 and IFNγ upon stimulation with EDP1503.
- EDP1503 induces a proinflammatory chemotactic signature within the TME.
- NK and CD8⁺ TILs display an activated phenotype post-EDP1503 treatment.
- Both NK and CD8⁺ T cells are required for a productive anti-tumor response induced by EDP1503. Moreover, NK cells are insufficient to mediate protection in the absence of CD8⁺ cytotoxic T lymphocytes.

Selected References

1. Sivan A., *et al*., Commensal Bifidobacterium promotes antitumor immunity and facilitates anti-PD-L1 efficacy. Science. 2015 Nov 27;350(6264):1084-9.
2. Routy B., *et al*., Gut microbiome influences efficacy of PD-1-based immunotherapy against epithelial tumors. Science. 2018 Jan 5;359(6371):91-97.
3. Gopalakrishnan V., *et al*., Gut microbiome modulates response to anti-PD-1 immunotherapy in melanoma patients. Science. 2018 Jan 5;359(6371):97-103.
4. Matson V., *et al*., The commensal microbiome is associated with anti-PD-1 efficacy in metastatic melanoma patients. Science. 2018 Jan 5;359(6371):104-108.

EVELO