EDP1503 induces antitumor responses via gut-mediated activation of both innate and adaptive immunity

P-325

Loise Francisco-Anderson PhD[1], Shamira Shariffudin[1], Humphrey Gardner MD[1], Michael Goldberg PhD[1], Shubhra Kashyap MS[1], Mary Abdou[1], Chris Davitt PhD[1], Shannon Argueta PhD[1], Pooja Parameswaran MS[1], Peter Sandy PhD[1], Holly Ponichtera PhD[1], Mark Carlson[1], Maria Sizova PhD[1], Valeria Kravitz[1], Erin Troy PhD[1], Sam Andrewes MS[1], Johanna C. Bendell MD[2,3], Judy S. Wang MD[2,4], Susanna V. Ulahannan MD[2,5], Michael Chisamore PhD[6], Mark Bodmer PhD[1], and Duncan McHale MD[1]

[1]Evelo Biosciences, Cambridge MA USA; [2]GI Cancer Research Program, Sarah Cannon Research Institute, Nashville, TN, USA, [3]Dept of Medical Oncology, Tennessee Oncology, Nashville, TN, USA; [4]Dept of Medical Oncology, Drug Dev Unit, Florida Cancer Specialists, Sarasota, FL, USA; [5]Dept of Medical Oncology & Hematology, Stephenson Cancer Center, University of Oklahoma Health Sciences Center, Oklahoma City, OK, USA; [6]Oncology Clinical Research, Merck & Co., Inc, Kenilworth, NJ, USA

INTRODUCTION & OBJECTIVES

INTRODUCTION: Systemic immunity is regulated by the small intestine via transient non-colonizing interactions of bacteria and immune cells along its axis, resulting in microbe-dependent modulation of inflammation. EDP1503 is a strain of *Bifidobacterium animalis lactis* that when administered orally, demonstrates effective preclinical anti-tumor responses due to activation of innate and adaptive immune mechanisms. The pleiotropic effects of EDP1503 are driven through interactions with multiple pattern recognition receptors inducing a proinflammatory signature in human PBMCs and activating CD8 and NK cell IFNγ production and cytolytic activity.

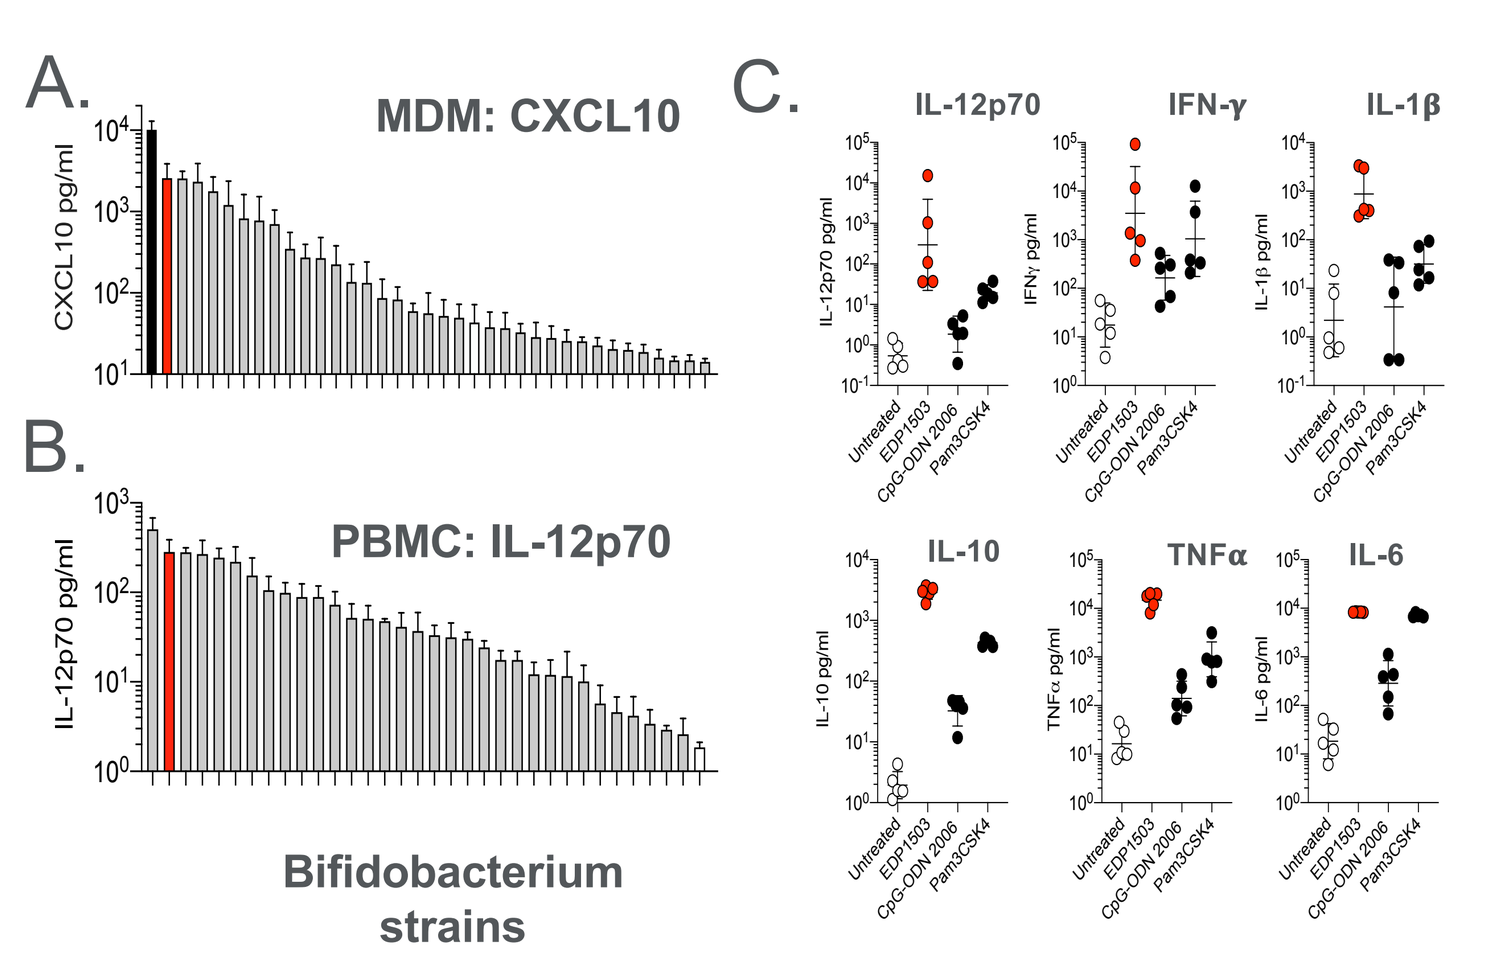
A.
MDM: CXCL10
B.
PBMC: IL-12p70
Bifidobacterium strains
C.
IL-12p70
IFN-γ
IL-1β
IL-10
TNFα
IL-6

Fig 1. EDP1503 Stimulates Type I Immune Responses
(A) Human monocyte-derived macrophages stimulated with EDP1503 (shown in red) *in vitro* lead to the strong induction of CXCL10 compared to other strains of *Bifidobacterium* (M1 polarized macrophages shown in black). **(B)** Similarly, human PBMCs stimulated with EDP1503 induced significant IL-12p70 in comparison to other *Bifidobacterium.* **(C)** EDP1503 stimulates the PBMC production of a pro-inflammatory cytokine signature significantly greater than individual TLR2 or TLR9 agonists (N = 5 donors).

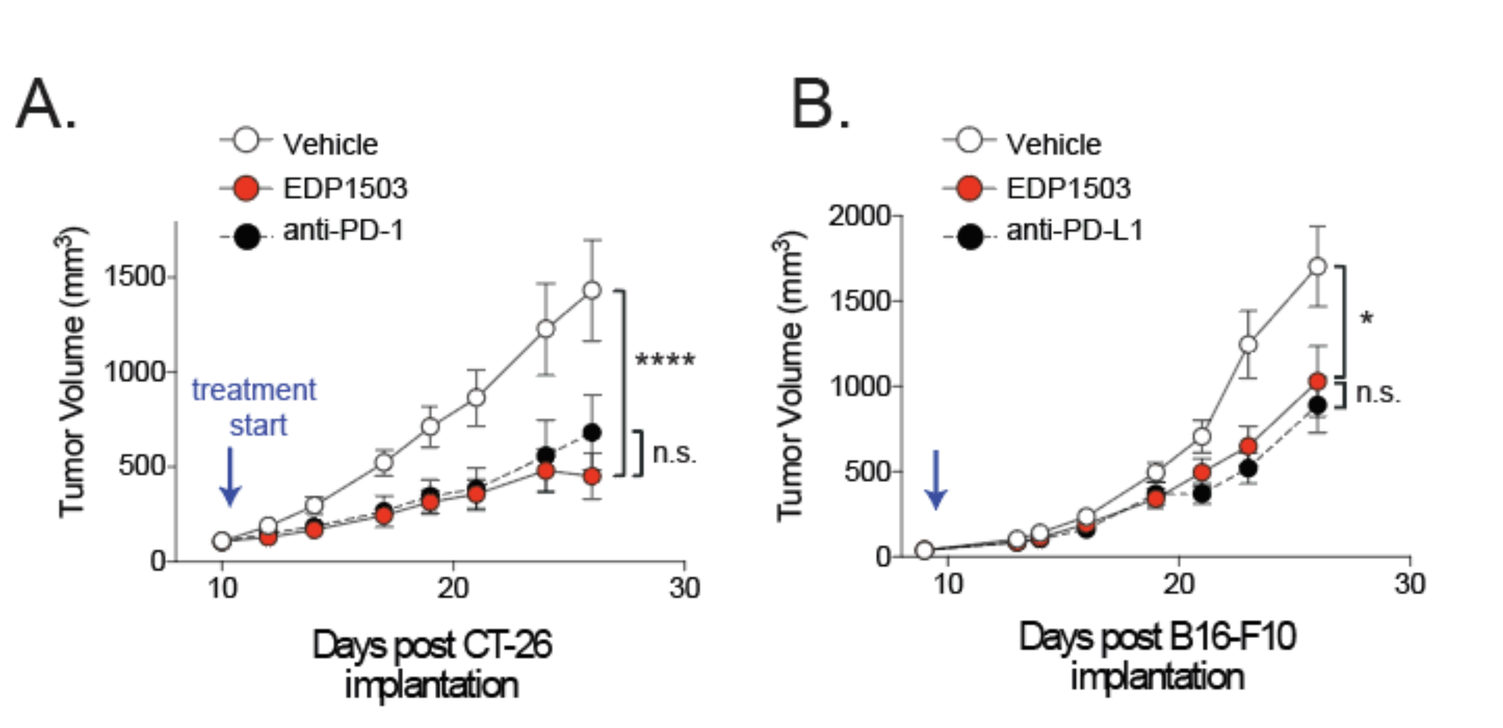
A.
Vehicle
EDP1503
anti-PD-1
Tumor Volume (mm³)
treatment start
Days post CT-26 implantation
B.
Vehicle
EDP1503
anti-PD-L1
Days post B16-F10 implantation

Fig 2. Oral delivery of EDP1503 is as effective as anti-PD-1 in multiple tumor models *in vivo*
Oral delivery of EDP1503 monotherapy leads to tumor control comparable to checkpoint inhibition in multiple subcutaneous syngeneic tumor models. **(A)** Syngeneic mice were inoculated with CT26 or **(B)** B16-F10 tumor cells at 10^5 subcutaneously. Tumors were established for 10 days (~100mm³) prior to initiation of treatment (indicated by blue arrow). Mice were treated with either sucrose vehicle or EDP1503 via oral gavage daily or treated with anti-PD-1/ anti-PD-L1 every four days for the duration of the study.

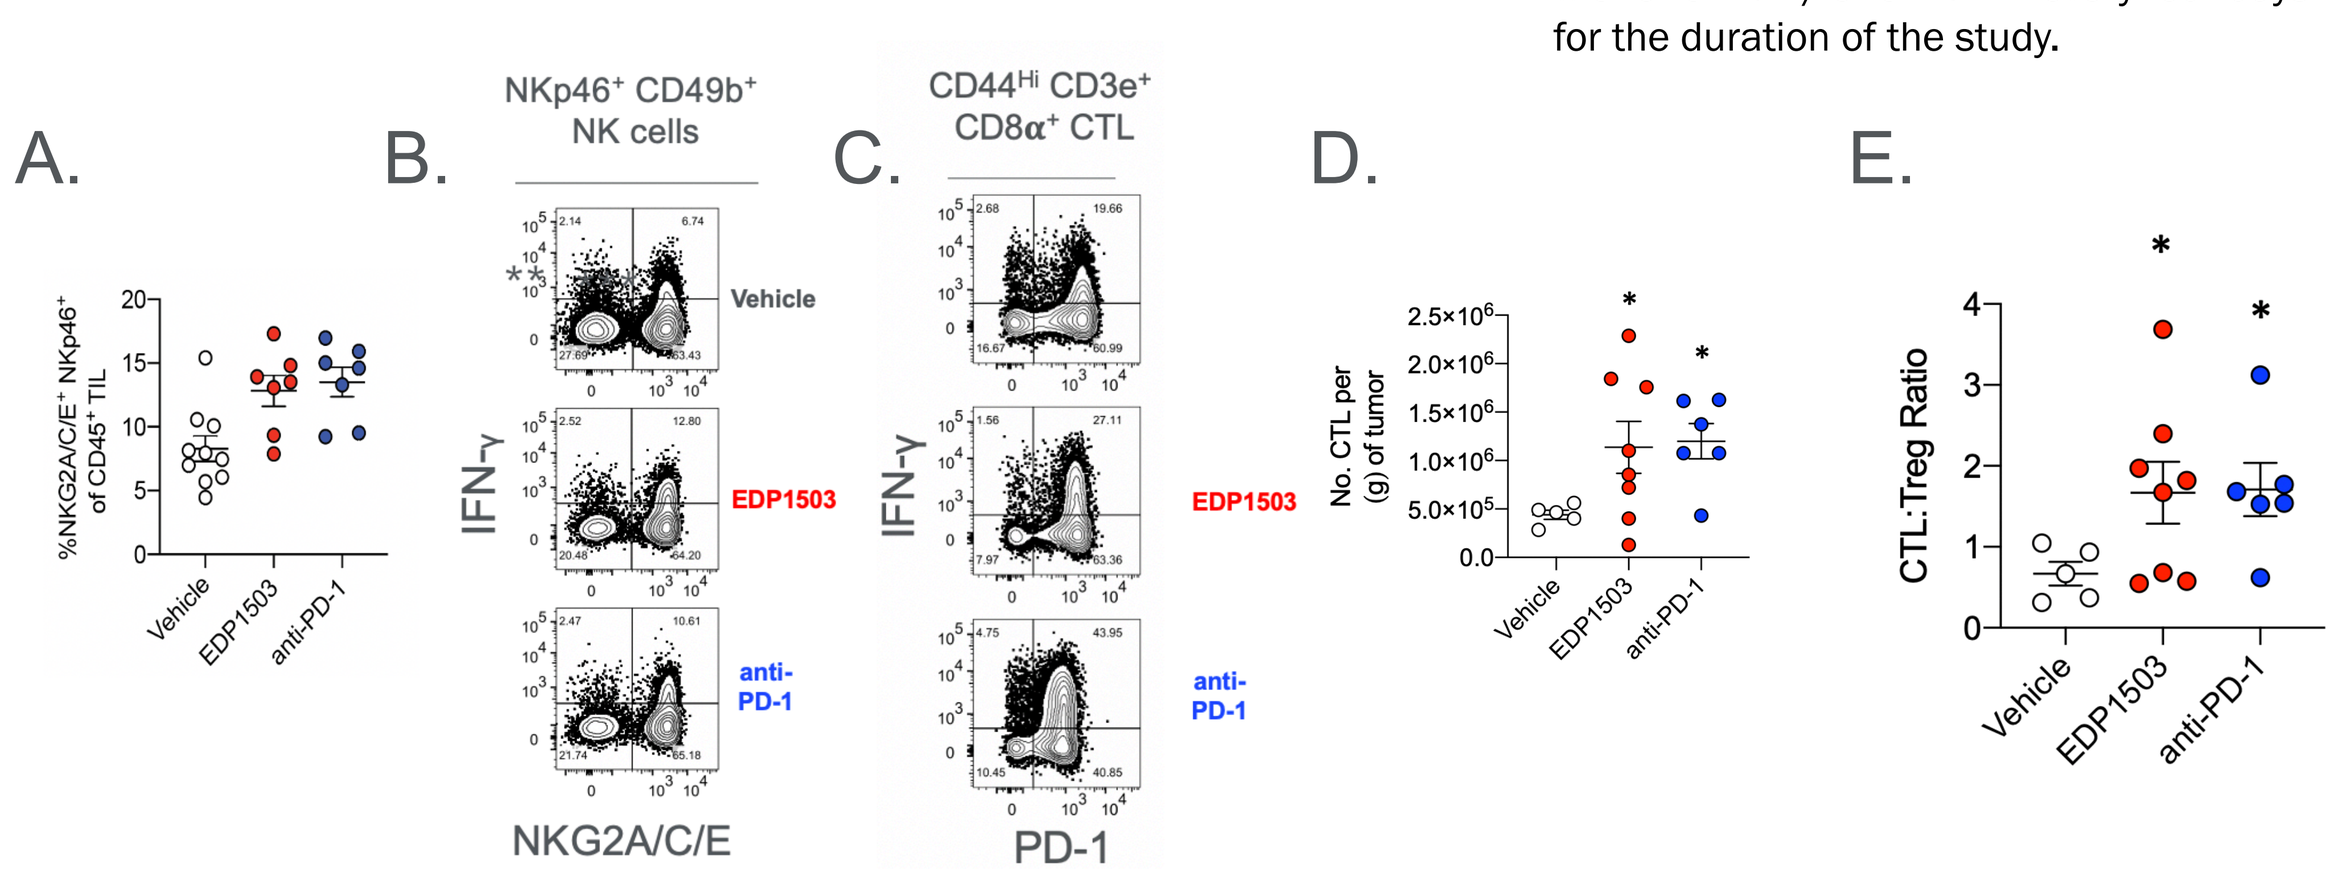
A.
B.
NKp46⁺ CD49b⁺ NK cells
Vehicle
EDP1503
anti-PD-1
NKG2A/C/E
C.
CD44^Hi CD3e⁺ CD8α⁺ CTL
PD-1
D.
E.
CTL:Treg Ratio

Fig 3. EDP1503 activates effector NK and CD8 TILs **(A)** EDP1503 treatment of CT26 bearing mice increased NKp46⁺NKG2A/C/E⁺ NK cells within the TME comparable to anti-PD-1 treatment. **(B)** EDP1503 induces IFNγ production by NKp46⁺NKG2A/C/E⁺ NK cells. **(C)** EDP1503 induces elevated IFNγ production and concomitant upregulation of PD-1 in CD44⁺CD8 CTL within the TME. **(D)** EDP1503 treatment results in increased tumor-infiltrating CTLs in responding tumors, comparable to anti-PD-1; reported as counts of CTL normalized per gram of tumor. **(E)** EDP1503 increases the CD8 CTL to Treg ratio within the TME of responding tumors, comparable to anti-PD-1. Responding tumors were identified as below the median tumor weight for each respective group. Tumors were analyzed following 5 days of EDP1503 treatment by flow cytometry and statistics were obtained using Brown-Forsythe and Welch ANOVA and Holm-Sidak post-hoc analysis.

OBJECTIVES: This study is being conducted to assess the safety, tolerability, and efficacy of EDP1503 alone and in combination with pembrolizumab in patients with advanced metastatic colorectal carcinoma, triple-negative breast cancer, and checkpoint inhibitor-relapsed tumors.

METHODS

CLINICAL TRIAL: A Phase I/II Open-label Study of EDP1503 in Combination With Pembrolizumab in Patients With Advanced Metastatic Colorectal Carcinoma, Triple-negative Breast Cancer, and Checkpoint Inhibitor Relapsed Tumors n=~120 (NCT03775850)

Primary Outcome Measures:

1. Safety and tolerability of EDP1503 in combination with pembrolizumab as assessed per CTCAE v5.0 [Time Frame: 2 years]. Number of participants with EDP1503 related adverse events as assessed per CTCAE v5.0
2. Safety and tolerability of EDP1503 in combination with pembrolizumab [Time Frame: 2 years]. Safety and tolerability of EDP1503 alone and in combination with pembrolizumab assessed via clinical laboratory evaluations
3. Evidence of anti-tumor activity of EDP1503 based on ORR [Time Frame: 2 years]. To determine preliminary evidence of anti-tumor activity of EDP1503 in patients

Study Design

Endpoints
- Safety and tolerability
- Overall response rates
- Biomarkers of immune response in paired tumor biopsies taken before and after 2-week EDP1503 monotherapy

Dosing
- Patients receive a 14-day run-in of EDP1503 monotherapy, following which they are treated with a combination of EDP1503 and pembrolizumab
- EDP1503 is given as 2-4 capsules taken by mouth twice daily; pembrolizumab (200 mg) is administered by intravenous (IV) infusion once every 3 weeks

MSS Colorectal Cancer (N=33)
Triple-negative Breast Cancer (N=11)
Additional tumor types[1] relapsed on prior PD-1/L1 therapy (N=18)

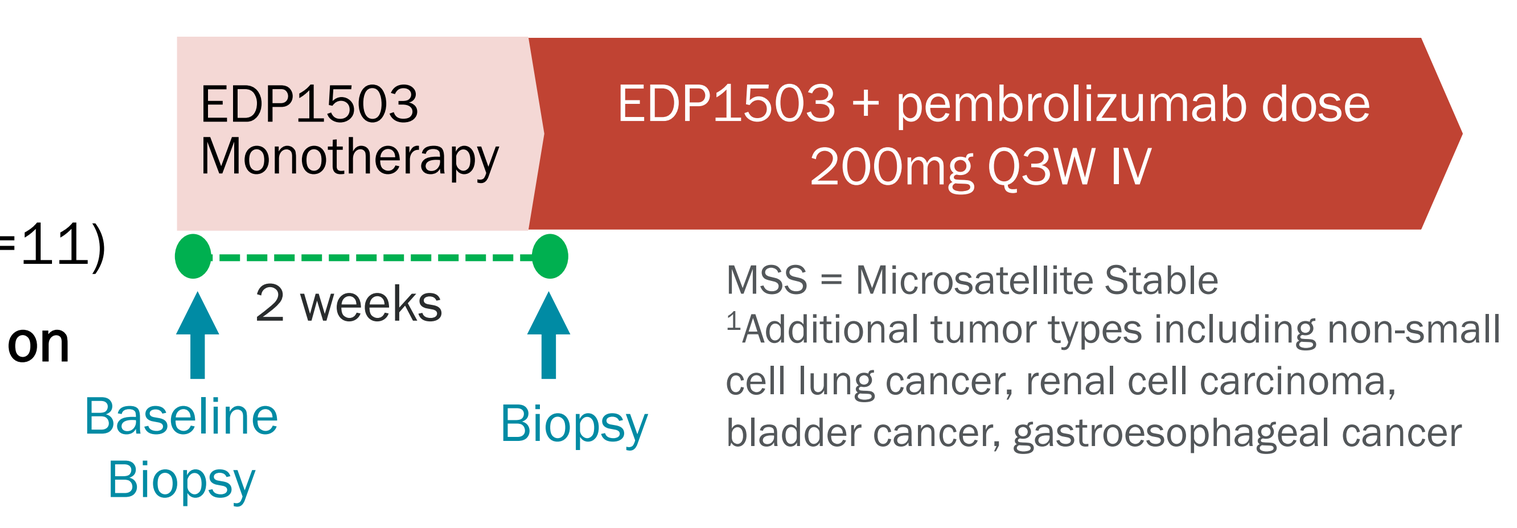
EDP1503 Monotherapy
EDP1503 + pembrolizumab dose 200mg Q3W IV
2 weeks
Baseline Biopsy
Biopsy

MSS = Microsatellite Stable
[1]Additional tumor types including non-small cell lung cancer, renal cell carcinoma, bladder cancer, gastroesophageal cancer

RESULTS

Table 1: Baseline Characteristics

Characteristics	n (%)
Gender	
Male	32 (52)
Female	30 (48)
Median age	61
> 65 years	21 (34)
ECOG	
0	19 (31)
1	43 (69)

Characteristics	n (%)
Tumor type	
CRC	33 (53)
TNBC	11 (18)
NSCLC	11 (18)
RCC	2 (3)
GEJ/Esophageal	2 (3)
Other	3 (5)
Prior anti-PD-(L)1 therapy	22 (35)

Table 2: Treatment-related Adverse Events (TRAE)

Adverse event, n (%)	Related to study treatment: All Gr.	Gr. 1-2	Gr. 3
≥ 1 any TRAE	34 (100)	34 (100)	3 (9)
TRAE led to discontinuation	0	0	0
Grade ≥3 TRAE	3 (9)	0	3 (9)
TRAE led to death	0	0	0
Diarrhea	10 (29)	10 (29)	1 (3)
Fatigue	10 (29)	9 (27)	1 (3)
Abdominal distension	7 (21)	7 (21)	0
Abdominal pain	7 (21)	7 (21)	0
Arthralgia	6 (18)	5 (15)	1 (3)
Flatulence	6 (18)	6 (18)	0
Nausea	6 (18)	6 (18)	0
Decreased appetite	4 (12)	4 (12)	0
Vomiting	4 (12)	4 (12)	0
Constipation	3 (9)	3 (9)	0
Dyspepsia	3 (9)	3 (9)	0
Pruritus	3 (9)	3 (9)	0
Eructation	2 (6)	2 (6)	0
Night sweats	2 (6)	2 (6)	0
Rash maculo-papular	2 (6)	2 (6)	0

Adverse event, n (%)	Related to study treatment: All Gr.	Gr. 1-2	Gr. 3
Abdominal discomfort	1 (3)	1 (3)	0
Arthritis	1 (3)	1 (3)	0
Aspartate aminotransferase increased	1 (3)	1 (3)	0
Blood alkaline phosphatase increased	1 (3)	1 (3)	0
Colitis	1 (3)	1 (3)	0
Dehydration	1 (3)	1 (3)	0
Dyspnea	1 (3)	1 (3)	0
Headache	1 (3)	1 (3)	0
Hypothyroidism	1 (3)	1 (3)	0
Infusion related reaction	1 (3)	1 (3)	0
Myalgia	1 (3)	1 (3)	0
Rectal discharge	1 (3)	1 (3)	0
Skin hyperpigmentation	1 (3)	1 (3)	0
Stomatitis	1 (3)	1 (3)	0
Tumor pain	1 (3)	1 (3)	0

Note: No treatment-related Grade 4-5 AEs or treatment-related serious AEs have been reported.

Fig 4. Microsatellite Stable (MSS) CRC

Duration of Follow-up and Percent Change from Baseline Over Time for Target Lesions

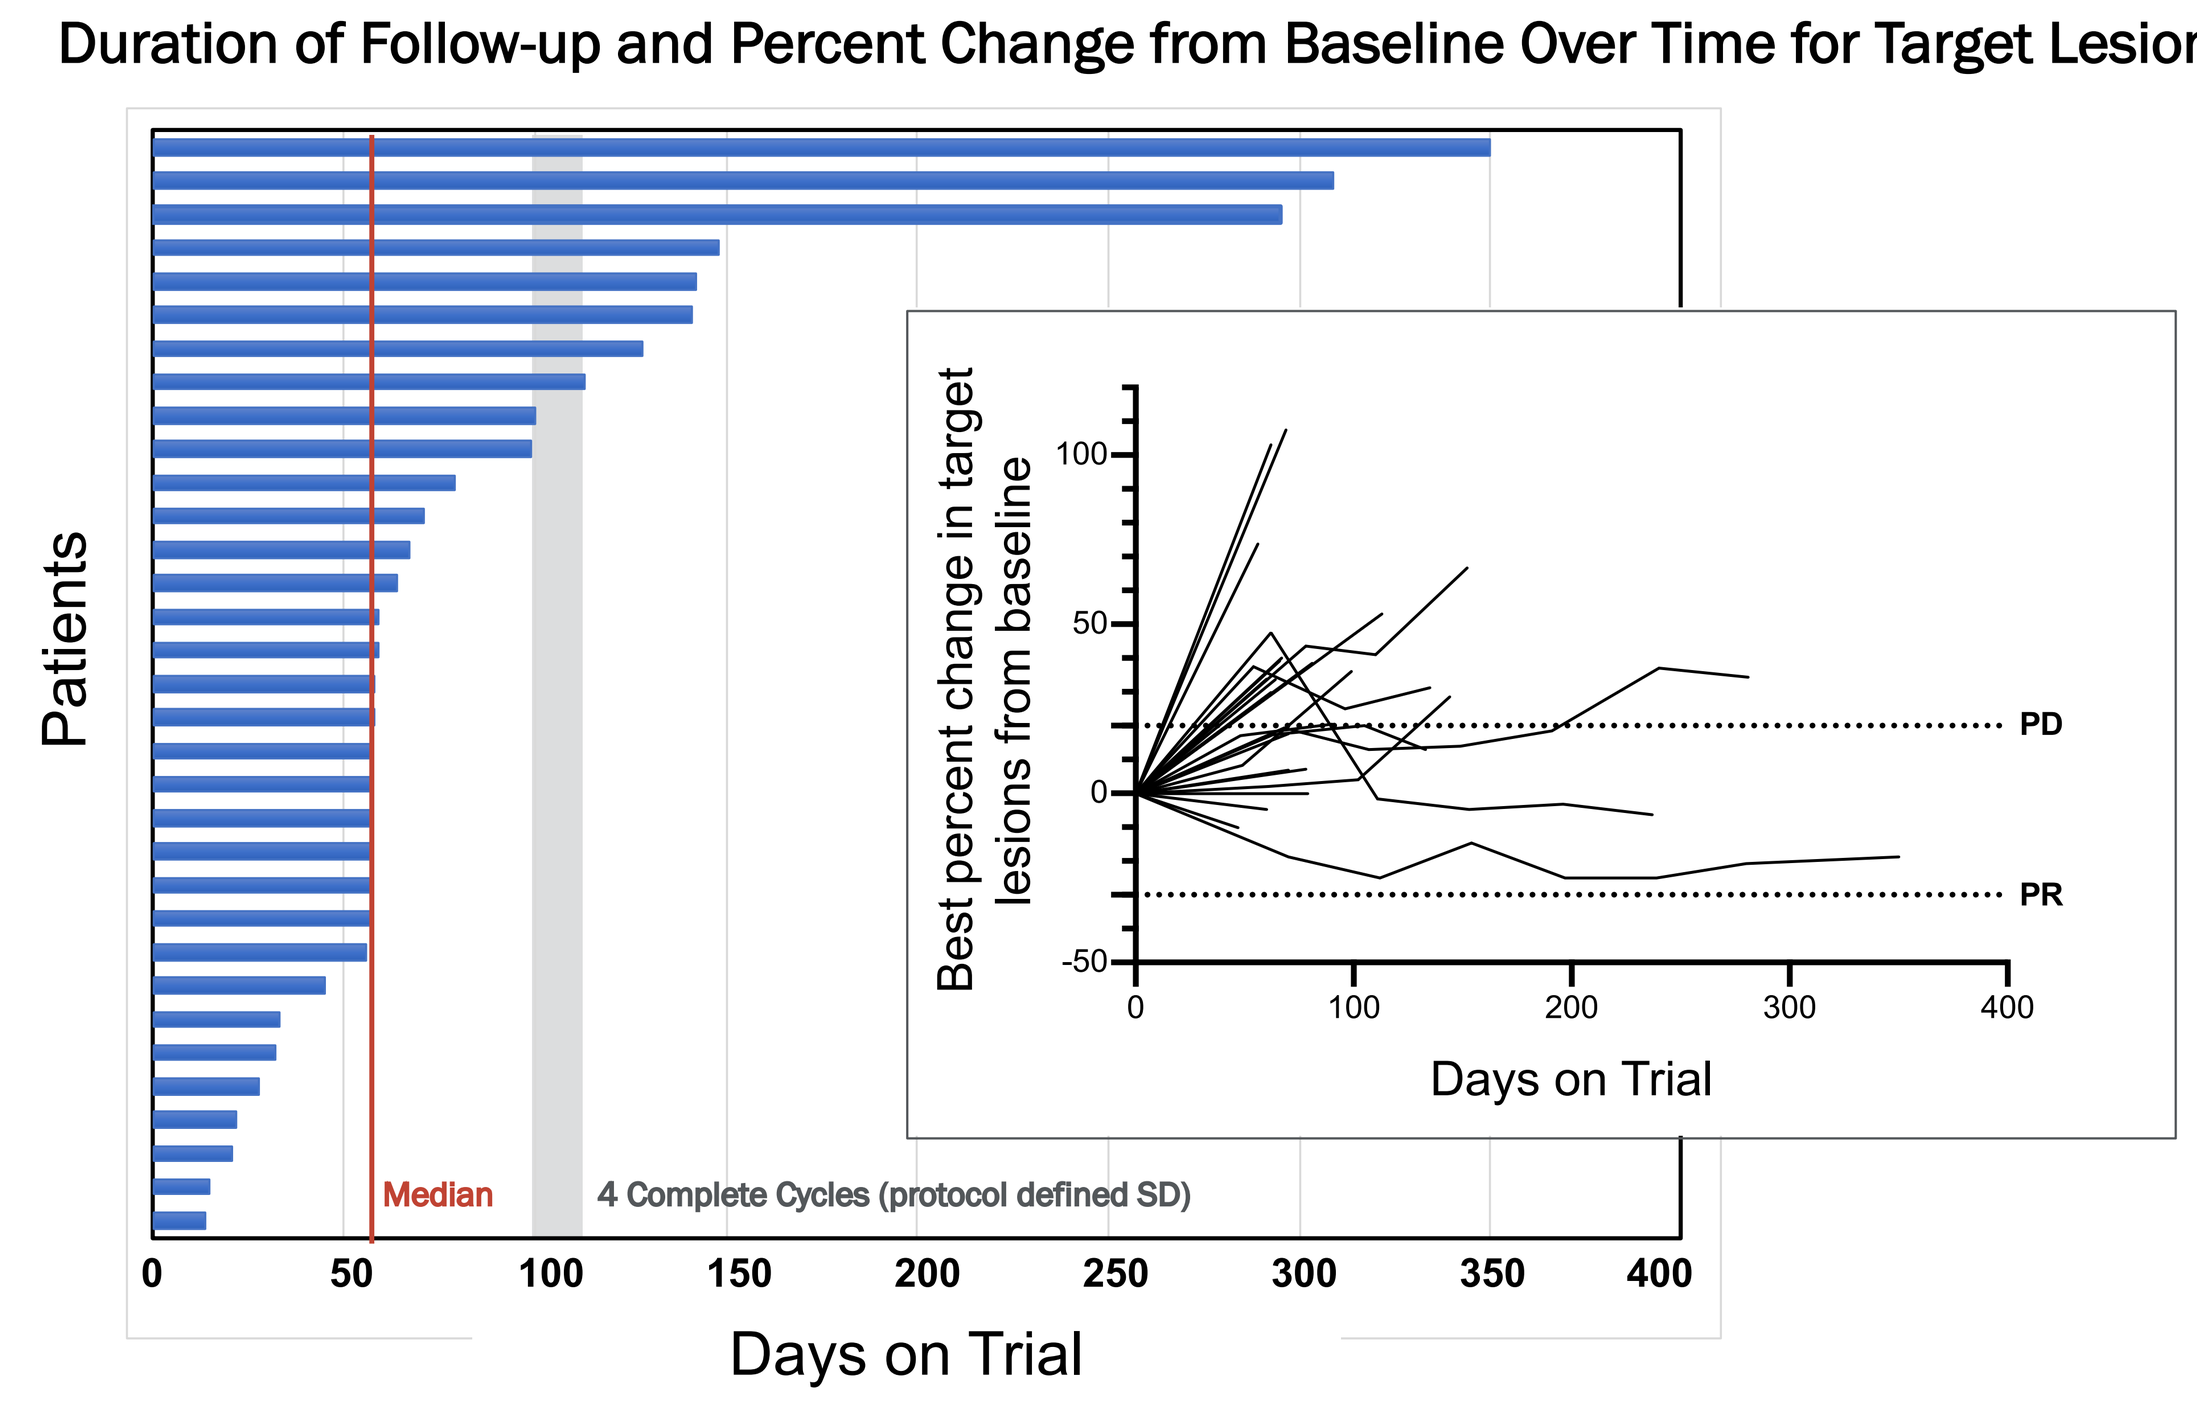
Patients
Median
4 Complete Cycles (protocol defined SD)
Days on Trial
Best percent change in target lesions from baseline
PD
PR
Days on Trial

Fig 4. MSS CRC patients (N=33) were treated with EDP1503 for 2 weeks prior to combination with pembrolizumab. All received EDP1503 at the low dose of 2 enteric capsules twice daily and are currently off study. Median on-treatment time: 58 days. 7 patients had best response of stable disease (SD). 3 had prolonged stable disease of > 9 months.

Preliminary data, investigator assessment of best percentage change in target lesions by RECIST. Data cutoff 29 May 2020. Does not include patients who were not evaluable due to early disease progression or termination prior to first response assessment.

Fig 5. Increased CD8:Treg ratio induced by EDP1503 correlates with treatment duration in MSS CRC

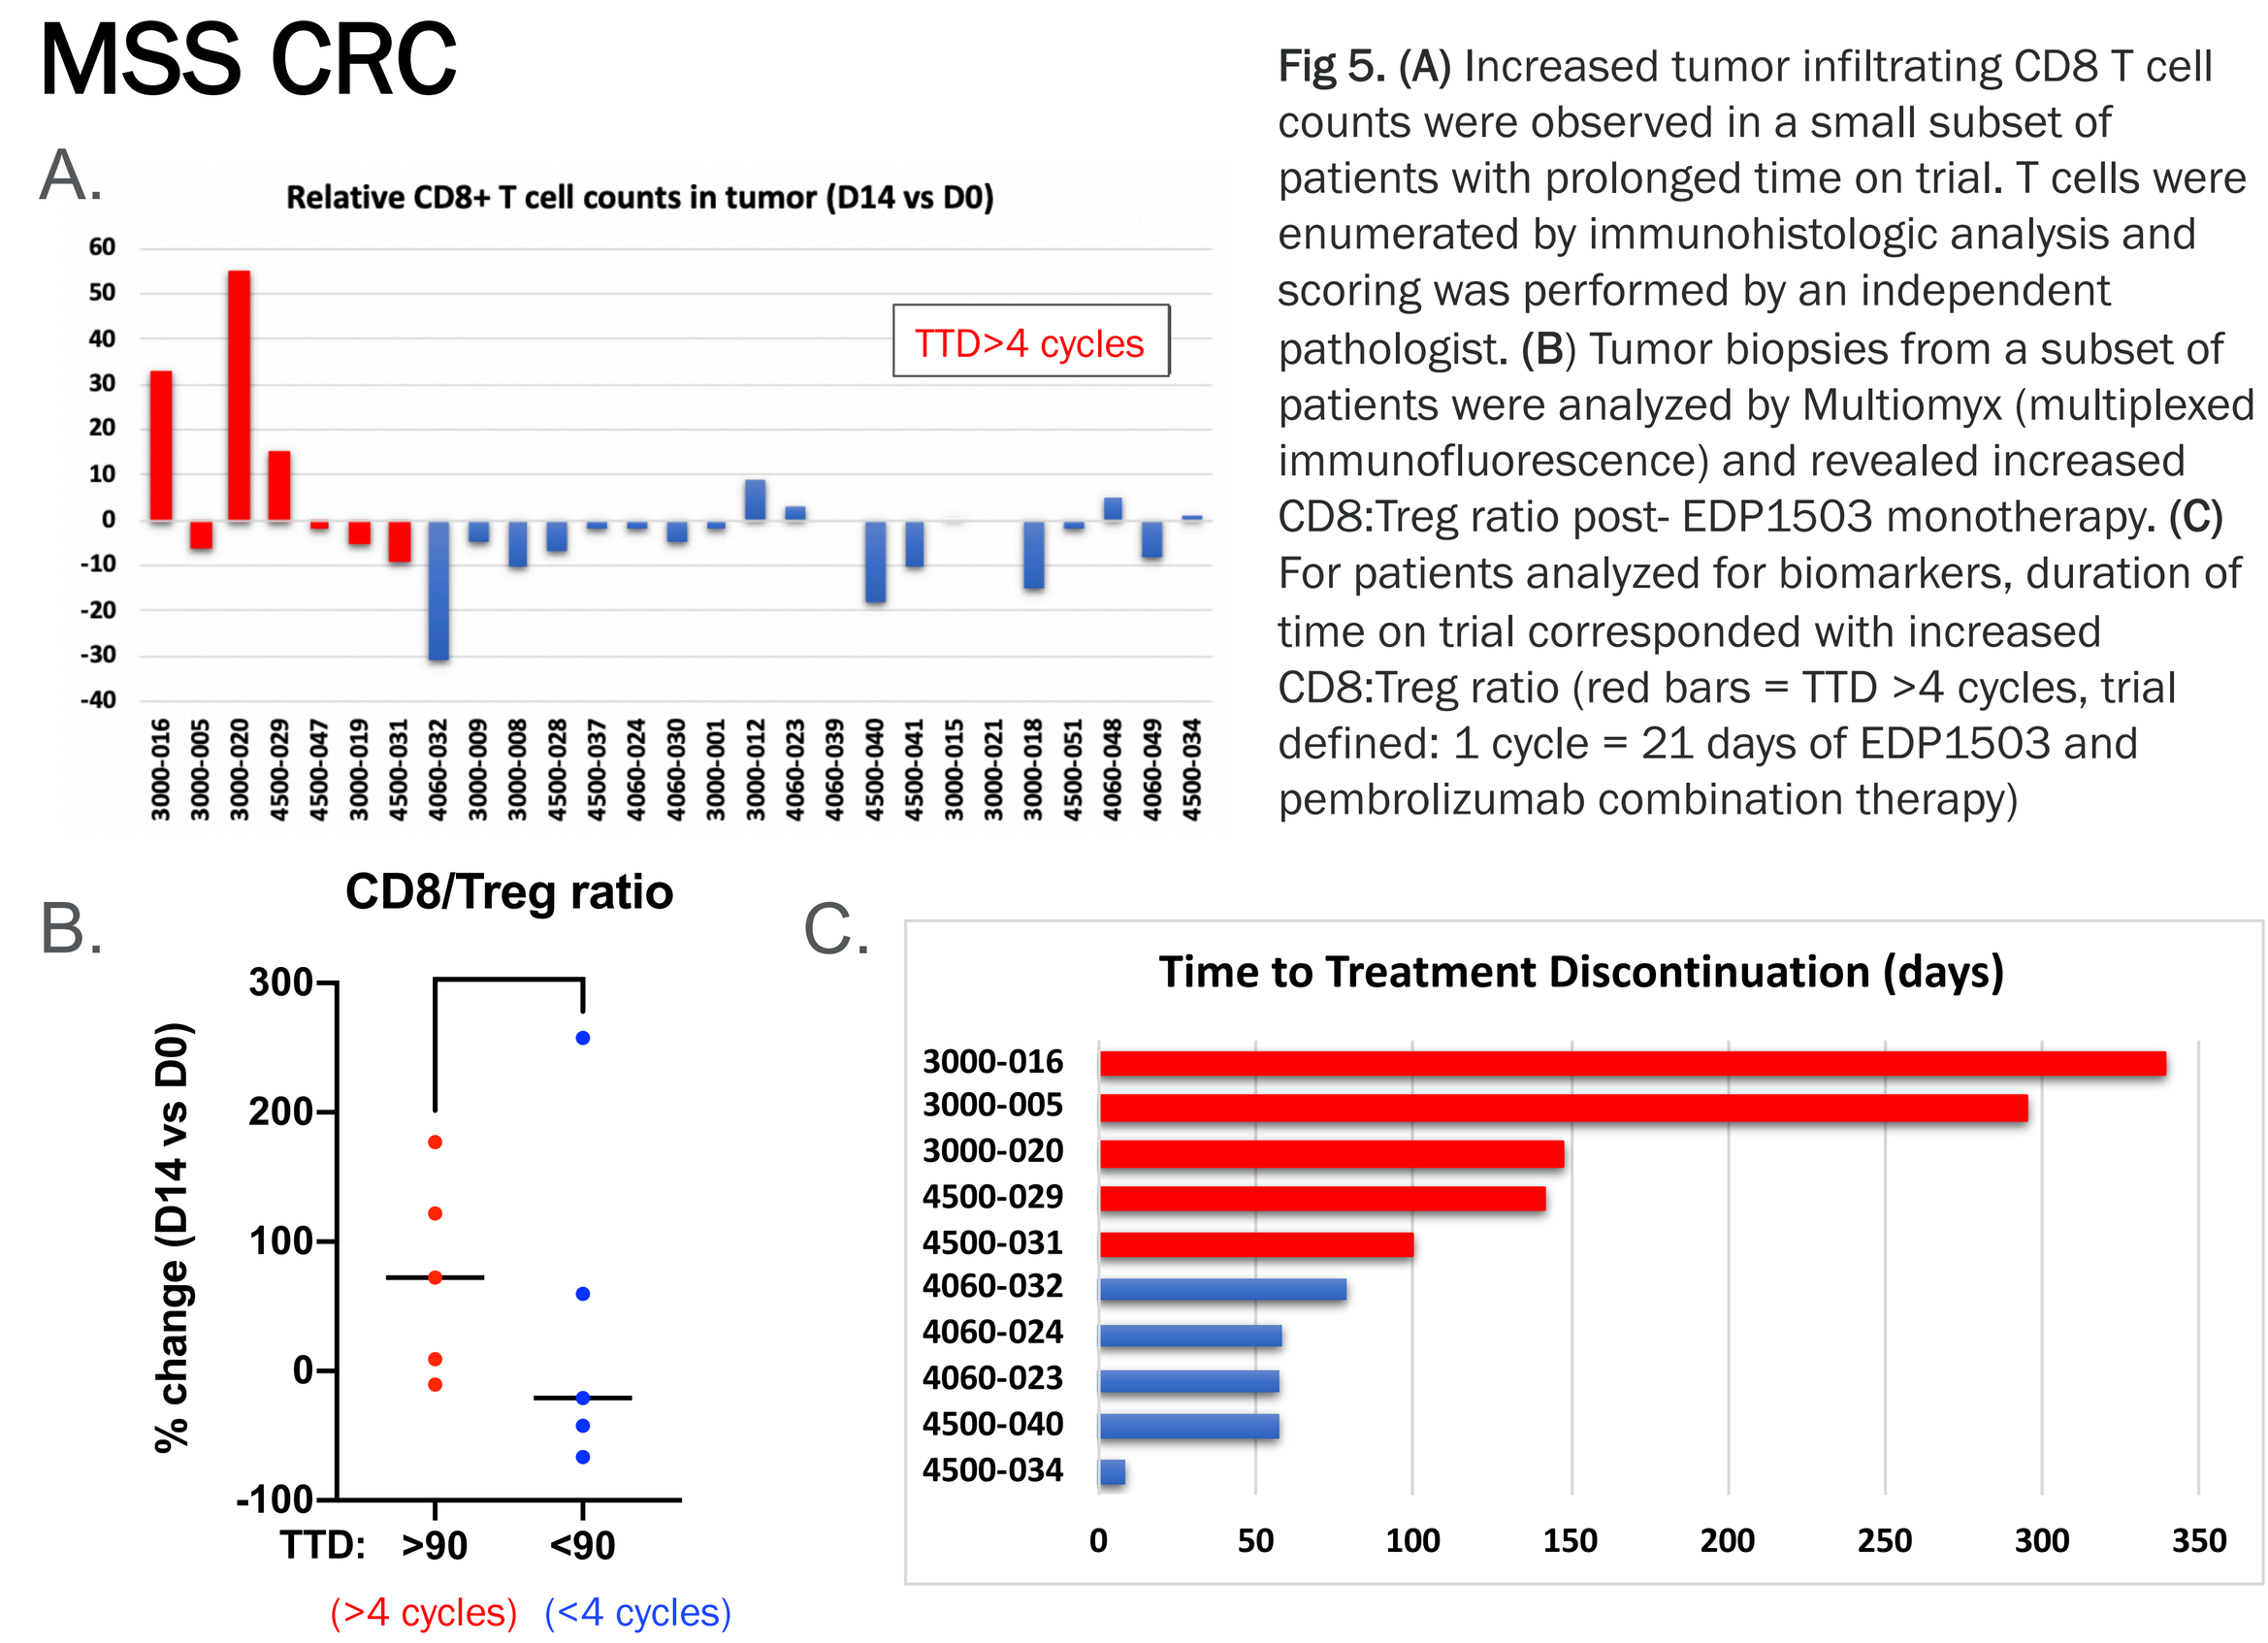
A.
Relative CD8+ T cell counts in tumor (D14 vs D0)
TTD>4 cycles
B.
CD8/Treg ratio
% change (D14 vs D0)
TTD: >90 <90
(>4 cycles) (<4 cycles)
C.
Time to Treatment Discontinuation (days)

Fig 5. (A) Increased tumor infiltrating CD8 T cell counts were observed in a small subset of patients with prolonged time on trial. T cells were enumerated by immunohistologic analysis and scoring was performed by an independent pathologist. **(B)** Tumor biopsies from a subset of patients were analyzed by Multiomyx (multiplexed immunofluorescence) and revealed increased CD8:Treg ratio post- EDP1503 monotherapy. **(C)** For patients analyzed for biomarkers, duration of time on trial corresponded with increased CD8:Treg ratio (red bars = TTD >4 cycles, trial defined: 1 cycle = 21 days of EDP1503 and pembrolizumab combination therapy)

Fig 6. Additional Tumor Types

A. Duration of Follow-up, Percent Change from Baseline Over Time for Target Lesions

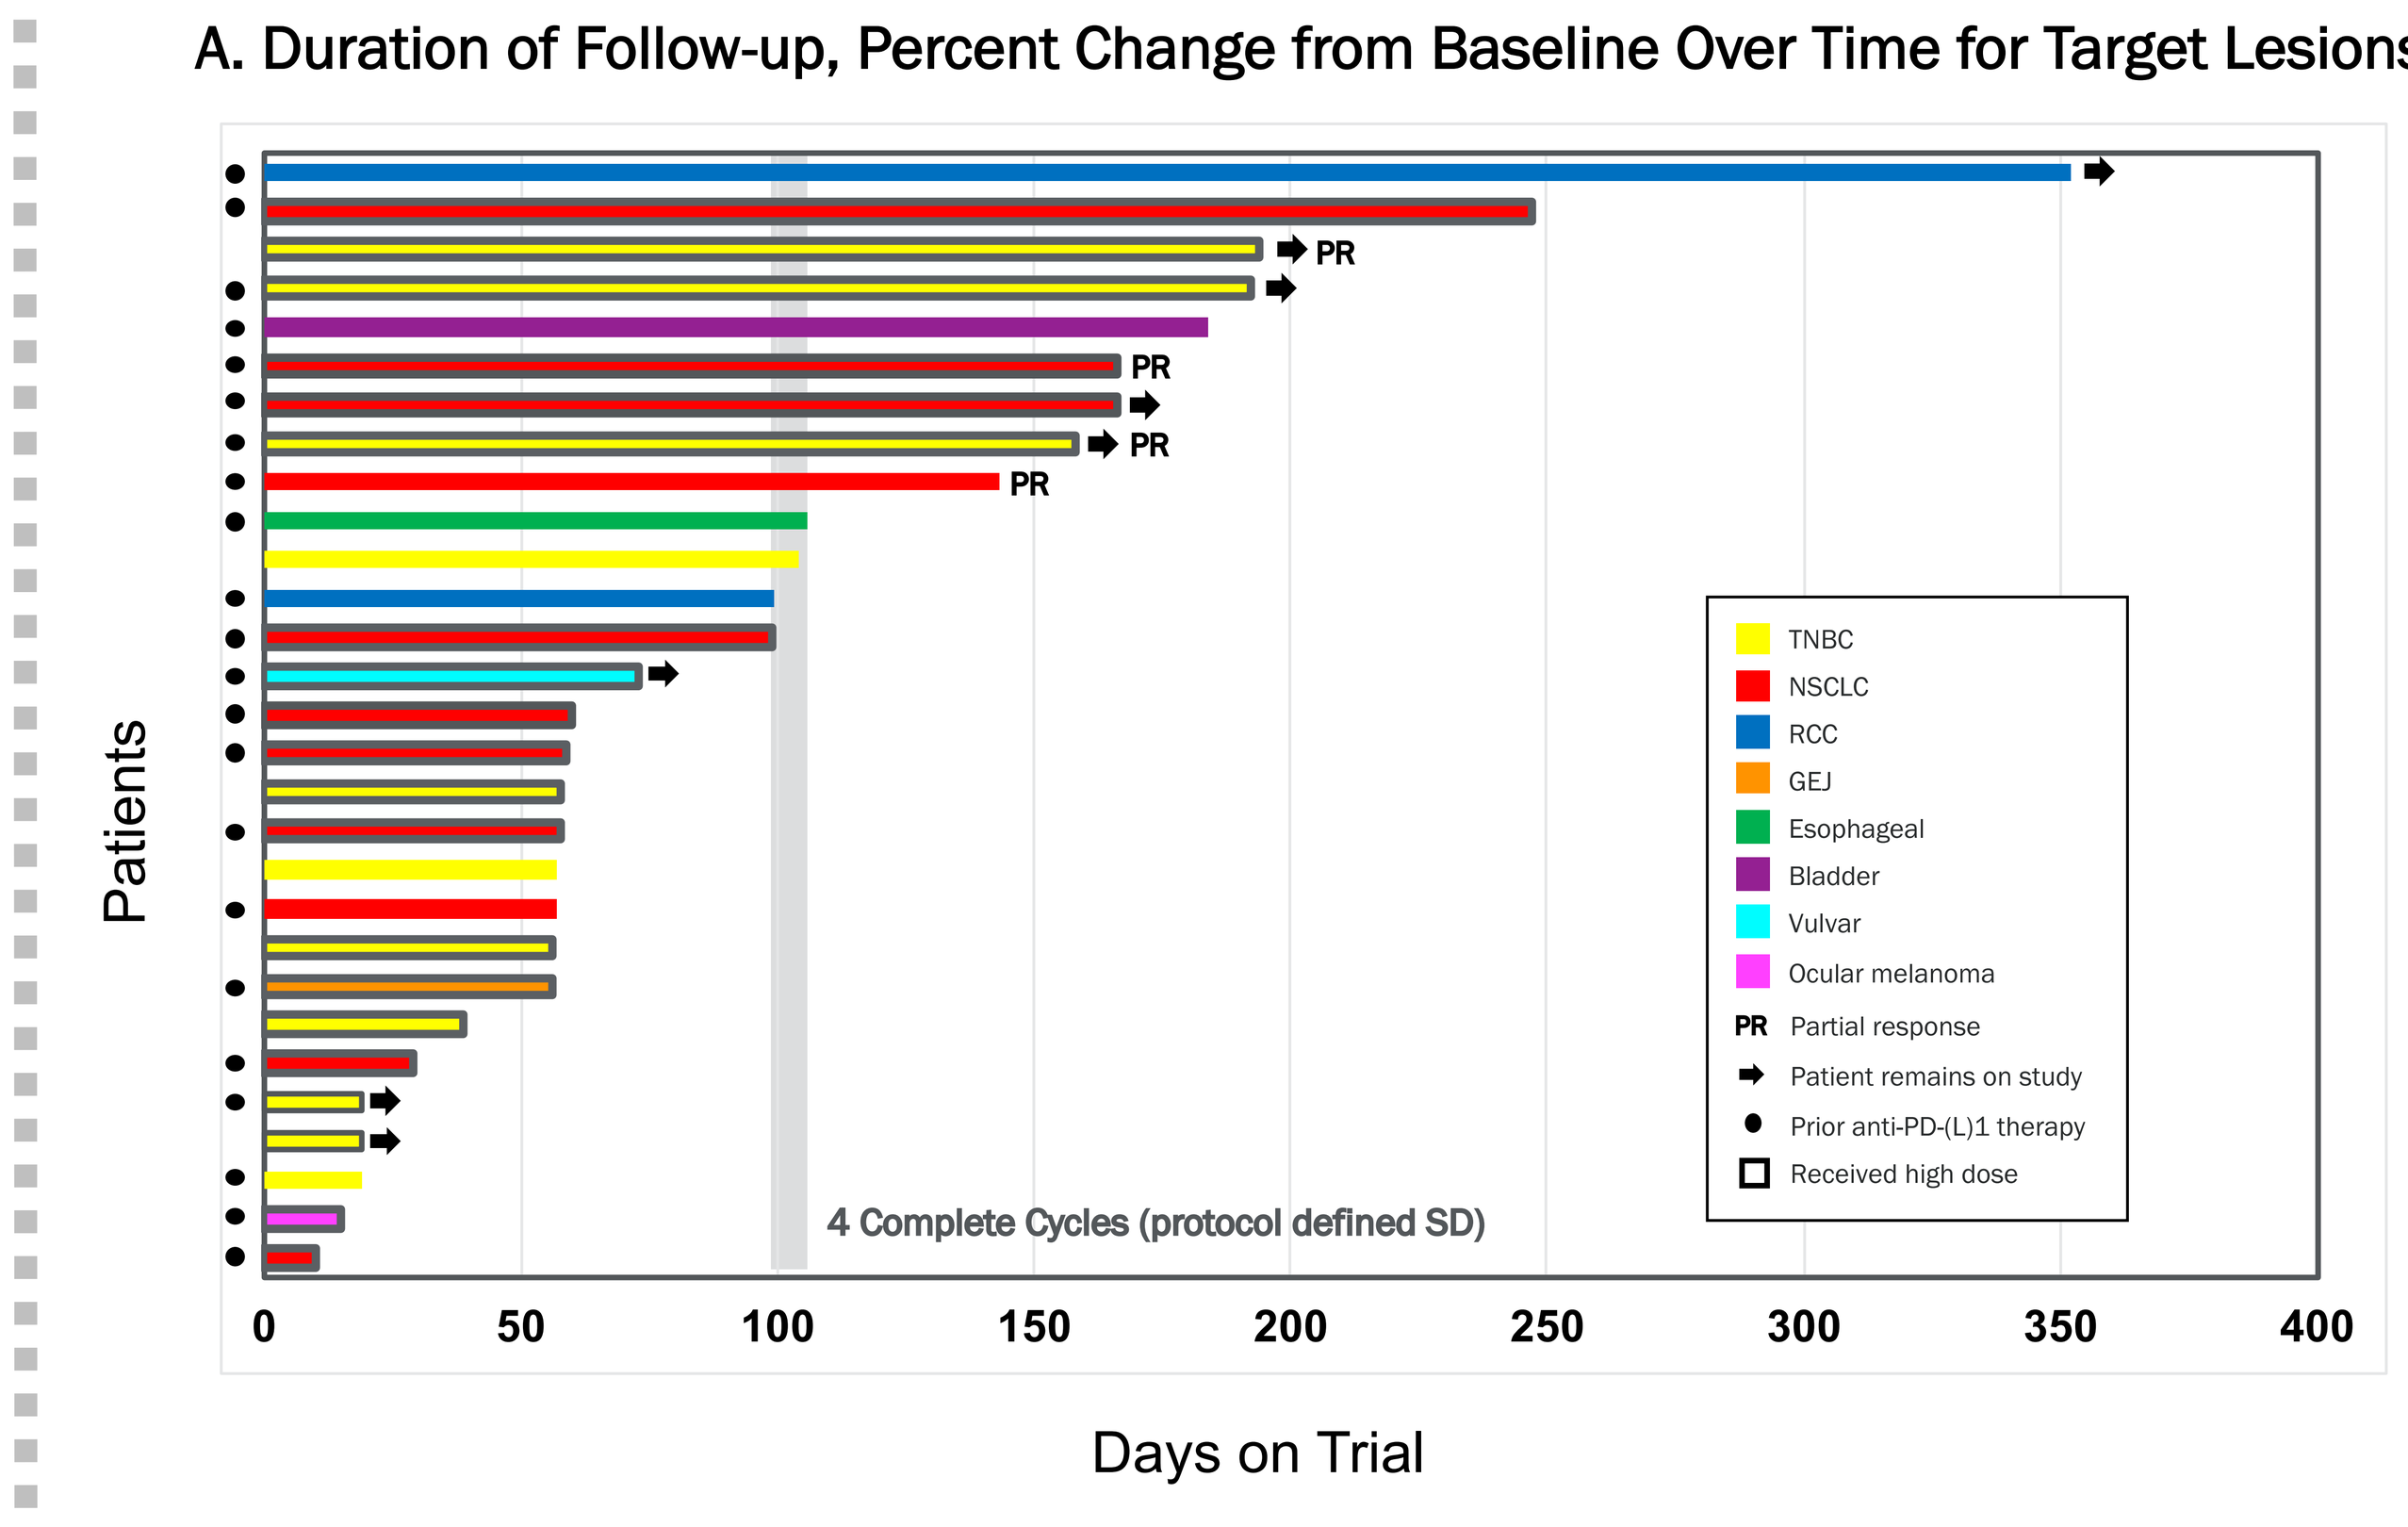
Patients
PR
TNBC
NSCLC
RCC
GEJ
Esophageal
Bladder
Vulvar
Ocular melanoma
PR Partial response
Patient remains on study
Prior anti-PD-(L)1 therapy
Received high dose
4 Complete Cycles (protocol defined SD)
Days on Trial

B. Percent Change from Baseline Over Time for Target Lesions

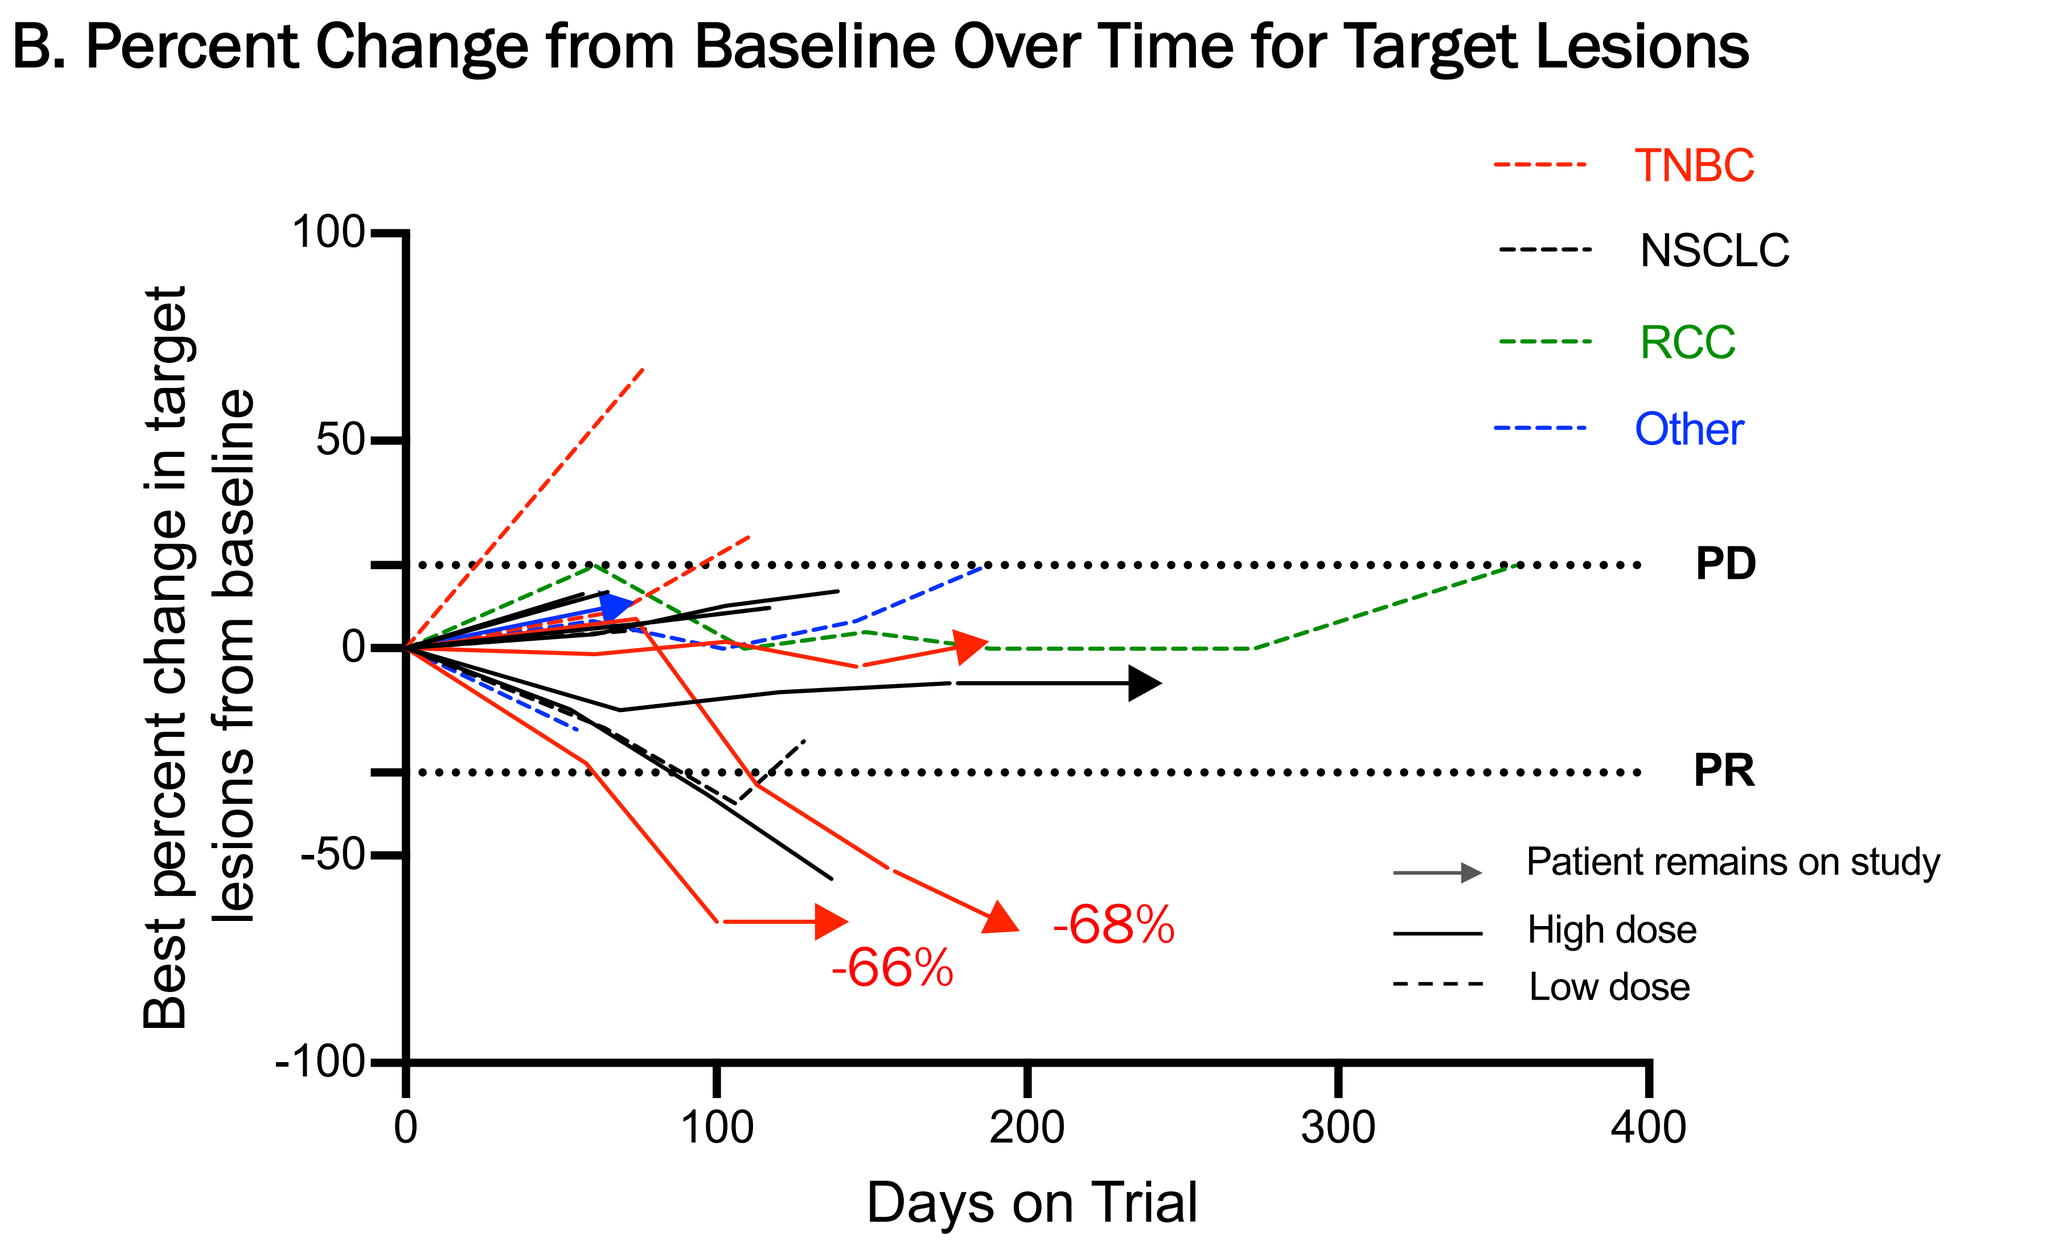
TNBC
NSCLC
RCC
Other
PD
PR
-66%
-68%
Patient remains on study
High dose
Low dose
Best percent change in target lesions from baseline
Days on Trial

Fig 6. Patients with additional tumor types (N=29) were treated with EDP1503 for 2 weeks prior to combination with pembrolizumab. Patients received EDP1503 at either a low dose of 2 enteric capsules twice daily or a high dose of 4 enteric capsules twice daily. 4 patients had partial responses (2 TNBC, 2 NSCLC) and 5 had stable disease (1 TNBC, 2 NSCLC, 1 RCC, 1 bladder).

Preliminary data, investigator assessment of best percentage change in target lesions by RECIST. Data cutoff 29 May 2020. Does not include patients not evaluable due to early disease progression or termination prior to assessment of first response assessment, or awaiting first response assessment at time of data cutoff.

CONCLUSIONS

- Single strains of microbes can alter systemic immunology without systemic exposure.
- Preclinically, orally delivered EDP1503 has multiple effects on the systemic immune system:
 - Induces a proinflammatory gene signature (CXCL10, IL12p70, IFNγ, IL-1β, TNFα and IL-6)
 - Activates effector lymphocyte populations (NK, NKT, CD4⁺, CD8⁺)
 - Induces a proinflammatory chemotactic signature within the TME (data not shown)
- The small intestine is the key site of action of EDP1503.
- Colonization is not required for pharmacological activity.
- The combination of EDP1503 and pembrolizumab is safe and well-tolerated with no treatment-related Grade 4/5 AEs or SAEs.
- Evidence of clinical translational of the preclinical data has been observed in subjects with TNBC and NSCLC.
- Partial responses were observed in 2 patients with TNBC and 2 with NSCLC, for an ORR of 14% (4/29) across additional tumor types. Both TNBC patients have ongoing partial responses.
- In patients with TNBC who received the higher EDP1503 dose of 4 capsules BID, an ORR of 25% (2/8) was observed.
- Although no objective responses were observed in patients with MSS CRC, a DCR of 21% (7/33) was observed, with 3 patients having prolonged stable disease of greater than 9 months.
- Biomarker analyses of MSS CRC patients pre- and post- 2 weeks of EDP1503 monotherapy indicated that increased CD8:Treg ratios induced by EDP1503 were correlated with treatment duration.
- The biomarker data also provides evidence that EDP1503 is driving part of the observed clinical responses.
- Based on preliminary efficacy results, this study is continuing to recruit TNBC patients at the high dose of EDP1503.

REFERENCES

1. Sivan A., *et al.*, **Commensal Bifidobacterium promotes antitumor immunity and facilitates anti-PD-L1 efficacy.** Science. 2015 Nov 27;350(6264):1084-9.
2. Routy B., *et al.*, **Gut microbiome influences efficacy of PD-1-based immunotherapy against epithelial tumors.** Science. 2018 Jan 5;359(6371):91-97.
3. Gopalakrishnan V., *et al.*, **Gut microbiome modulates response to anti-PD-1 immunotherapy in melanoma patients.** Science. 2018 Jan 5;359(6371):97-103.
4. Matson V., *et al.*, **The commensal microbiome is associated with anti-PD-1 efficacy in metastatic melanoma patients.** Science. 2018 Jan 5;359(6371):104-108.

Acknowledgements: This study was sponsored by Evelo Biosciences in collaboration with Merck & Co., Inc., Kenilworth, NJ USA. We thank the patients and their families, as well as all investigators and site personnel, for participating in this study.