Orally-administered EDP1815, a single strain of *Prevotella histicola*, has potent systemic anti-inflammatory effects in Type 1, Type 2, and Type 3 inflammatory models

Andrea Itano, Taylor Cormack, Kritika Ramani, Kristie Barth, Iris Wang, Ayonika Mukherjee, Holly Ponichtera, Collin McKenna, Mehmedalija Jahic, Mark Bodmer
Evelo Biosciences, Cambridge, MA, USA

INTRODUCTION

The small intestine plays a central role in governing the body's immune, metabolic and neurological systems. Cells in the small intestine, including epithelial and dendritic cells, continuously sample the contents of the lumen. Depending on what they detect, these cells can change their activation states in the small intestinal mucosa. These cells can in turn traffic around the body via immune networks, and subsequently modulate inflammation in sites distal to the gut.

EDP1815 is a pharmaceutical preparation of a single strain of *Prevotella histicola* from the duodenum of a human donor. Given orally, it has luminally-restricted action in the small intestine that leads to systemic anti-inflammatory effects. It was tested in preclinical mouse models of TH1 (Type 1), TH2 (Type 2), and TH17 (Type 3) T cell-mediated inflammation. In these models, EDP1815 treatment resulted in robust inhibition of tissue inflammation, with concomitant reduction in disease-related cytokines.

Since EDP1815 is non-colonizing and non-viable its pharmacological effect is mediated by direct interaction with cells in the small intestine, obviating the need to modify the microbiome. We are investigating whether systemic T-cell mediated inflammatory diseases such as psoriasis may be treated with orally-administered agents which harness this effect, acting luminally in the small intestine without systemic exposure.

RESULTS

EDP1815 enhances IL-10 and IL-27 cytokine production by human inflammatory M1-type APCs

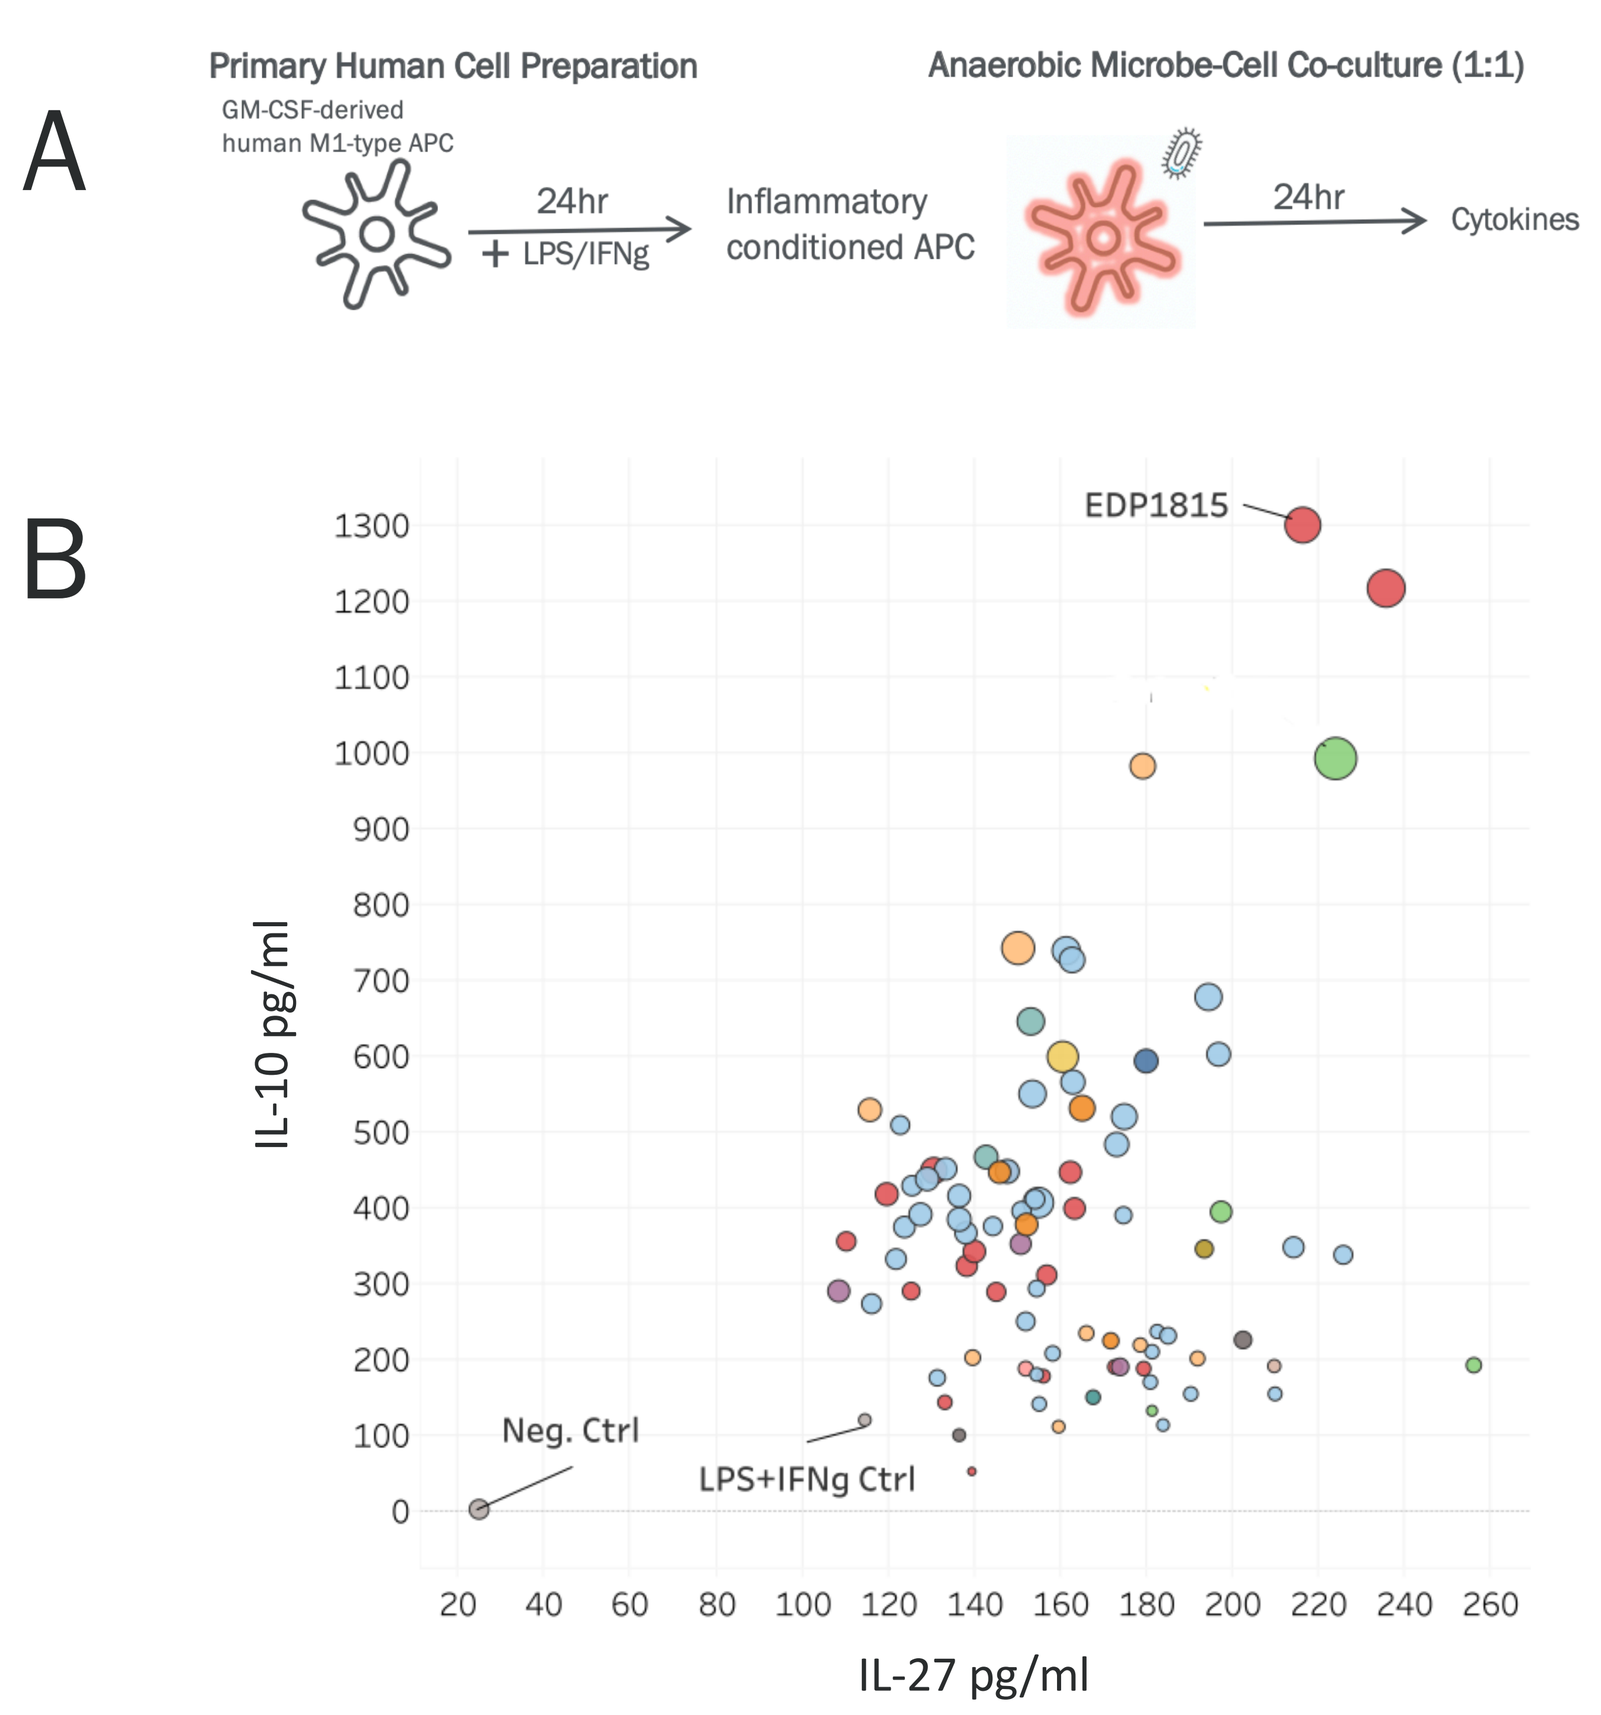
A
Primary Human Cell Preparation
GM-CSF-derived human M1-type APC
24hr
+ LPS/IFNg
Inflammatory conditioned APC
Anaerobic Microbe-Cell Co-culture (1:1)
24hr
Cytokines
B
EDP1815
Neg. Ctrl
LPS+IFNg Ctrl
IL-10 pg/ml
IL-27 pg/ml

Figure 1. (A) Primary human cell assay. Human, CD14+ PBMCs are grown in GM-CSF to induce an M1-type pro-inflammatory phenotype. Cells are then activated for 24 hrs with LPS + IFNg. Cells are incubated with individual strains of microbes for 24 hrs, after which cytokines in the supernatant are measured by Luminex.
(B) Eighty-eight obligate anaerobes were tested in the primary human cell assay. Each point represents the average value from 3 individual healthy donors. Each color represents an individual family of microbes. The size of the circle represents the IL-10/TNF ratio. EDP1815 induced high amounts of anti-inflammatory cytokine IL-10 and IL-27 from M1-type skewed macrophages. These data also demonstrate that each strain has a unique cytokine profile, and that taxonomy is not a guide to function.

EDP1815 is efficacious in a model of peripheral TH1-mediated inflammation (KLH DTH)

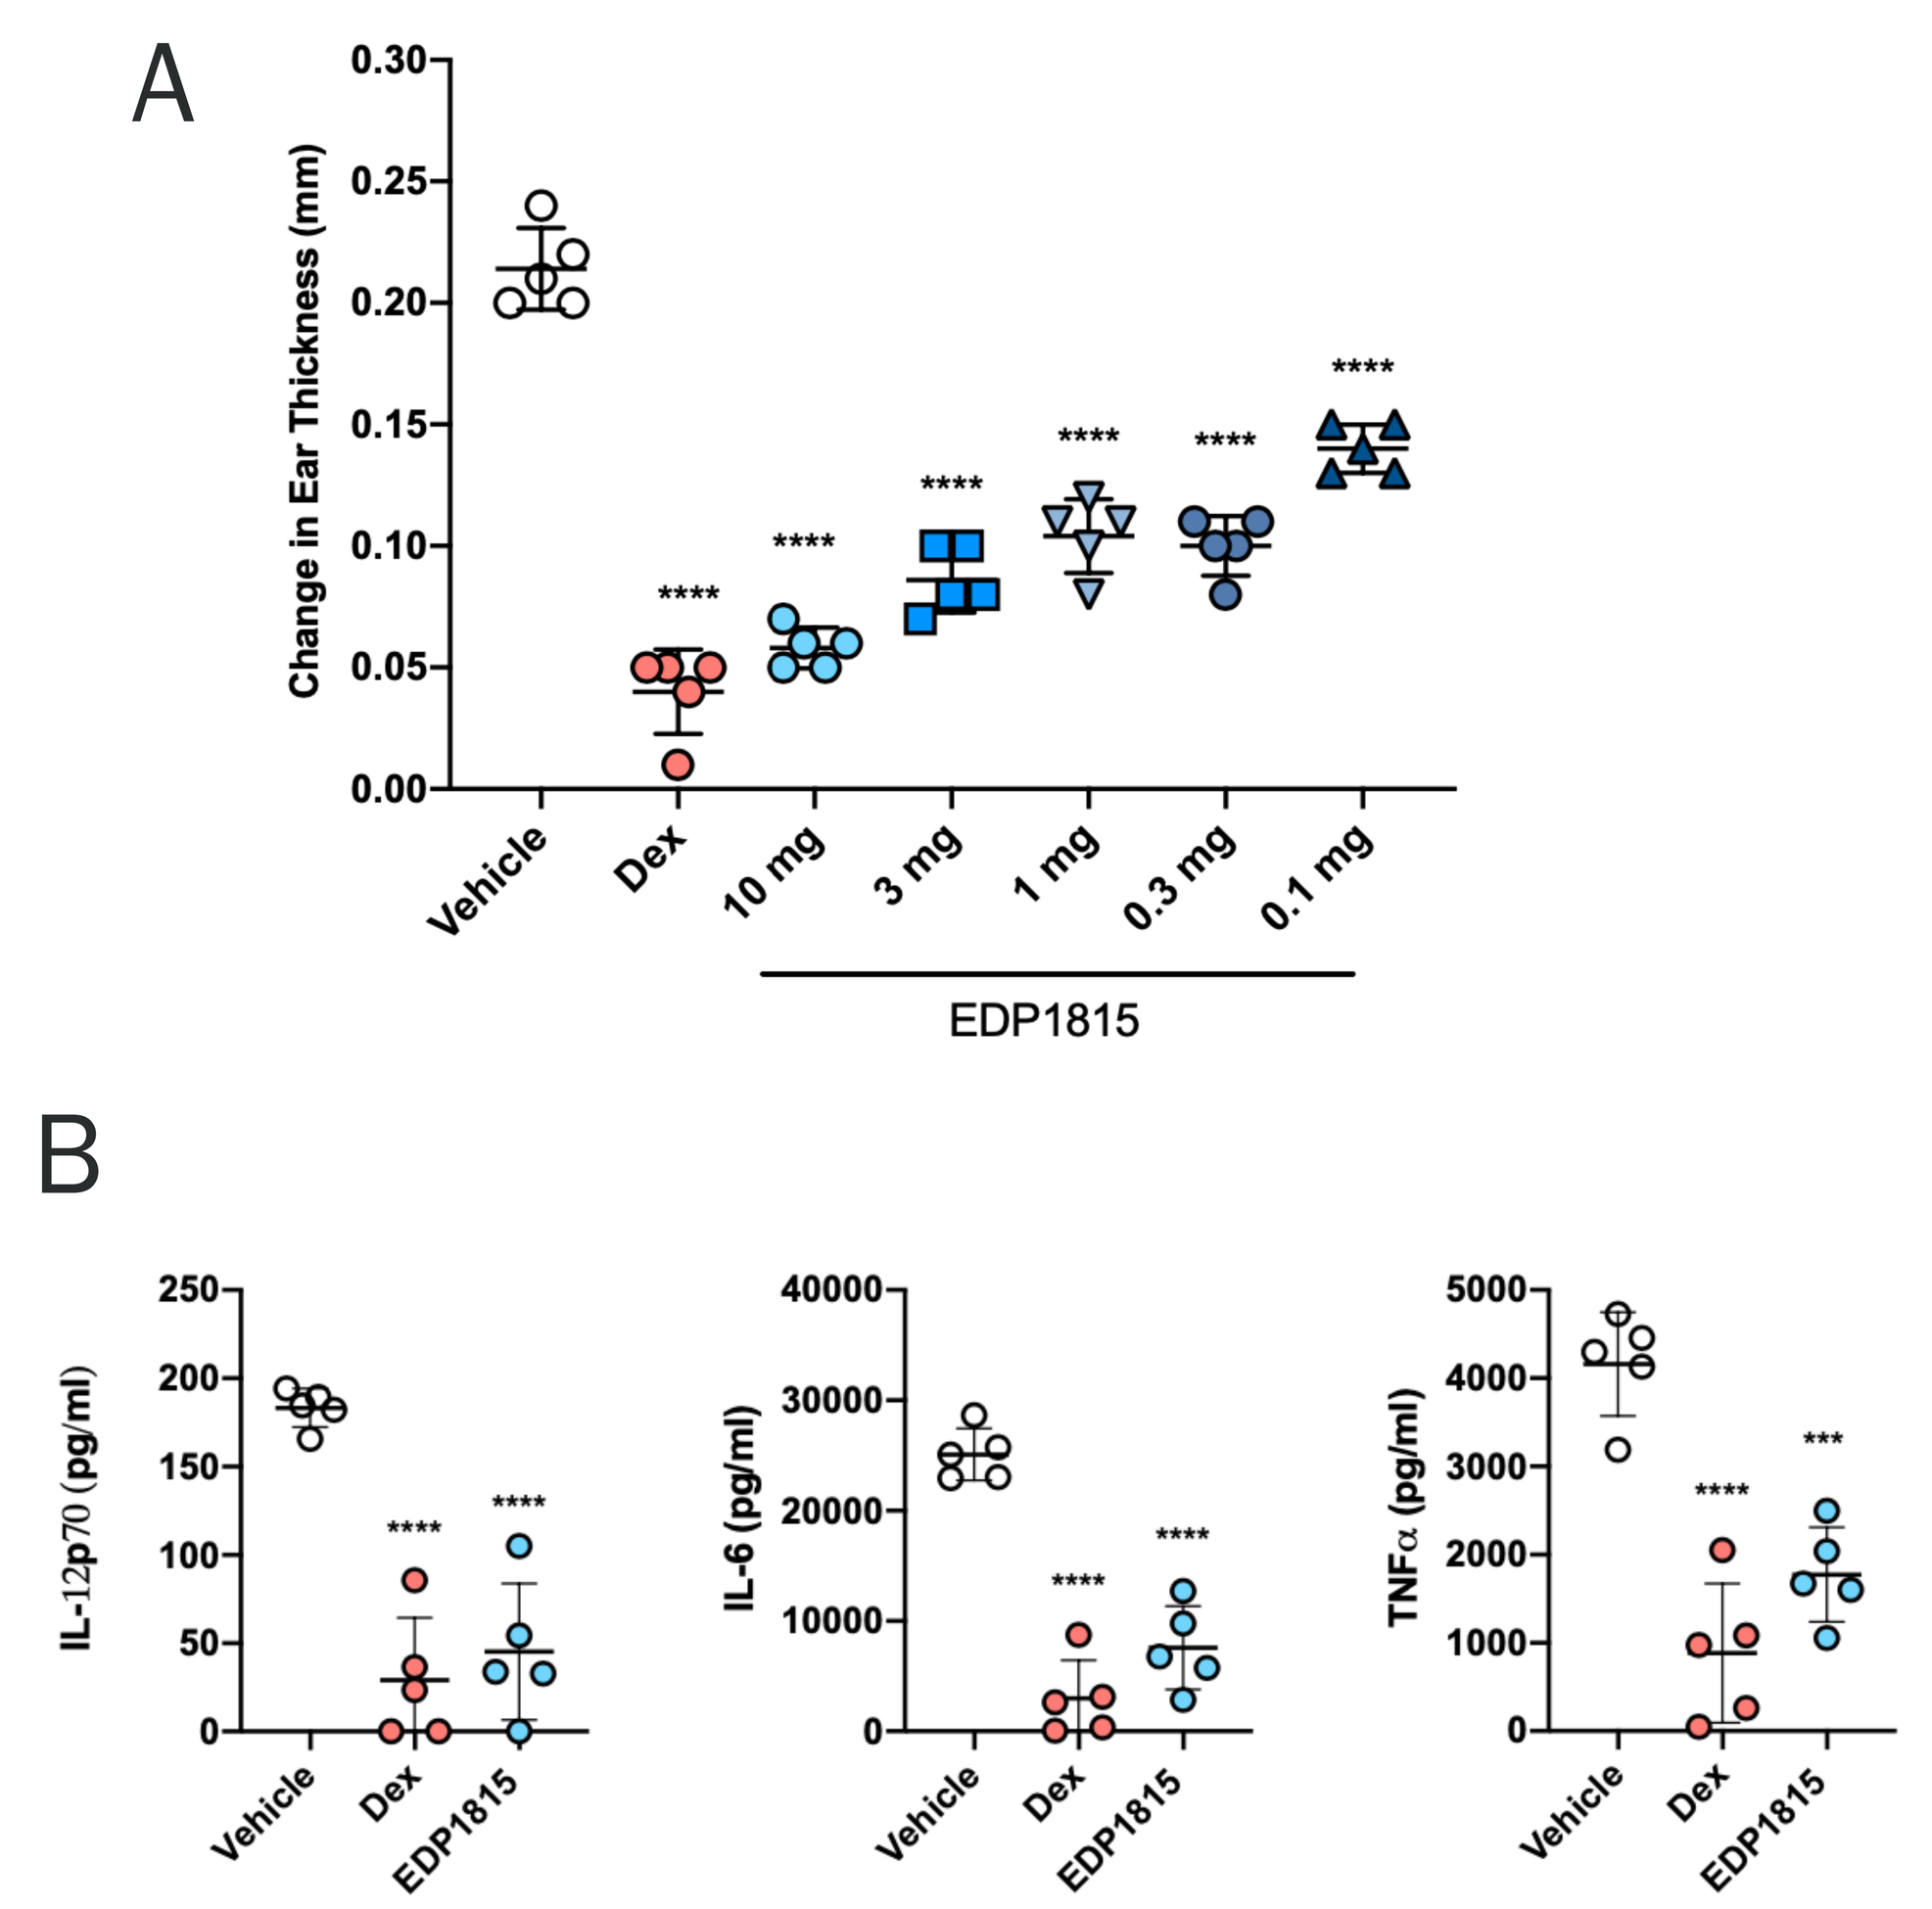
A
Change in Ear Thickness (mm)
Vehicle
Dex
10 mg
3 mg
1 mg
0.3 mg
0.1 mg
EDP1815
B
IL-12p70 (pg/ml)
IL-6 (pg/ml)
TNFα (pg/ml)
Vehicle
Dex
EDP1815

Figure 2. Delayed-type hypersensitivity (DTH) response to Keyhole Limpet Hemocyanin (KLH). C57Bl/6 mice were immunized with KLH and CFA and challenged intradermally in the ear 9 days later with KLH. Mice were treated from the day after immunization through ear challenge with placebo, dexamethasone (1 mg/kg IP QD), or EDP1815 (PO QD). (A) Ear inflammation was measured on day 9. (B) Ex vivo stimulation of draining lymph node or spleen cells with LPS. At the end of the DTH study, mice were sacrificed and total cells from ear draining lymph nodes were incubated with LPS for 2 days. Cytokines from supernatants were measured by MSD.

Oral treatment of EDP1815 is efficacious in an IL-17-driven model of psoriasis (Imiquimod)

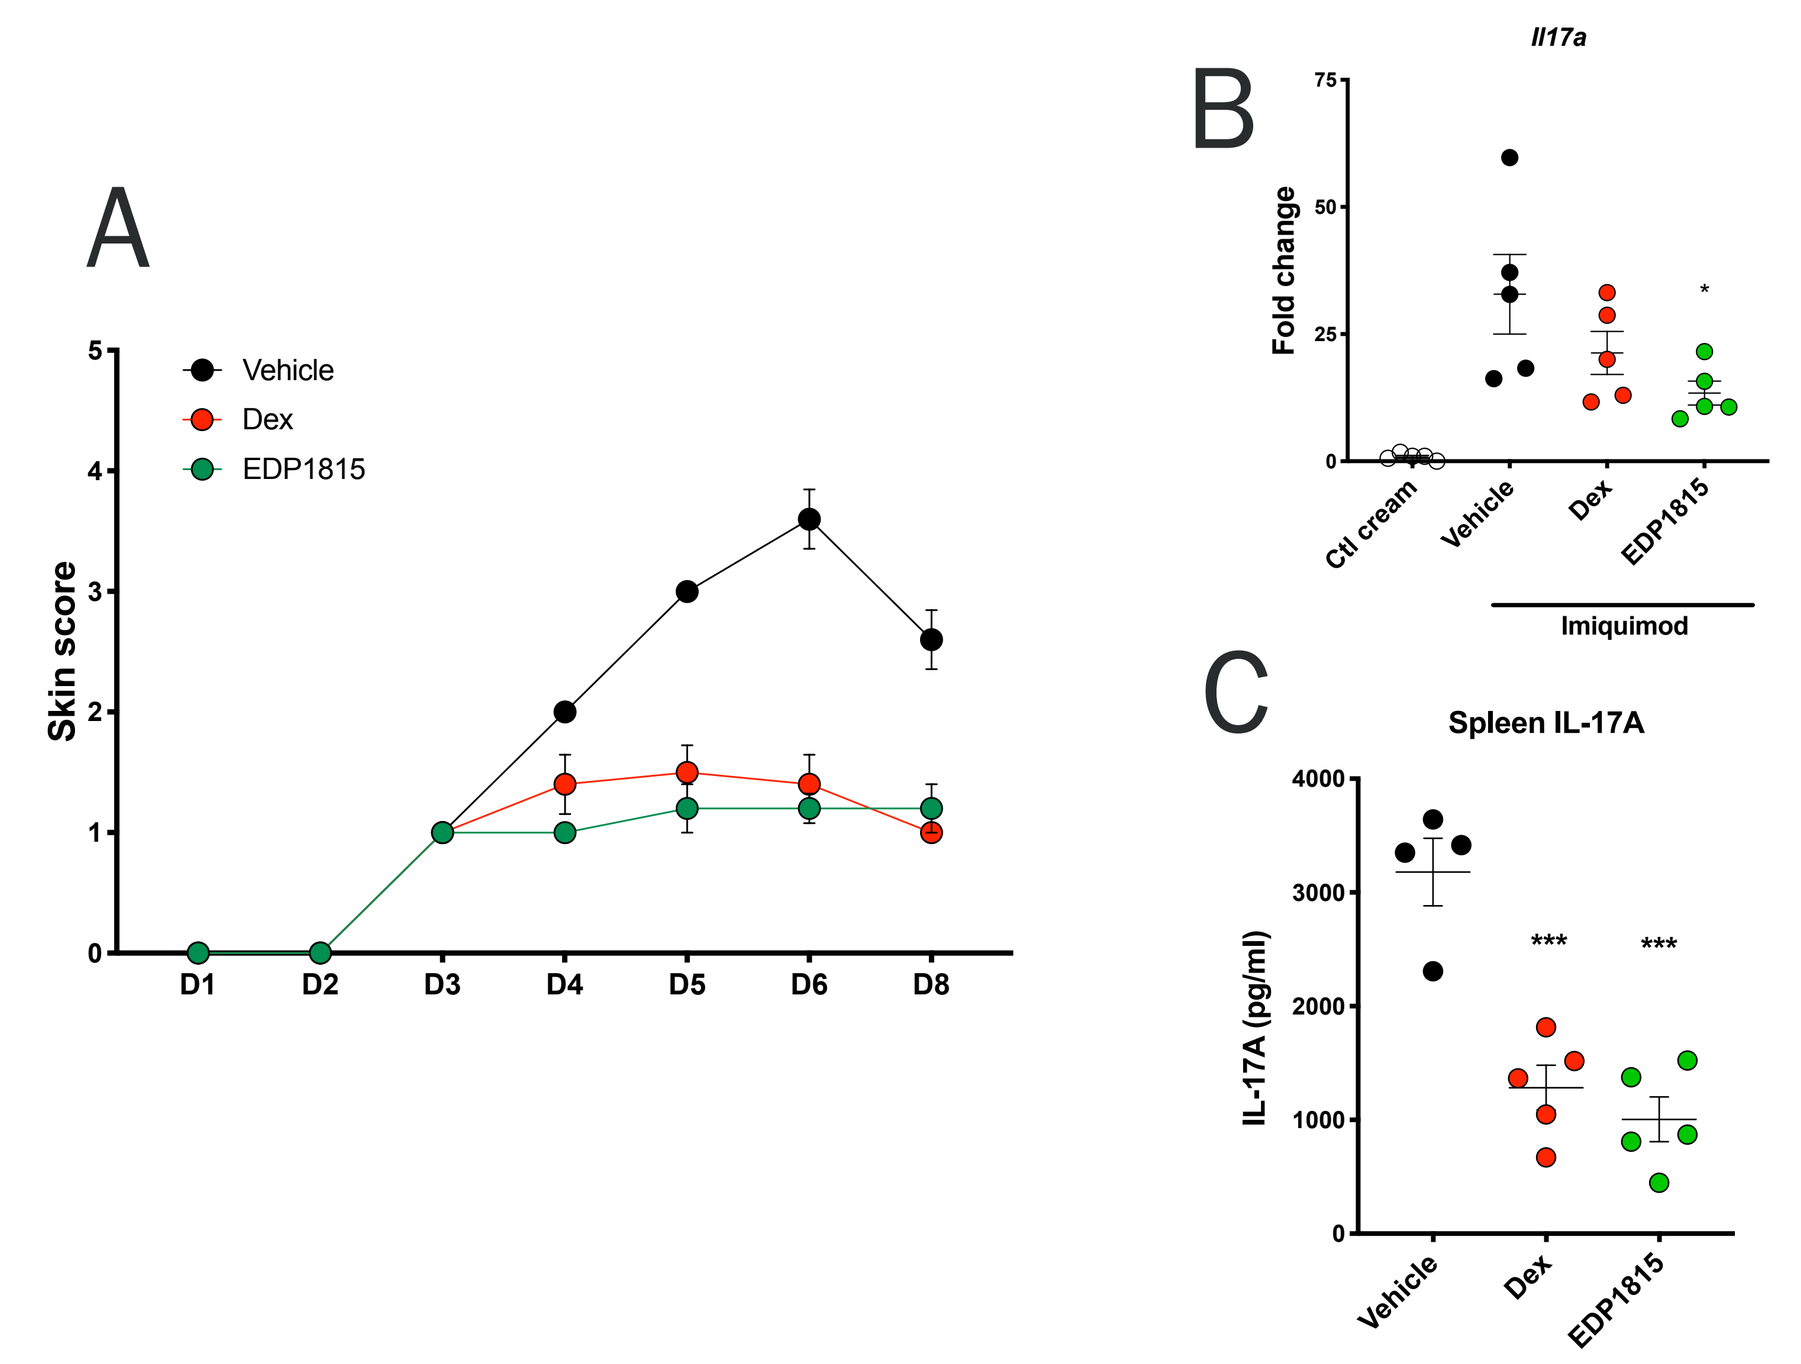
A
Vehicle
Dex
EDP1815
Skin score
D1
D2
D3
D4
D5
D6
D8
B
Il17a
Fold change
Ctl cream
Vehicle
Dex
EDP1815
Imiquimod
C
Spleen IL-17A
IL-17A (pg/ml)
Vehicle
Dex
EDP1815

Figure 3. Imiquimod driven psoriasis mouse model. BALB/c mice were topically treated with 5% imiquimod, a TLR7 and TLR8 agonist for 7 days on the back skin and ear. Mice were treated daily from day 1 through 7 with placebo, dexamethasone (1mg/kg IP) or EDP1815 (10mg PO). (A) Back scores were recorded daily to measure erythema and scaling associated with psoriasis. (B) *Il17a* mRNA transcripts from the psoriatic skin of the mice were measured by RT-qPCR (C) Ex vivo stimulation of splenocytes. At termination of the study, mice were sacrificed and splenocytes were stimulated with PMA/Ionomycin for 48hrs. IL-17A was measured from supernatants by MSD.

EDP1815 reduces inflammation and cytokine production in the skin in the MC903 model of TH2-mediated allergic dermatitis

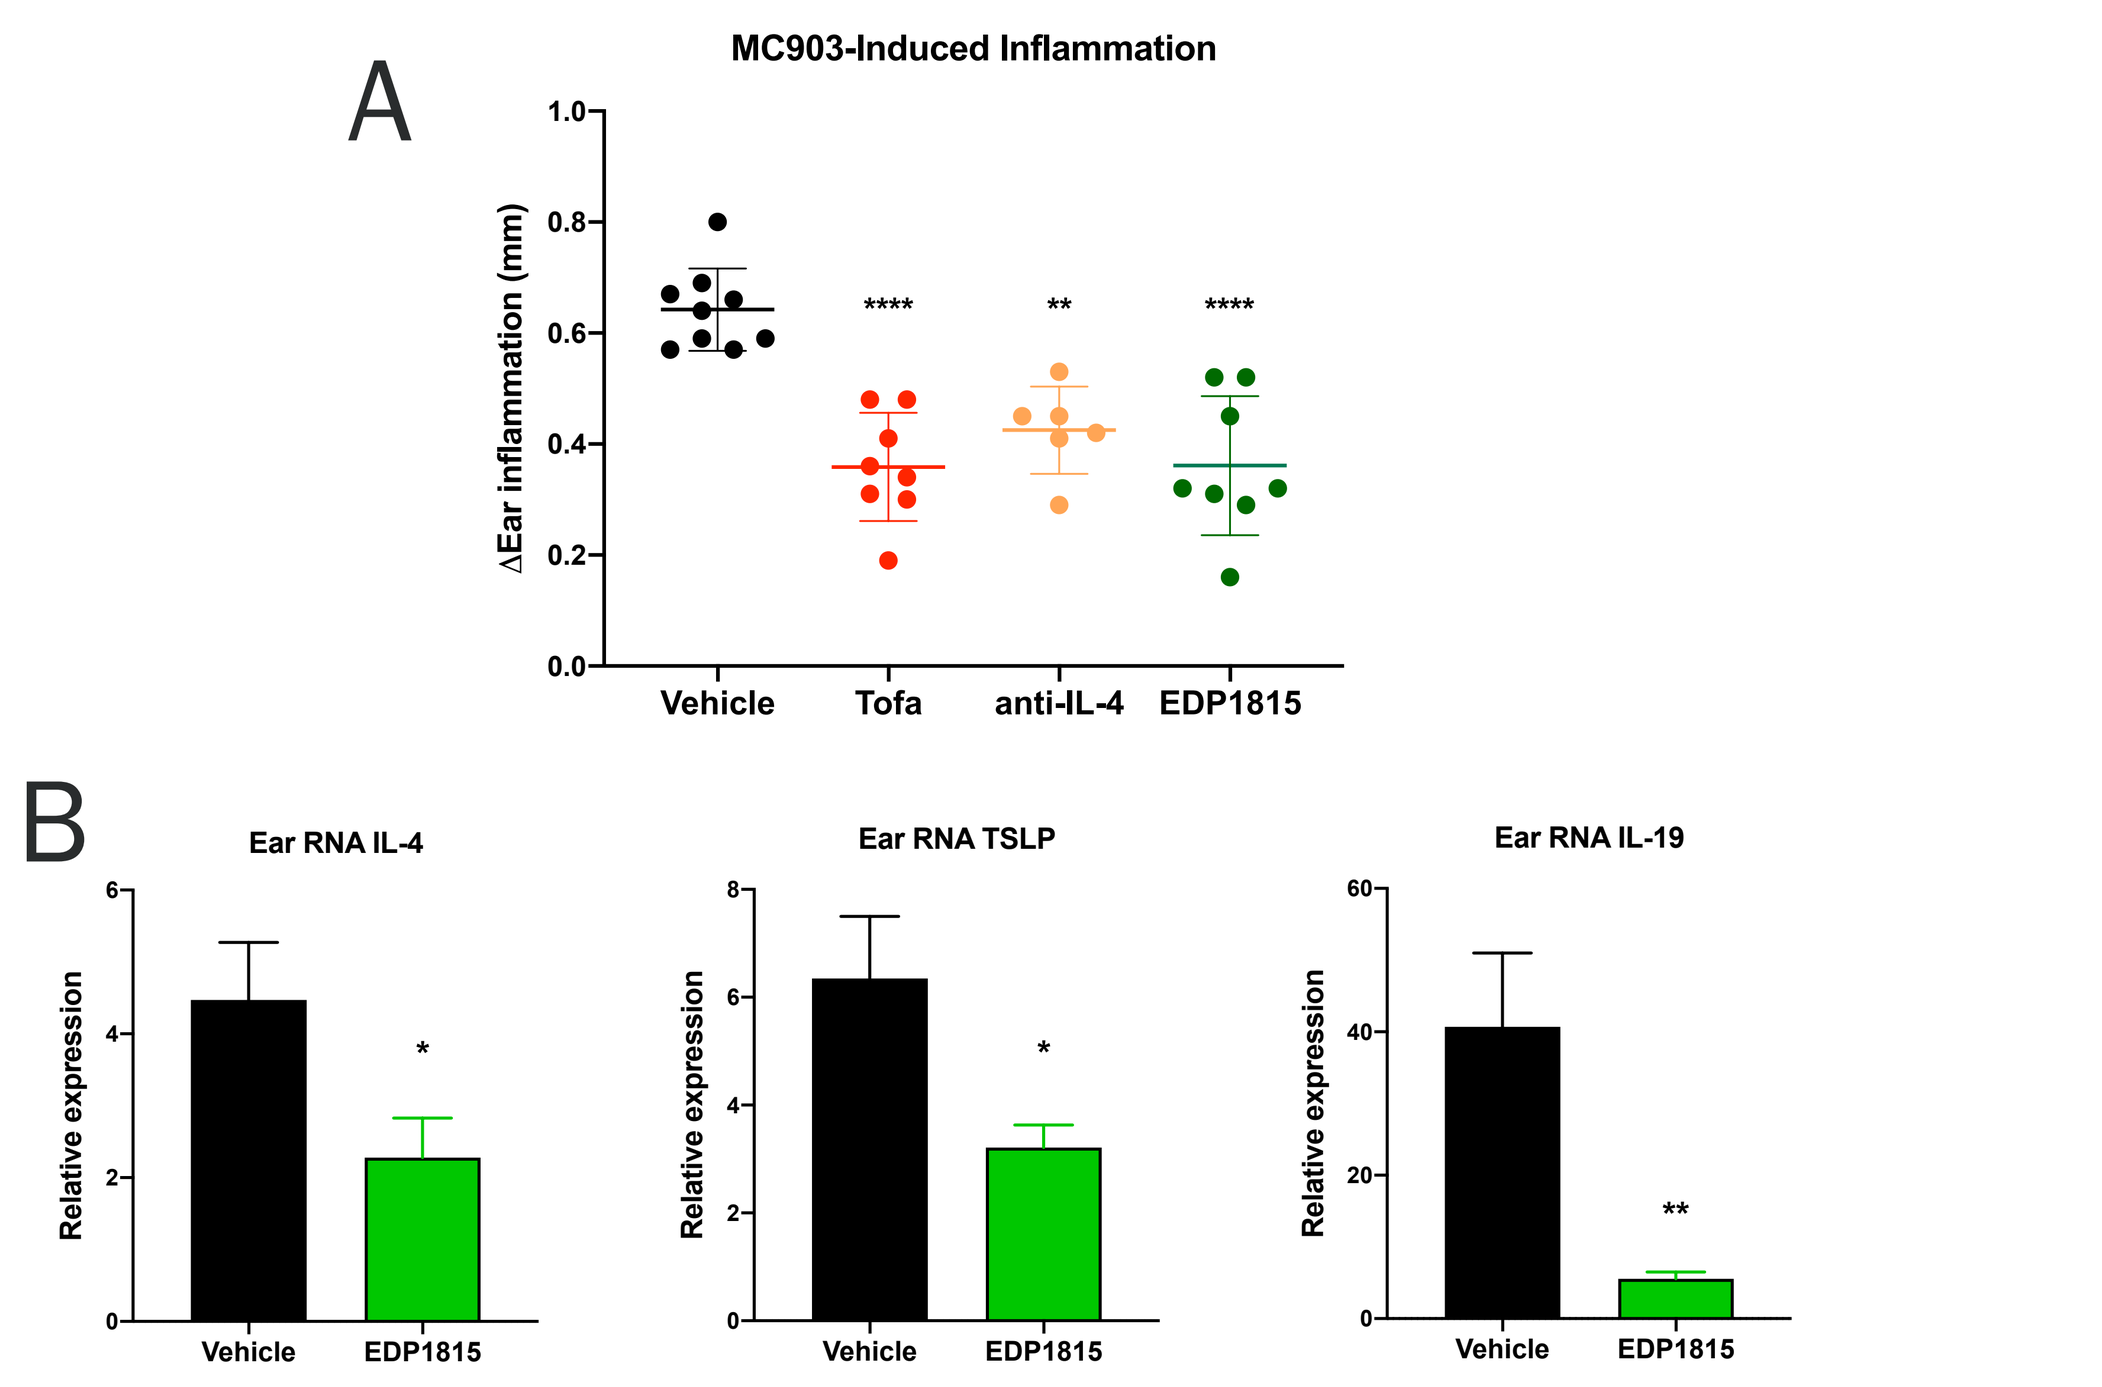
A
MC903-Induced Inflammation
ΔEar inflammation (mm)
Vehicle
Tofa
anti-IL-4
EDP1815
B
Ear RNA IL-4
Ear RNA TSLP
Ear RNA IL-19
Relative expression
Vehicle
EDP1815

Figure 4. MC903 model of allergic dermatitis. Mice were sensitized daily for 14 consecutive days with 2 nmol of MC903 (calcipotriol) in 20 uL of 100% EtOH on ears. Mice were gavaged daily from Day 1 to Day 6 with 10 mg of EDP1815, vehicle, or tofacitinib (20 mg/kg). Mice were treated with tofacitinib daily (20 mg/kg PO daily) or an anti-IL-4 antibody (200 ug/mouse IP every 3 days) for comparison. Baseline ear measurements were taken prior to the first ear sensitization on day 1. On day 14, ear thickness was measured. (A) Change in ear thickness was expressed as ear thickness at day 14 minus ear thickness at baseline. (B) Expression of IL-4, TSLP, and IL-19 from ear tissues was measured by qPCR.

Oral treatment of EDP1815 inhibits central nervous system inflammation in a TH17-driven EAE model

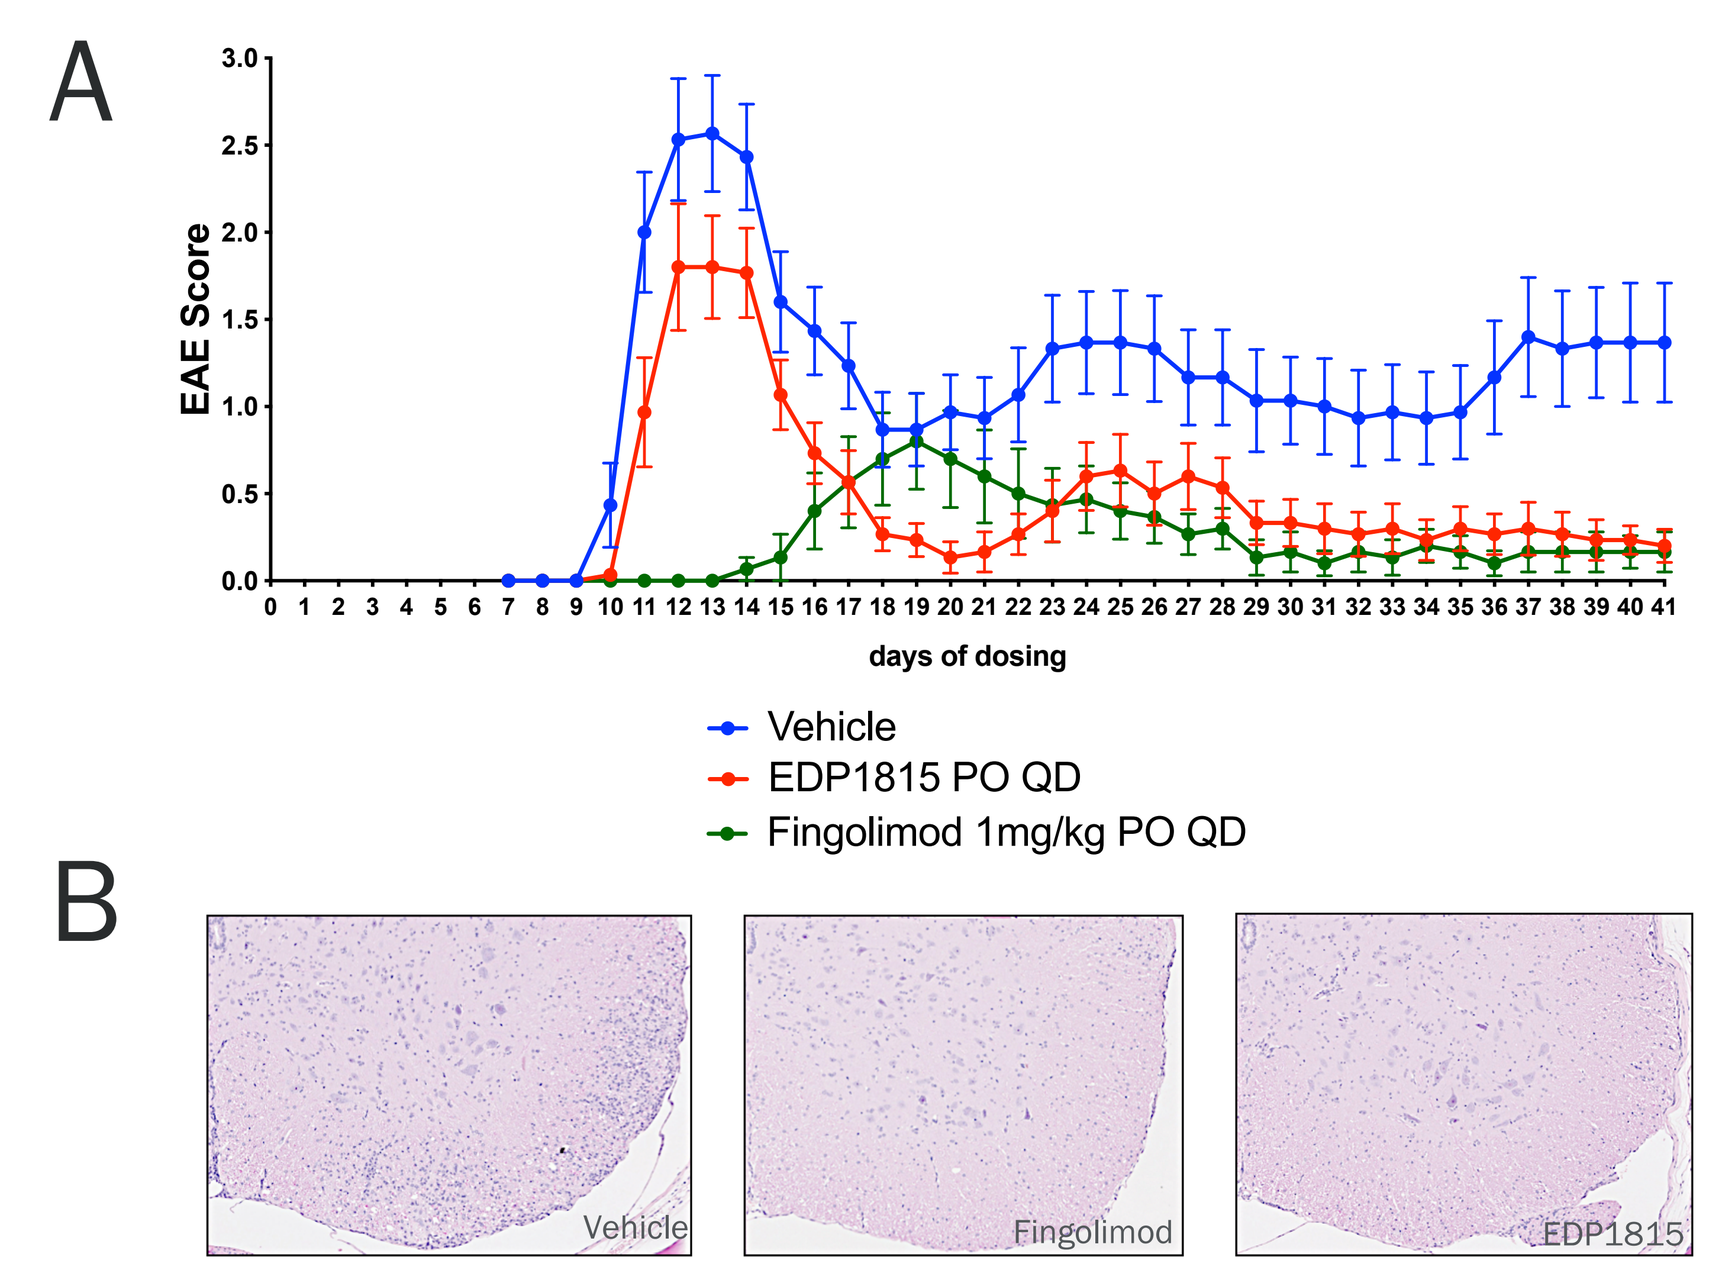
A
EAE Score
days of dosing
Vehicle
EDP1815 PO QD
Fingolimod 1mg/kg PO QD
B
Vehicle
Fingolimod
EDP1815

Figure 5. PLP/SJL model of relapsing-remitting experimental autoimmune encephalomyelitis (EAE). SJL mice were immunized with PLP peptide and CFA on Day 0. Mice were dosed daily from day of immunization. (A) Clinical score was read daily from day 7 - 41, on a scale from 0 to 4, with 0 being no obvious changes in motor function, and 4 being limp tail, complete hind leg and partial front leg paralysis. (B) Histopathology of spinal cord taken on day 41.

CONCLUSIONS

Orally administered EDP1815 is luminally-active with anti-inflammatory effects in murine models of TH1-, TH2-, and TH17-mediated inflammation, including a model of chronic central nervous system inflammation. There are two main conclusions from this work. The first is that the intestinal mucosa is a central controller of systemic inflammation, operating at a distance in response to signals generated by substances passing through the gut. The effects appear to represent broad-based resolution of inflammation that re-establishes normal homeostatic inflammatory status across multiple pathways. The second is that resolution of inflammation by a non-absorbed oral agent acting via the small intestinal axis has the potential to create a new class of effective, safe, oral medicines which can be manufactured at reasonable cost for the treatment of inflammatory diseases suffered by millions of patients around the globe. These results support a series of clinical studies currently under way with EDP1815. Results from the first clinical study of EDP1815 will be described in oral presentation "FC02.05 Safety and efficacy of an orally administered, single strain commensal microbe in psoriasis after 28 days of therapy: EDP1815".

EVELO